Outcomes in Treatment-Experienced People With HIV and Detectable Viremia Switching to B/F/TAF in Routine Clinical Practice: 24-Month Results From the BICSTaR Study

TUPEB092

BICSTaR

Berend van Welzen[1], Sam McConkey[2], Marta Boffito[3], Alexander Wong[4], Giulia Marchetti[5], Alexy Inciarte[6], Daniel Elbirt[7], Christoph Stephan[8], Yeşim Taşova[9], Hung-Chin Tsai[10], Jun Yong Choi[11], Claudine Duvivier[12], Chiaw Yee Choy[13], David Thorpe[14], Andrea Marongiu[14], Tali Cassidy[14], Katsuji Teruya[15]

[1]University Medical Centre Utrecht, Utrecht, the Netherlands; [2]Beaumont Hospital, Dublin, Ireland; [3]Chelsea and Westminster Hospital, London, UK; [4]Regina General Hospital, Regina, SK, Canada; [5]University of Milan, ASST Santi Paolo e Carlo, Milan, Italy; [6]Hospital Clínic de Barcelona, Barcelona, Spain; [7]Kaplan Medical Center, Rehovot, Israel; [8]Universitätsklinikum Frankfurt, Frankfurt, Germany; [9]Cukurova University Hospital, Adana, Türkiye; [10]Kaohsiung Veterans General Hospital, Kaohsiung, Taiwan; [11]Yonsei University Severance Hospital, Seoul, Republic of Korea; [12]AP-HP, Hôpital Necker-Enfants Malades, Necker-Pasteur Infectiology Center, Paris, France; [13]National Centre for Infectious Diseases, Singapore; [14]Gilead Sciences Ltd, Uxbridge, UK; [15]National Center for Global Health and Medicine, Tokyo, Japan

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- In treatment-experienced people with HIV and uncontrolled viremia, switching to bictegravir/emtricitabine/tenofovir alafenamide (B/F/TAF) was associated with high rates of virologic suppression and was generally well tolerated
- An increase towards the population norm in 36-Item Short Form Health Survey (SF-36) Mental Component Summary scores indicated improvements in mental wellbeing after switching to B/F/TAF, and participants reported increased satisfaction with HIV treatment

Plain Language Summary

- B/F/TAF is a once-daily tablet used to treat human immunodeficiency virus (HIV). It combines three medicines: bictegravir (B), emtricitabine (F), and tenofovir alafenamide (TAF)
- The BICSTaR study looked at how well B/F/TAF worked and how safe it was for people with HIV in healthcare settings around the world
- In this part of the study, researchers focused on people who still had detectable HIV in their blood when they switched to B/F/TAF from other HIV medicines
- Researchers found that B/F/TAF was highly effective at reducing the amount of HIV in the blood, and most participants had undetectable HIV levels after 2 years of treatment
- People who switched to B/F/TAF had few side effects, which were mostly mild or moderate
- People reported that their mental wellbeing improved after switching to B/F/TAF
- Most people in the study said they were happier with their HIV treatment after switching to B/F/TAF

Introduction

- B/F/TAF is a single tablet regimen recommended in international clinical guidelines for people with HIV[1-3]
- Since marketing approval, cumulative exposure to B/F/TAF is estimated to be 5,807,885 person-years[4]
- BICtegravir Single Tablet Regimen (BICSTaR) was a prospective, observational, 24-month cohort study evaluating the safety and effectiveness of B/F/TAF in people with HIV receiving real-world clinical care[5]
 - The study enrolled 2379 treatment-naïve and treatment-experienced people with HIV-1 from routine clinical practice across 14 countries[5]
 - The pooled analysis showed that B/F/TAF was highly effective and well tolerated through 24 months among a broad range of people with HIV-1[5]
- Multiple studies have demonstrated that B/F/TAF is effective at maintaining virologic suppression in treatment-experienced individuals who are virologically suppressed at the time of switching to B/F/TAF, but data are limited in those with active viremia at the time of switching[6-8]
- Here, we present data on the effectiveness, safety profile, and patient-reported outcomes through 24 months for people with HIV-1 who had detectable viremia at the time of switching to B/F/TAF

Objective

- To describe the effectiveness, safety profile, and patient-reported outcomes through 24 months in treatment-experienced people with HIV-1 with detectable viremia who switched to B/F/TAF during routine clinical care in the BICSTaR study

Methods

Study Design

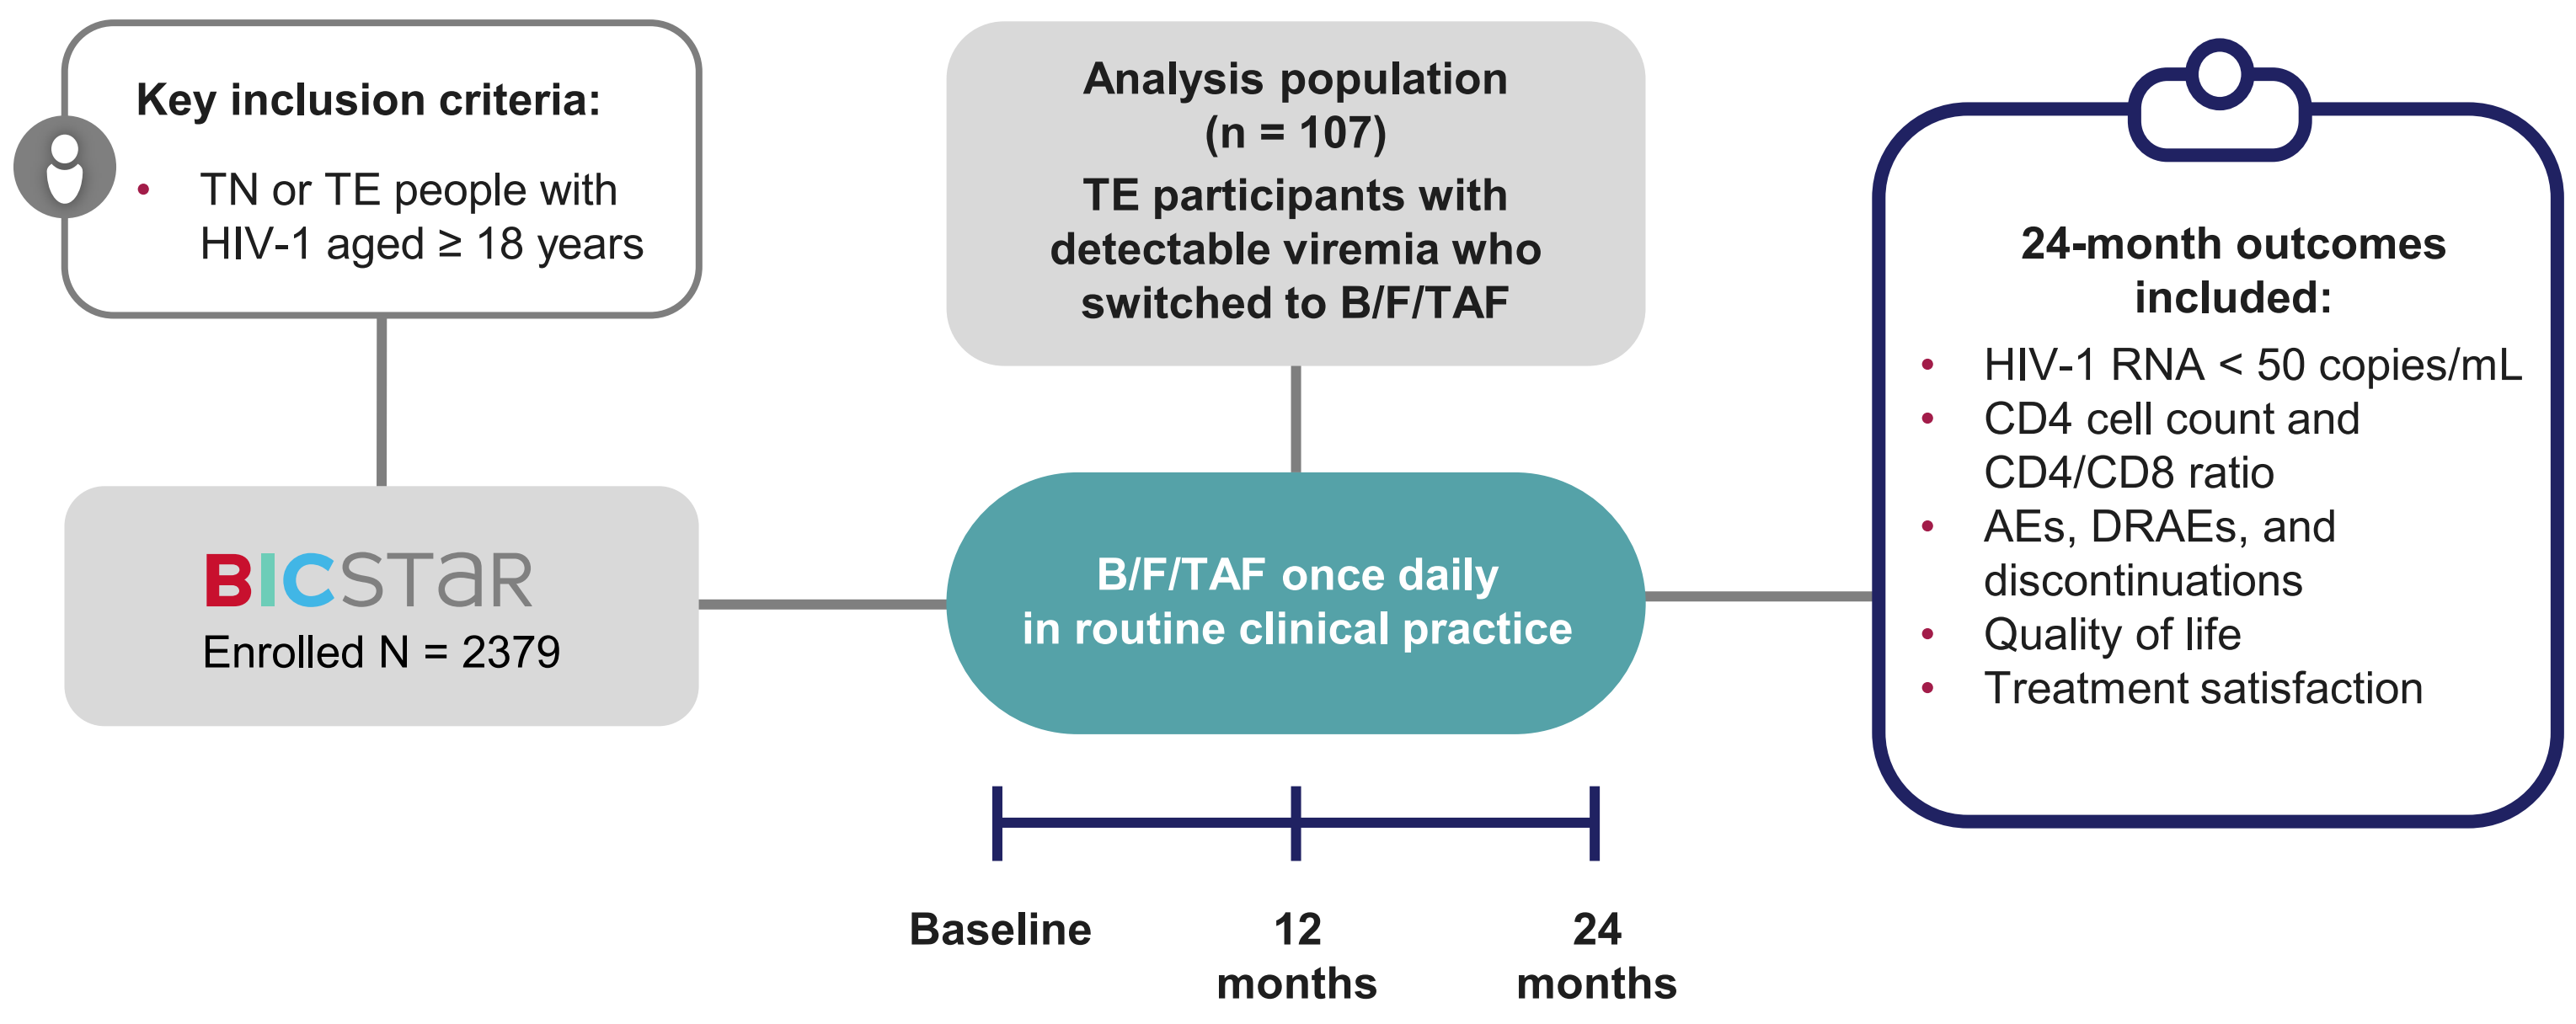

Key inclusion criteria:
TN or TE people with HIV-1 aged ≥ 18 years
BICSTaR
Enrolled N = 2379
Analysis population (n = 107)
TE participants with detectable viremia who switched to B/F/TAF
B/F/TAF once daily in routine clinical practice
Baseline
12 months
24 months
24-month outcomes included:
HIV-1 RNA < 50 copies/mL
CD4 cell count and CD4/CD8 ratio
AEs, DRAEs, and discontinuations
Quality of life
Treatment satisfaction

Outcomes in this analysis[a]:

- Effectiveness assessments: proportion of participants with HIV-1 RNA < 50 copies/mL (M = E), CD4 cell count, and CD4/CD8 ratio
- Safety profile assessments: AEs, DRAEs, B/F/TAF discontinuations
- Quality of life (SF-36 scores)
- Treatment satisfaction (HIVTSQ status and change[b] scores)

[a]Outcomes were evaluated at 24 months, except where indicated. [b]HIVTSQ status: Measures a participant's satisfaction with their current treatment. Scores range from 0 (very dissatisfied) to 60 (very satisfied). HIVTSQ change: Assesses change in treatment satisfaction between a participant's previous and current treatment (evaluated after 12 months in this study to minimize recall bias). Scores range from -30 (deterioration) to +30 (improvement).
AE, adverse event; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; BICSTaR, BICtegravir Single Tablet Regimen; CD, cluster of differentiation; DRAE, drug-related adverse event; HIVTSQ, HIV Treatment Satisfaction Questionnaire; M = E, missing = excluded; SF-36, 36-Item Short Form Health Survey; TE, treatment-experienced; TN, treatment-naïve.

Results

Baseline Demographics

	TE People With HIV-1 and Viremia at Baseline n = 107
Age at B/F/TAF initiation, years, median (Q1, Q3)	46 (34, 52)
≥ 50 years, n (%)	38 (36)
≥ 65 years, n (%)	3 (3)
Sex assigned at birth, n (%)	
Male	85 (79)
Female	22 (21)
Race/ethnicity, n (%)	
White	66 (62)
Black	25 (23)
Asian	10 (9)
Other[a]	6 (6)

[a]Includes American Indian or Alaska Native (n = 1), Hispanic/Latine (n = 1), First Nations Indigenous (n = 1), North African (n = 1), Afro and White (n = 1), and Argentina (n = 1).
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; Q, quartile; TE, treatment-experienced.

Clinical Characteristics

	TE People With HIV-1 and Viremia at Baseline n = 107
HIV-1 RNA at baseline, log_{10} copies/mL, median (Q1, Q3)	2.34 (1.89, 3.65)
HIV-1 RNA viral load, n (%)	
≥ 50 to < 200 copies/mL	50 (47)
≥ 200 to < 1000 copies/mL	21 (20)
≥ 1000 to < 100,000 copies/mL	26 (24)
≥ 100,000 copies/mL	10 (9)
CD4 count at baseline, cells/µL, median (Q1, Q3)	504 (248, 727)
Reason for switching to B/F/TAF, n (%)[a]	
Simplification of ART	46 (43)
Participant's preference	15 (14)
Side effects of current ART	20 (19)
Other	36 (34)
Number of comorbidities ongoing at B/F/TAF initiation, n (%)	
1	27 (25)
2	21 (20)
≥ 3	26 (24)
Any concomitant medication at B/F/TAF initiation, n (%)	64 (62)
Number of previous ART regimens, n (%)[b]	
1	39 (38)
2	23 (22)
≥ 3	41 (40)
History of virologic failure on any ART regimen, n (%)[c]	
No	64 (60)
Yes	27 (25)
At least one primary resistance mutation at baseline, n (%)[d]	
No	40 (38)
Yes	11 (10)

[a]Multiple reasons for switching to B/F/TAF are possible. [b]Data were missing for four participants. [c]Data were missing for one participant and unknown for 15 (14%). [d]Data were missing for one participant and unavailable for 55 (52%).
ART, antiretroviral therapy; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CD, cluster of differentiation; Q, quartile; TE, treatment-experienced.

Virologic Effectiveness Through 24 Months

- The majority of participants transitioned from having uncontrolled viremia at baseline to an undetectable viral load at 24 months
- In the missing = excluded analysis, at 24 months, 87% (65/75) of participants with available data had undetectable viremia[a]
- Among the 10 participants with detectable viremia at 24 months, the median (quartile [Q]1, Q3) HIV-1 viral load was 1.93 (1.79, 3.37) log_{10} copies/mL

Sankey Flow Diagram of Viral Load Trajectories (n = 75)[b]

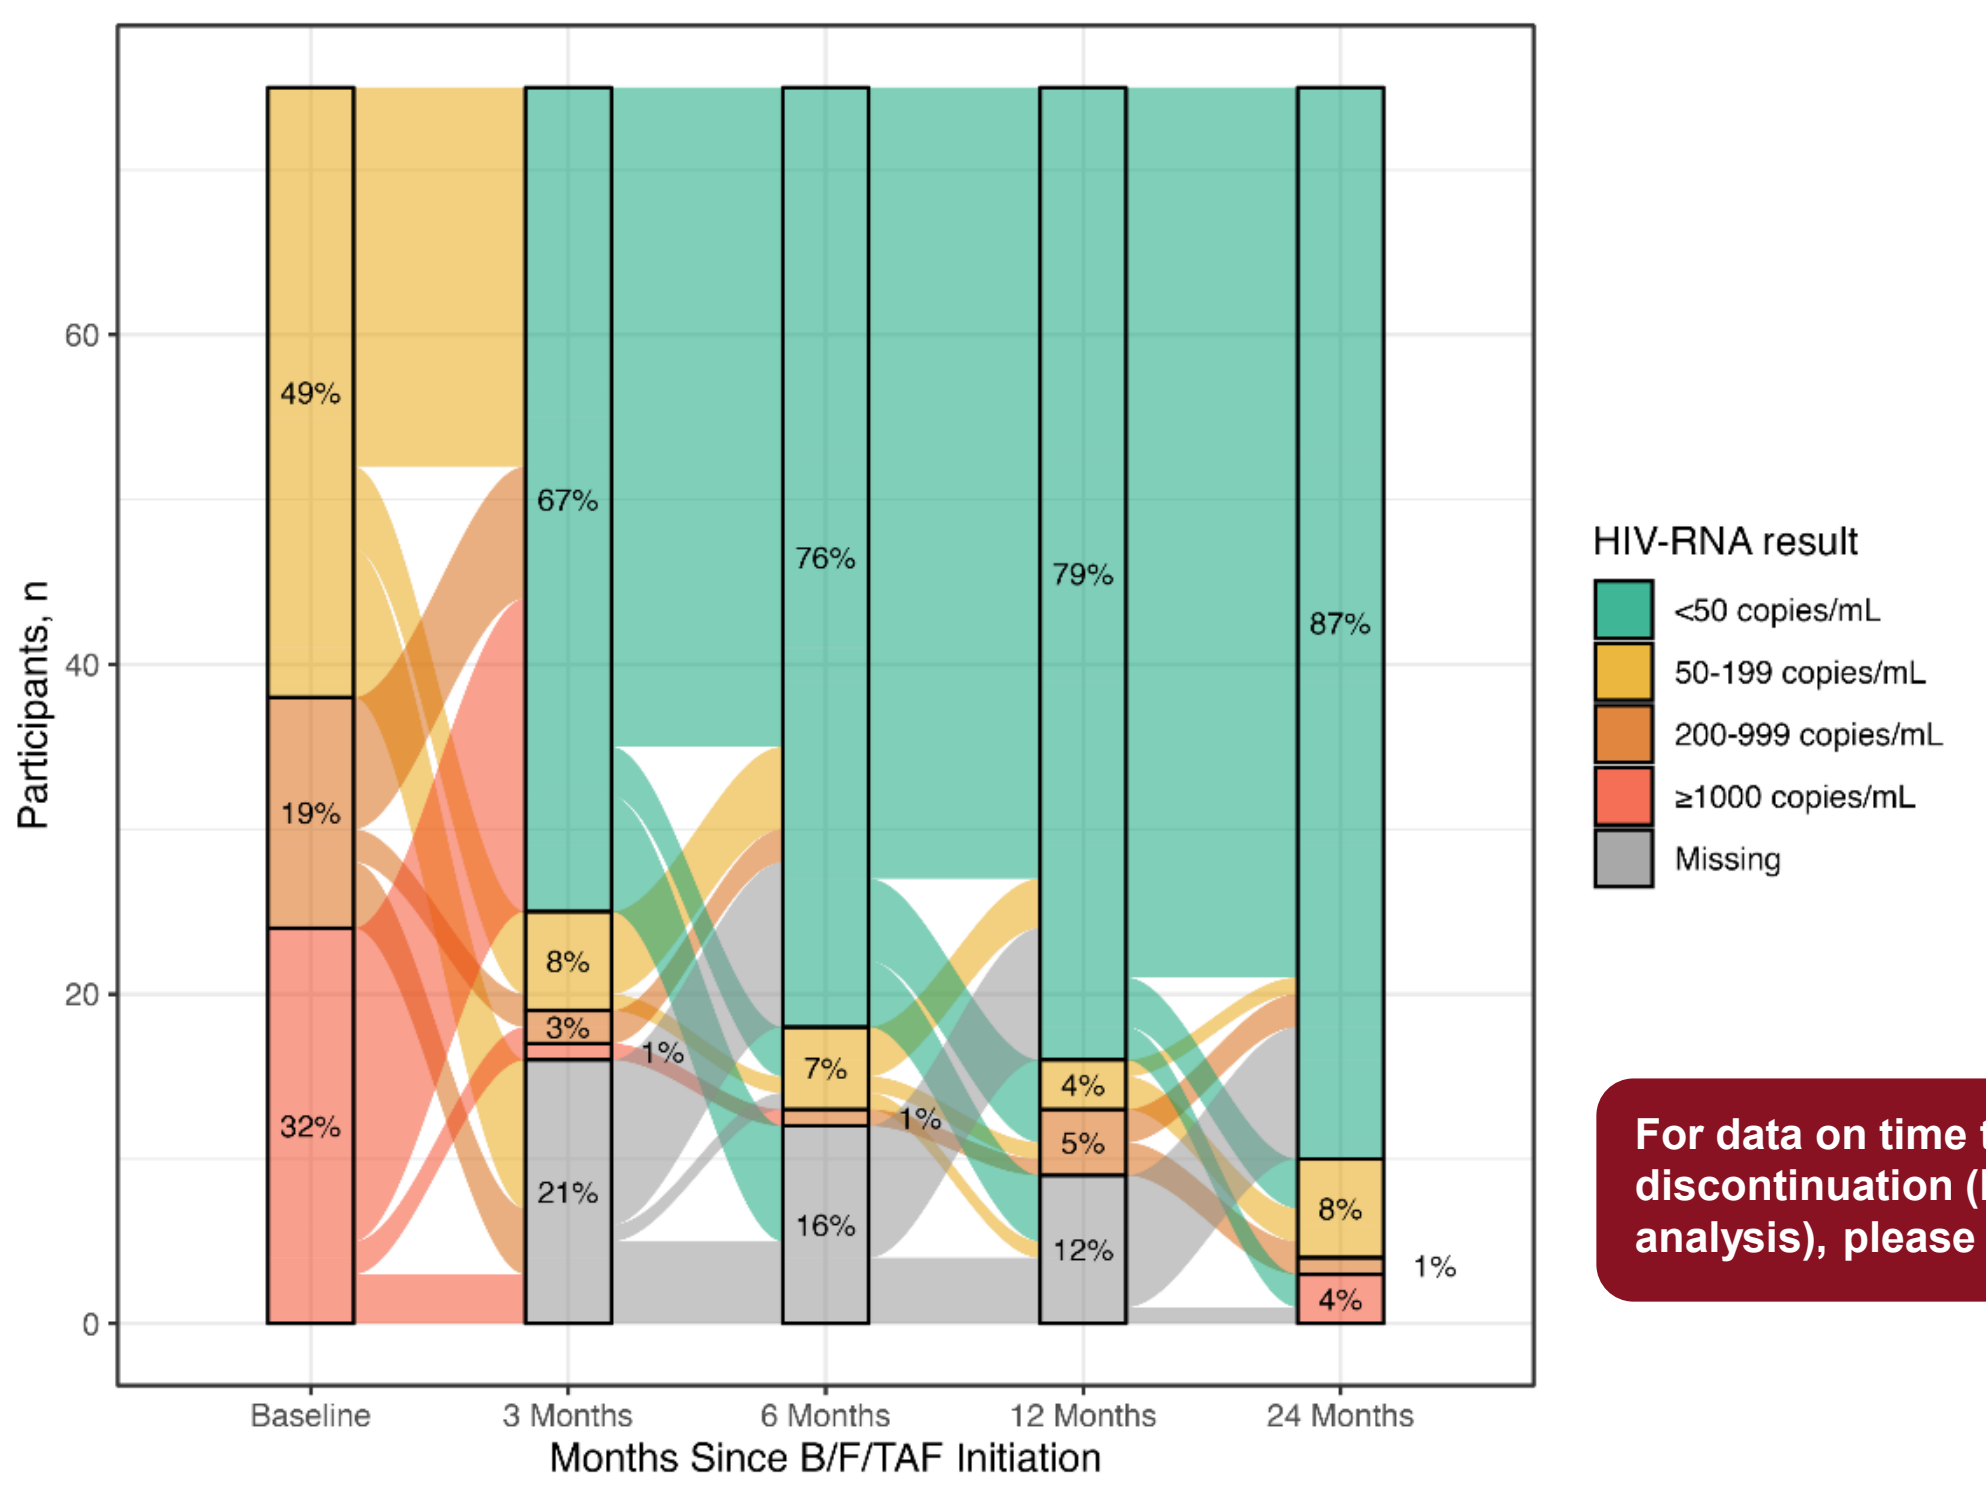

Participants, n
0
20
40
60
49%
19%
32%
67%
8%
3%
1%
21%
76%
7%
1%
16%
79%
4%
5%
12%
87%
8%
4%
1%
Baseline
3 Months
6 Months
12 Months
24 Months
Months Since B/F/TAF Initiation
HIV-RNA result
<50 copies/mL
50-199 copies/mL
200-999 copies/mL
≥1000 copies/mL
Missing

For data on time to B/F/TAF discontinuation (Kaplan-Meier analysis), please scan the QR code

QR CODE

[a]Missing = excluded: participants who discontinued the study and/or B/F/TAF before the 24-month window (18-30 months) and those without viral load data in the 24-month window were excluded (no imputation). [b]The Sankey diagram provides a visualization of the evolution of the population viral load over 24 months. Data shown are for participants with viral load data available at baseline and 24 months. Thicker links represent a larger transition of participants over time. For participants who discontinued B/F/TAF during study follow-up and remained in the study, results collected after discontinuation of B/F/TAF were not considered for analysis.
B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide.

Change in Immunologic Outcomes

	Median (Q1, Q3) at baseline		Median (Q1, Q3) change at 24 months
CD4 count, cells/µL	551 (267, 698)	n = 59[a]	+113 (-25, 205) *P* = 0.002[b]
CD4/CD8 ratio	0.62 (0.40, 0.93)	n = 53[a]	+0.20 (-0.01, 0.30) *P* = 0.002[b]

[a]Partcipants with viremia at baseline and data available at baseline and 24 months. [b]Sign test.
CD, cluster of differentiation; Q, quartile.

Safety Outcomes

- Overall, 11% (12/107) of participants reported ≥ 1 drug-related adverse event (DRAE) within 24 months of B/F/TAF initiation
- DRAEs led to B/F/TAF discontinuation in 6/107 (6%) participants; five had moderate DRAEs and one had a mild DRAE
- No serious DRAEs were reported

Participants, n (%)	TE People With HIV-1 and Viremia at Baseline n = 107
≥ 1 AE	69 (64)
≥ 1 DRAE	12 (11)
Mild	7 (7)
Moderate	6 (6)
Severe	1 (1)
Discontinued B/F/TAF due to DRAEs[a]	6 (6)
Discontinued B/F/TAF due to a "lack of efficacy"[b]	5 (5)
Death	2 (2)[c]

Data are shown for participants with AEs occurring within 24 months of B/F/TAF initiation. [a]The only DRAE occurring in ≥ 1% of participants that led to discontinuation was depression (n = 2; 2%). [b]Among participants who discontinued B/F/TAF, last on-treatment HIV-1 viral load values were 180 copies/mL (Day 165), 55 copies/mL (Day 203), 57 copies/mL (Day 267), 131 copies/mL (Day 272), and 105 copies/mL (Day 309). All switched to another ART regimen. [c]Pulmonary insufficiency following trauma (n = 1); unknown (n = 1).
AE, adverse event; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; DRAE, drug-related adverse event; TE, treatment-experienced.

Quality-of-Life Outcomes

- An increase in median SF-36 Mental Component Summary score was observed at 24 months following the switch to B/F/TAF (*P* = 0.006[a])
- Median SF-36 Physical Component Summary scores remained stable

Change From Baseline to Month 24 in SF-36 Scores

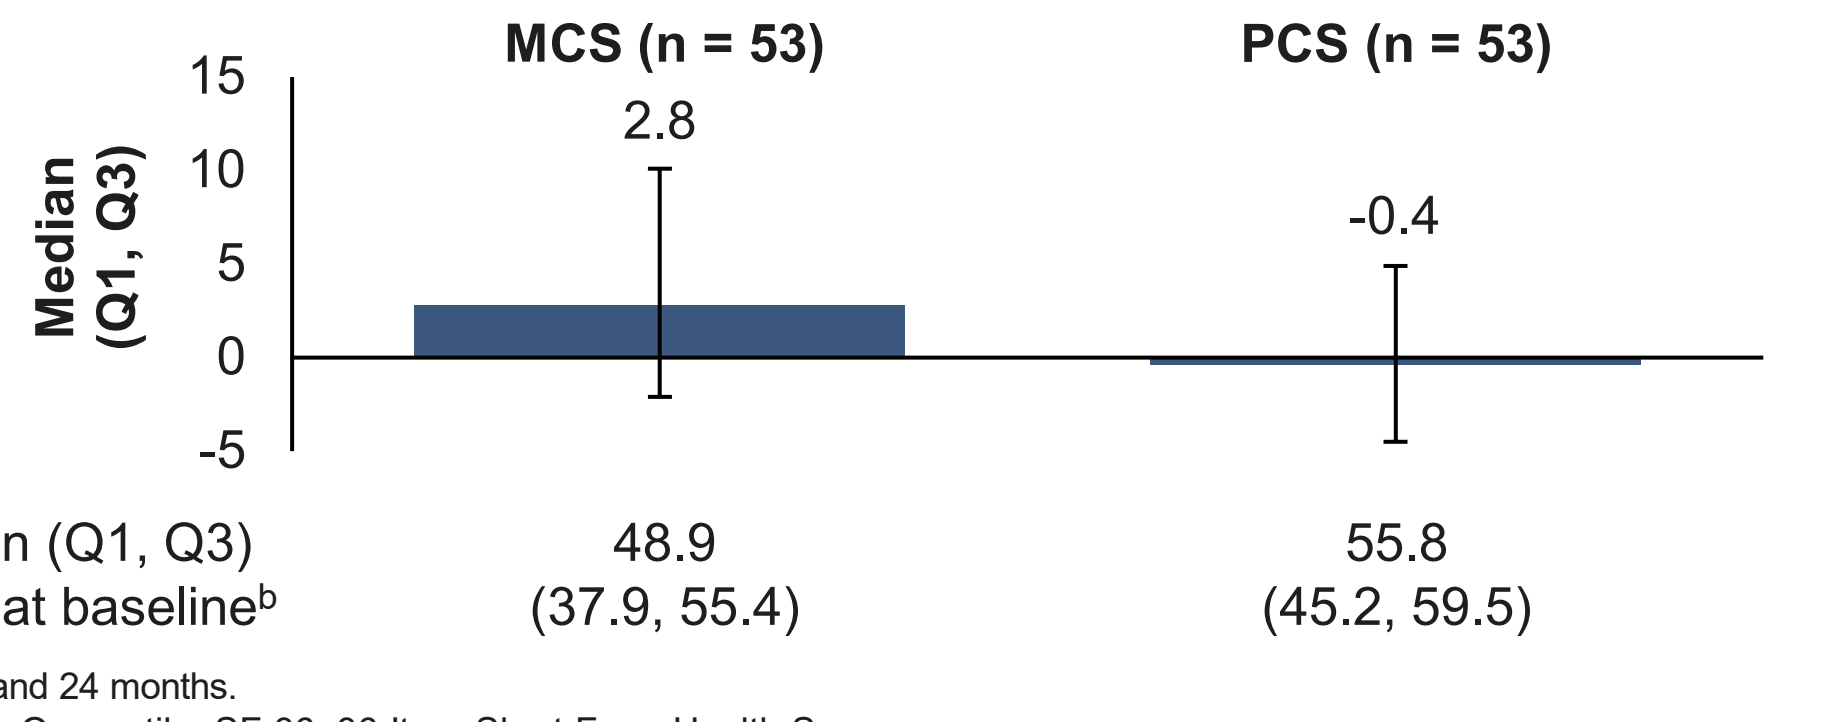

MCS (n = 53)
PCS (n = 53)
Median (Q1, Q3)
15
10
5
0
-5
2.8
-0.4
Median (Q1, Q3) score at baseline[b]
48.9 (37.9, 55.4)
55.8 (45.2, 59.5)

[a]Wilcoxon signed-rank test. [b]For participants with data available at baseline and 24 months.
MCS, Mental Component Summary; PCS, Physical Component Summary; Q, quartile; SF-36, 36-Item Short Form Health Survey.

HIV Treatment Satisfaction

- Median HIV Treatment Satisfaction Questionnaire (HIVTSQ) status[a] score was high at baseline and increased at 24 months
- Median (Q1, Q3) HIVTSQ change[b] score at 12 months was +26.0 (21.0, 29.0) (*P* < 0.001[c]), indicating an improvement in treatment satisfaction following a switch to B/F/TAF compared with the prior antiretroviral regimen

HIVTSQ Status Scores

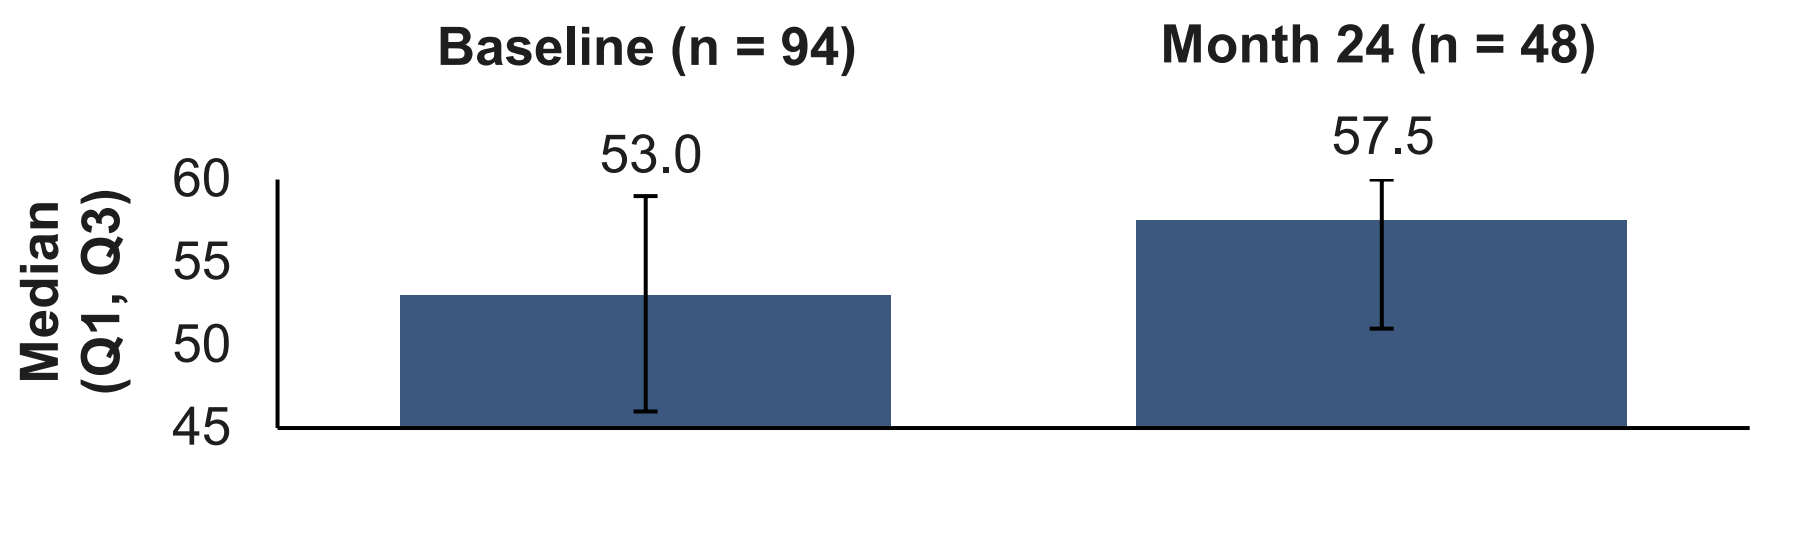

Baseline (n = 94)
Month 24 (n = 48)
Median (Q1, Q3)
60
55
50
45
53.0
57.5

[a]HIVTSQ status scores range from 0 to 60, with 60 representing highest treatment satisfaction. [b]HIVTSQ change scores range from -30 to +30: the higher the score, the greater the improvement in satisfaction with treatment compared with the previous regimen. [c]Sign test.
HIVTSQ, HIV Treatment Satisfaction Questionnaire; Q, quartile.

References: **1.** EACS. https://eacs.sanfordguide.com/eacs-part1/art/eacs-initial-regimens-arv-naive-adults (accessed April 10, 2026). **2.** Gandhi RT, et al. *JAMA*. 2023;329:63-84. **3.** DHHS. https://clinicalinfo.hiv.gov/sites/default/files/guidelines/documents/adult-adolescent-arv/guidelines-adult-adolescent-arv.pdf (accessed April 10, 2026). **4.** Gilead Sciences, Inc. Data on file. **5.** Trottier B, et al. *HIV Res Clin Pract*. 2025;26:2456890. **6.** Daar ES, et al. *Lancet HIV*. 2018;5:e347-56. **7.** Brar I, et al. *Medicine*. 2025;104:e41482. **8.** Gagliardini R, et al. *Int J Infect Dis*. 2025;158:107961.

Acknowledgments: We thank all study participants, investigators, and staff. These studies were funded by Gilead Sciences, Inc. Medical writing support was provided by Jack Seymour, BSc (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Disclosures: **BvW** reports grants or contracts from ViiV Healthcare (paid to institution); consulting fees from Gilead Sciences, Inc. and ViiV Healthcare; and payment or honoraria from MSD (paid to institution). **SM**, **DE**, **YT**, and **JYC** have no conflicts of interest to report. **MB** reports research grants from Gilead Sciences, Inc., Moderna, MSD, Sanofi, Valneva, ViiV Healthcare, and the Medical Research Council; payment or honoraria from AstraZeneca, Gilead Sciences, Inc., MSD, Pfizer, and ViiV Healthcare; and support for attending meetings and/or travel from Gilead Sciences, Inc. **AW** reports grants for clinical outreach work from ViiV Healthcare; and consulting and speaker fees from Gilead Sciences, Inc., Merck, and ViiV Healthcare.

Disclosures (cont.): **GM** reports support for attending meetings and/or travel and payment or honoraria from Gilead Sciences, Inc.; and has participated on data safety monitoring or advisory boards for Gilead Sciences, Inc. and ViiV Healthcare. **AI** reports consulting fees from Bayer and IQVIA; payment or honoraria from AbbVie and GSK; equipment, materials, drugs, medical writing, gifts, or other services from Cactus Communications; and has participated on data safety monitoring or advisory boards for Janssen, Merck, and Pfizer. **CS** reports grants or contracts from the Federal Ministry for Economic Cooperation and Development and the Society for Intercultural Collaboration; consulting fees from Gilead Sciences, Inc., Janssen, MSD, Shionogi, and ViiV Healthcare; and support for attending meetings from AbbVie, Gilead Sciences, Inc., and Janssen. **H-CT** has received speaker honoraria, and travel and registration support for attending AIDS 2026, from APAC. **CD** has received travel grants from Gilead Sciences, Inc., Merck, and ViiV Healthcare. **CYC** reports grants from Catalyst and NICID; payment or honoraria and speaker fees for the Multi-Speciality Medical Mega Conference 2024 and IDIC Theme Based Training; and serves as an unpaid board member for Action for AIDS. **DT**, **AM**, and **TC** are employees of, and hold stock in, Gilead Sciences, Inc. **KT** has received payment for presentations from Shionogi and ViiV Healthcare.

Correspondence: David Thorpe, david.thorpe@gilead.com.

AIDS 2026, the 26th International AIDS Conference; July 26–31, 2026; Rio de Janeiro, Brazil