Weight Change in Trial of Switching from Second-line bPI to B/F/TAF in a Context of Food Insecurity

0896

Fabienne Homeus[1], Samuel Pierre[1], Jean Bernard Marc[1], Jodany Bernadin[1], Vanessa Rivera[1], Sean E. Collins[2], Letizia Trevisi[3], Abigail Zion[4], Adina Zhang[4], Elizabeth Fox[5], Jean W. Pape[1,6], Serena Koenig[3,7], Paul Sax[3,7], Vanessa Rouzier[1]

[1]Les Centres GHESKIO, Port-au-Prince, Haiti, [2]Gilead Sciences, Inc., Foster City, CA, [3]Harvard Medical School, Boston, MA, [4]Analysis Group, Inc. Boston, MA, [5]Cornell University, Ithaca, NY, [6]Weill Cornell Medical College, New York, NY, [7]Brigham and Women's Hospital, Boston, MA

BACKGROUND

Data are limited on weight gain after switching from second-line boosted PI-based regimens to B/F/TAF in low and middle-income countries (LMICs). We assessed weight in a clinical trial that compared continuing boosted PI + NRTIs vs. B/F/TAF during a period of severe civil unrest in Haiti.

METHODS

The parent study was a prospective, open-label trial conducted at GHESKIO in Port-au-Prince, Haiti. Adults (≥18 years) with viral suppression on second-line PI/r-based ART were randomized to continue their current regimen vs. switch to B/F/TAF. In this secondary analysis, we assessed weight gain and food insecurity.

TABLE 1. BASELINE CHARACTERISTICS BY GROUP

	B/F/TAF	bPI
Age—median (IQR)	49·5 (43·6, 56·2)	48·0 (40·5, 57·4)
Female—n (%)	90 (58·8)	83 (56·1)
BMI—median (IQR)	23·3 (20·3, 27·0)	23·3 (21·5, 27·1)
<18.5	15 (9·8)	12 (8·1)
18.5 to 24.9	85 (55·6)	75 (50·7)
25 to 29.9	33 (21·6)	40 (27·0)
≥30	19 (12·4)	21 (14·2)
CD4 count (cells/mm³)—median (IQR)	531 (393, 719)	552 (390, 698)
CD4 count category—n (%)		
<100	1 (0·7)	1 (0·7)
100 to 349	27 (17·7)	28 (18·9)
350 to 499	38 (24·8)	34 (23·0)
≥500	79 (51·6)	80 (54·1)
Total duration of ART (years)—median (IQR)	10·8 (8·0, 12·8)	9·6 (7·1, 12·0)
Duration of ritonavir-boosted PI regimen (years)—median (IQR)	3·4 (1·9, 5·5)	4·1 (2·5, 6·2)
Ritonavir-boosted Protease Inhibitor—n (%)		
Atazanavir	63 (41·2)	58 (39·2)
Lopinavir	90 (58·8)	90 (60·8)
NRTIs—n (%)		
TDF+FTC/3TC	118 (77·1)	116 (78·4)
AZT+3TC	30 (19·6)	24 (16·2)
ABC+3TC	5 (3·3)	8 (5·4)

At Week 48, the median change in weight was significantly higher in the B/F/TAF group than in the bPI group and weight gain was higher in females than males.

The proportion of participants with severe food insecurity increased significantly during the study, yet most of the B/F/TAF group gained weight while most of the bPI group had weight loss between baseline and week 48.

FIGURE 1. MEDIAN CHANGE IN WEIGHT FROM BASELINE TO 48 WEEKS

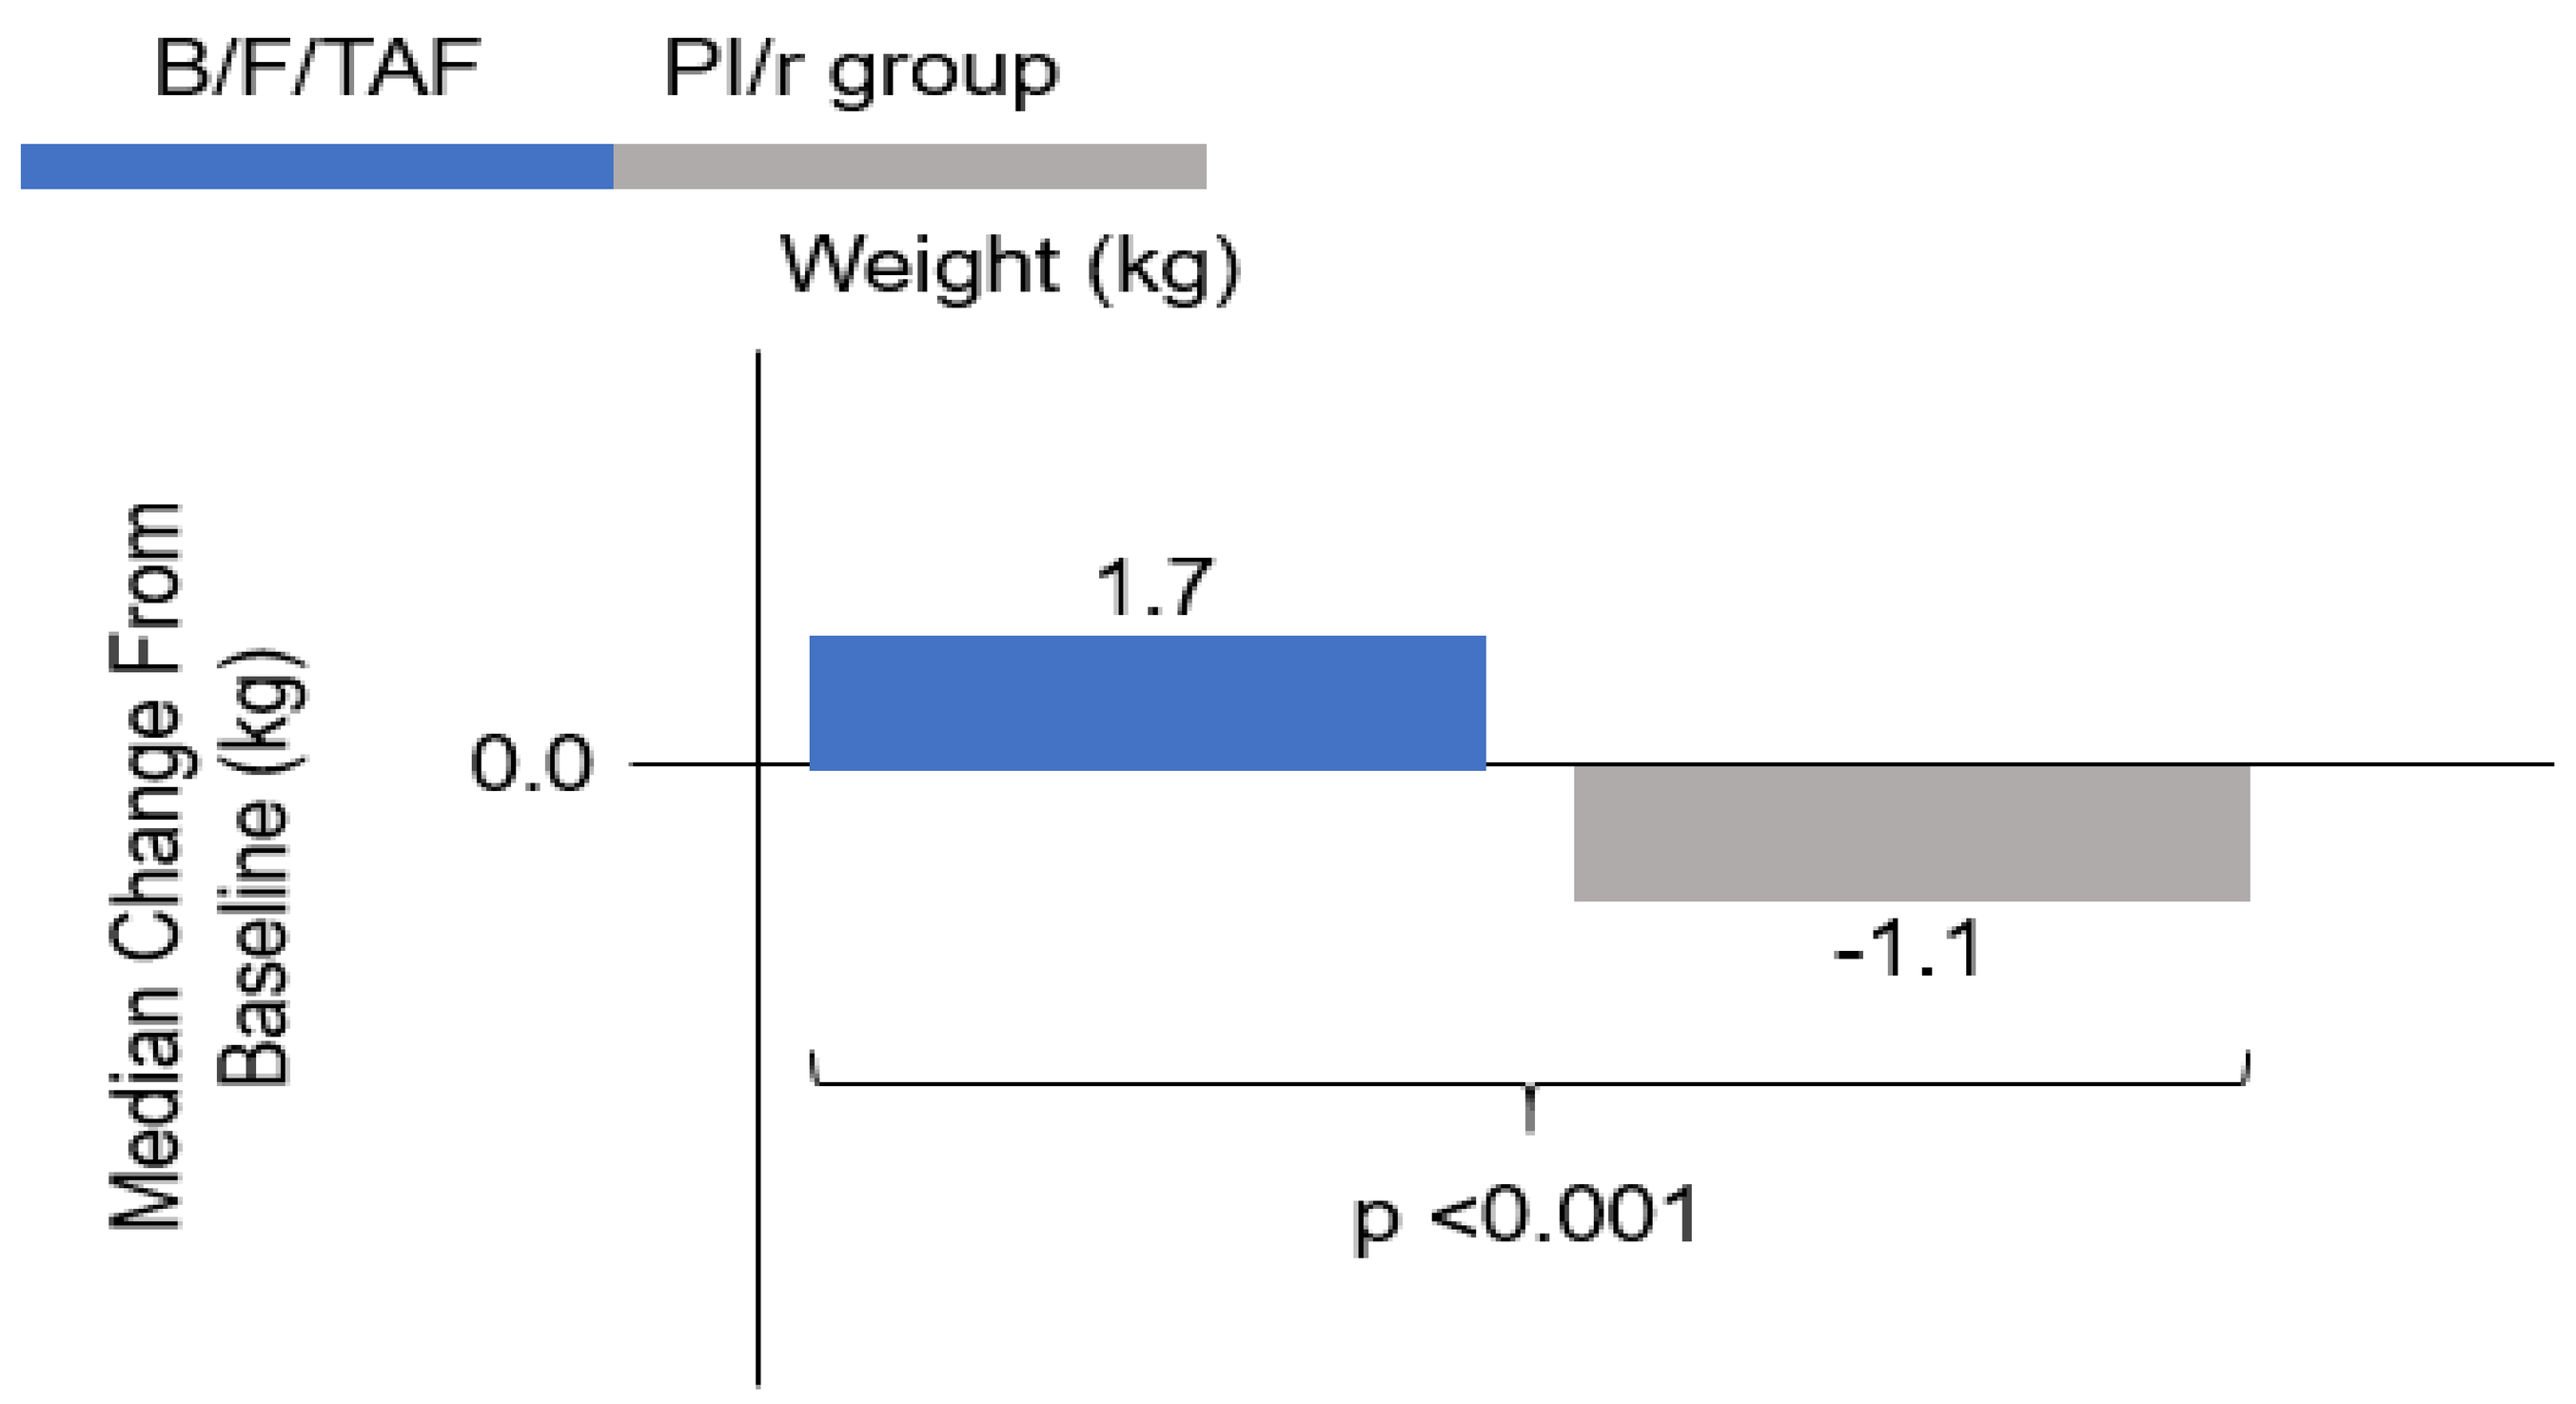
B/F/TAF
PI/r group
Weight (kg)
Median Change From Baseline (kg)
0.0
1.7
-1.1
p <0.001

TABLE 2. WEIGHT CHANGE AND FOOD INSECURITY AT 48 WEEKS

	Females			Males		
	B/F/TAF	**bPI**	**p-value**	**B/F/TAF**	**bPI**	**p-value**
Median change in weight (kg)	+2.90	-0.80	<0.001	+0.60	-1.70	<0.001
Median change in BMI (kg/m²)	+1.16	-0.35	<0.001	+0.18	-0.59	<0.001
≥5% change in body weight	42 (50.6%)	10 (13.2%)	<0.001	13 (21.3%)	2 (3.3%)	<0.004
≥10% change in body weight	18 (21.7%)	2 (2.6%)	<0.001	5 (8.2%)	2 (3.3%)	p=0.439
Low food insecurity	15 (17.9%)	14 (19.4%)		9 (15.5%)	12 (21.4%)	
Moderate food insecurity	27 (32.1%)	21 (29.2%)		24 (41.4%)	15 (26.8%)	
Severe food insecurity	42 (50.0%)	37 (51.4%)		25 (43.1%)	29 (51.8%)	

RESULTS

Between October 2020 and April 2023, 301 participants were enrolled and randomized (B/F/TAF: 153; bPI: 148). Median age was 49 years (IQR 42, 56), 173 (57%) were women, the median BMI was 23.3 kg/m2 (IQR 20.8-27.1) and 28 (9%) had BMI <18.5. At enrollment, 175 (60%) were taking lopinavir/r and 115 (40%) atazanavir/r; 226 (78%) were taking tenofovir disoproxil fumarate, 51 (18%) zidovudine, and 13 (4%) abacavir; all were taking lamivudine or emtricitabine. At Week 48, the median change in weight was +1.7kg (IQR: -0.6, +5.1) in the B/F/TAF and -1.1kg (IQR: -2.9, +1.1) in the bPI group (p<0.001). In the B/F/TAF group, median weight gain was higher in females than males (+2.9 kg vs. +0.6 kg; p<0.001), and 21.7% of females vs. 8.2% of males (p<0.001) gained □10% body weight (Table 1). In contrast, the bPI group had median weight loss of -0.8kg in females and -1.7 kg in males. In the total cohort, the proportion of participants with severe food insecurity increased from 40.1% at enrollment to 49.2% at 48 weeks (p<0.001). There was no difference in the proportion with severe food insecurity at enrollment or 48 weeks by group.

CONCLUSIONS

Weight gain was common among females who switched from PI/r-based regimens (78% included TDF) to B/F/TAF, in the context of high rates of food insecurity. This is consistent with findings of similar drug switches in other settings. In contrast, we observed weight loss in the standard of care group, which may have different clinical implications in settings with high rates of food insecurity. Understanding the implications of weight gain after stopping TDF is essential to guide global policies on TDF-sparing regimens.

ACKNOWLEDGEMENTS

We would like to thank:

- The study participants and their families and friends for their time and participation in this study.
- GHESKIO study, clinical and laboratory staff whose dedication to clients and research participants has been unwavering in the face of unprecedented challenges
- This study was funded by Gilead Sciences, Inc