Long-Term Safety and Efficacy of Elvitegravir/Cobicistat/ Emtricitabine/Tenofovir Alafenamide in Treatment-Naïve Adolescents and Virologically Suppressed Children With HIV-1

WEPEB114

NCT01854775

Natella Rakhmanina[1], Aditya Gaur[2], Pope Kosalaraksa[3], Elizabeth Hellström[4], Renate Strehlau[5], Suvaporn Anugulruengkitt[6], Afaaf Liberty[7], Matthew R Hendricks[8], Kathryn Kersey[8], Xiaopeng Liu[8], Brenda Okware[8], Vinicius Adriano Vieira[8], Eva Natukunda[9]

[1]Children's National Hospital, The George Washington University, Washington, DC, USA; [2]St. Jude Children's Research Hospital, Memphis, TN, USA; [3]Faculty of Medicine, Khon Kaen University, Khon Kaen, Thailand; [4]Be Part Yoluntu Centre, Paarl, South Africa; [5]Wits VIDA Nkanyezi Research Unit, University of the Witwatersrand, Johannesburg, South Africa; [6]Faculty of Medicine, Chulalongkorn University, Bangkok, Thailand; [7]Chris Hani Baragwanath Hospital, Johannesburg, South Africa. [8]Gilead Sciences, Inc., Foster City, CA, USA; [9]Joint Clinical Research Centre, Kampala, Uganda

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- In children with HIV-1 who were virologically suppressed and adolescents with HIV-1 who were treatment naïve, long-term treatment with elvitegravir/cobicistat/emtricitabine/tenofovir alafenamide (E/C/F/TAF) maintained viral suppression in over 90% of participants for up to 456 weeks (approximately 9 years) with no new safety findings
- Most adverse events (AEs) were Grade 1-2, with low rates of study drug–related discontinuations due to AEs
- There were no clinically significant changes in height, weight, and body mass index (BMI)
- Spine and total body less head (TBLH) bone mineral density (BMD) increased over time, while height-for-age–adjusted BMD Z-scores increased or remained stable, consistent with normal growth
- There was no clinically significant impact of E/C/F/TAF on renal function

Plain Language Summary

- Researchers studied how well and how safely a combination human immunodeficiency virus (HIV) medicine (elvitegravir/cobicistat/emtricitabine/tenofovir alafenamide) works in children and teenagers with HIV
- They followed three groups over several years:
 - — Teenagers aged 12 to 18 years who weighed 35 kg (about 77 lb) or more and were starting HIV treatment for the first time
 - — Children aged 6 to 12 years who weighed 25 kg (about 55 lb) or more and already had their HIV under control
 - — Children aged 2 years and older who weighed 14 to 25 kg (about 31-55 lb) and already had their HIV under control
- In all groups, more than 90% of participants kept the virus at very low levels in their blood over several years
- The medicine was safe and well tolerated in all age groups studied

Introduction

- E/C/F/TAF is a single-tablet regimen for the treatment of HIV-1[1]
- E/C/F/TAF contains the nucleoside/nucleotide reverse transcriptase inhibitor (NRTI) tenofovir alafenamide (TAF), which has improved renal and bone safety compared with tenofovir disoproxil fumarate (TDF)[2]
- Children with HIV-1 require lifelong treatment necessitating treatment options that are safe and efficacious over the long term[3]
- GS-US-292-0106 (NCT01854775) was a Phase 2/3, multicenter, open-label, multicohort study that evaluated E/C/F/TAF in adolescents with HIV-1 aged ≥ 12 years and weighing ≥ 35 kg and children with HIV-1 aged ≥ 2 to < 12 years and weighing ≥ 14 kg
 - — The primary and secondary pharmacokinetic, efficacy, and safety endpoints were previously reported[4-6]

Objective

- This analysis reports long-term safety and efficacy of E/C/F/TAF in adolescents who were treatment naïve and in children with virologically suppressed HIV-1 at baseline

Methods

Cohort 1 (TN)
- Age ≥ 12 to < 18 years
- Weight ≥ 35 kg
- Plasma HIV-1 RNA ≥ 1000 c/mL at screening
- No genotypic resistance to EVG, FTC, or TDF

Cohort 2 (VS)
- Age 6 to < 12 years
- Weight ≥ 25 kg
- Plasma HIV-1 RNA < 50 c/mL[a]
- No documented or suspected resistance to any component of E/C/F/TAF

Cohort 3 (VS)
- Age ≥ 2 years
- Weight ≥ 14 to < 25 kg
- Plasma HIV-1 RNA < 50 c/mL[a]
- No documented or suspected resistance to any component of E/C/F/TAF

Phase 2/3, open-label study (NCT01854775)

Cohorts 1 and 2: E/C/F/TAF 150/150/200/10 mg QD with food

Cohort 3: E/C/F/TAF 90/90/120/6 mg QD with food

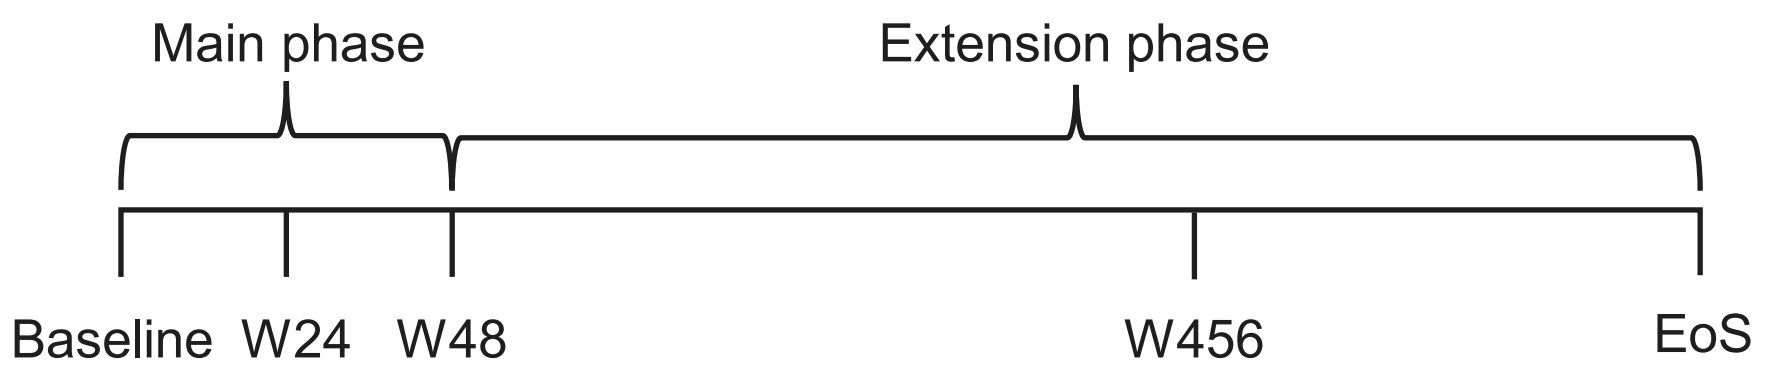

Main phase
Extension phase
Baseline
W24
W48
W456
EoS

Outcomes up to W456[b]
- HIV-1 RNA < 50 c/mL
- Change in CD4 count and percent
- Change in growth parameters
- Change in BMD

Outcomes through EoS[c]
- Resistance
- TEAEs
- Discontinuations

[a]Plasma HIV-1 RNA < 50 c/mL for ≥ 180 consecutive days before screening on a stable ART regimen. [b]Outcomes are reported for each cohort up to the week in which ≥ 50% of participants for each cohort were continuing follow-up in the optional open-label extension and had available data. [c]EoS was when the last participant data were collected (up to W600).
ART, antiretroviral therapy; BMD, bone mineral density; c, copies; C, cobicistat; CD4, cluster of differentiation 4; E/EVG, elvitegravir; EoS, end of study; F/FTC, emtricitabine; QD, once daily; TAF, tenofovir alafenamide; TDF, tenofovir disoproxil fumarate; TEAE, treatment-emergent adverse event; TN, treatment naïve; VS, virologically suppressed; W, Week.

Results

Demographics and Baseline Characteristics

	Cohort 1 ≥ 12 to < 18 years; ≥ 35 kg; TN n = 50	Cohort 2 6 to < 12 years; ≥ 25 kg; VS n = 52	Cohort 3 ≥ 2 years; ≥ 14 to < 25 kg; VS n = 27
Age, years, median (Q1, Q3)	15 (13, 17)	10 (9, 11)	6 (4, 8)
Weight, kg, median (Q1, Q3)	52.0 (41.0, 61.0)	30.9 (28.1, 33.7)	19.3 (17.0, 20.5)
Weight Z-score, median (Q1, Q3)	-0.31 (-1.08, 0.47)	-0.48 (-1.01, 0.14)	-0.88 (-1.72, -0.32)
Height Z-score, median (Q1, Q3)	-0.75 (-1.79, 0.08)	-0.73 (-1.26, 0.10)	-0.28 (-1.42, 0.23)
BMI Z-score, median (Q1, Q3)	0.17 (-0.56, 0.79)	-0.02 (-0.72, 0.42)	-1.24 (-1.79, -0.03)
Female sex assigned at birth, n (%)	28 (56.0)	30 (57.7)	17 (63.0)
Race, n (%)			
Black	44 (88.0)	37 (71.2)	24 (88.9)
Asian	6 (12.0)	13 (25.0)	3 (11.1)
White	0	2 (3.8)	0
CD4 count/μL, median (Q1, Q3)	456 (332, 574)	926 (795, 1129)	1,061 (895, 1315)
CD4 %, median (Q1, Q3)	23.0 (17.5, 30.0)	38.4 (33.7, 42.4)	37.4 (30.6, 40.3)
HIV-RNA < 50 c/mL, n (%)	0	52 (100)	27 (100)
Asymptomatic HIV disease, n (%)	42 (84.0)	48 (92.3)	26 (96.3)
Time since HIV diagnosis, years, median (Q1, Q3)	1.0 (0.0, 4.0)	8.0 (7.5, 10.0)	5.0 (4.0, 7.0)

BMI, body mass index; c, copies; CD4, cluster of differentiation 4; Q, quartile; TN, treatment naïve; VS, virologically suppressed.

Summary of Long-Term Efficacy

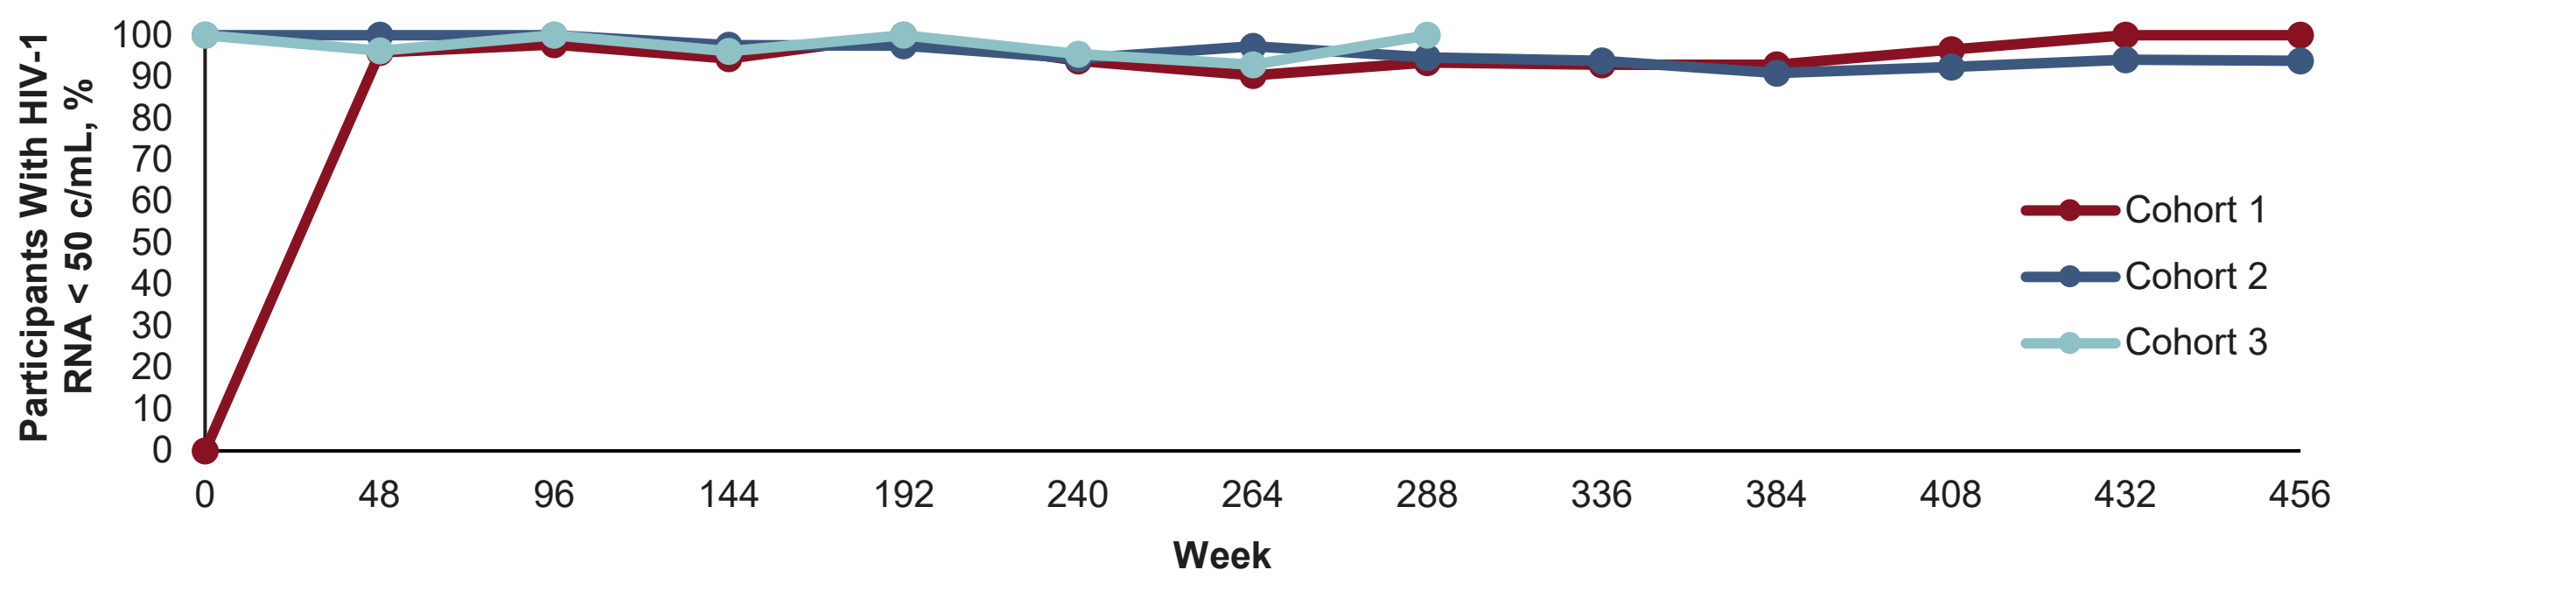

Participants With HIV-1 RNA < 50 c/mL, %
100
90
80
70
60
50
40
30
20
10
0
Cohort 1
Cohort 2
Cohort 3
0
48
96
144
192
240
264
288
336
384
408
432
456
Week

	0	48	96	144	192	240	264	288	336	384	408	432	456
Cohort 1 n =	50	48	46	36	32	32	31	31	28	28	29	25	25
Cohort 2 n =	52	52	46	41	40	36	37	37	33	33	26	17	16
Cohort 3 n =	27	27	18	26	24	22	14	1					

The cohorts were enrolled sequentially and therefore have variable follow up durations and different assessment timepoints. c, copies.

- Median (Q1, Q3) exposure was 446 (133, 565), 407 (214, 490), and 265 (263, 281) weeks in Cohorts 1, 2, and 3, respectively
- Median (Q1, Q3) adherence was 98.6 (95.6, 99.5), 98.0 (96.5, 98.8), and 98.2 (96.3, 99.2) % in Cohorts 1, 2, and 3, respectively

Median (Q1, Q3)	Cohort 1 ≥ 12 to < 18 years; ≥ 35 kg; Week 456[a] n = 25	Cohort 2 6 to < 12 years; ≥ 25 kg; Week 384[a] n = 30	Cohort 3 ≥ 2 years; ≥ 14 to < 25 kg; Week 264[a] n = 14
Change from baseline in CD4 count, cells/μL	309 (152, 398)	-294 (-517, -42)	-302 (-531, -67)
Change from baseline in CD4%	12.8 (7.9, 15.4)	1.0 (-4.9, 4.4)	-1.1 (-5.1, 0.5)

[a]Visits were included up to last timepoint where ≥ 50% of participants for each cohort were continuing follow-up in the optional open-label extension and had available data. Q, quartile.

- Resistance developed in one participant in Cohort 1 at Week 552 with NRTI resistance mutations K65R and M184V and integrase strand-transfer inhibitor (INSTI) resistance mutation E92Q
 - — Participant resuppressed HIV-1 RNA to < 50 copies/mL by their last study visit at Week 576 while continuing E/C/F/TAF
- Resistance was identified in one participant in Cohort 2 at Week 192 with the NRTI resistance mutation M184V
 - — Participant did not suppress HIV-1 RNA to < 50 copies/mL by their last study visit and no additional resistance mutations to study drugs other than M184V were identified in the corresponding sample

Safety

n (%)	Cohort 1 ≥ 12 to < 18 years; ≥ 35 kg; TN (n = 50)	Cohort 2 6 to < 12 years; ≥ 25 kg; VS (n = 52)	Cohort 3 ≥ 2 years; ≥ 14 to < 25 kg; VS (n = 27)	Total (N = 129)
Any TEAE	48 (96.0)	48 (92.3)	25 (92.6)	121 (93.8)
Study drug–related TEAE	22 (44.0)	14 (26.9)	4 (14.8)	40 (31.0)
Any Grade 3 or 4 TEAE	8 (16.0)	4 (7.7)	0	12 (9.3)
Study drug–related Grade 3 or 4 TEAE	1[a] (2.0)	0	0	1 (0.8)
Any serious TEAE	10 (20.0)	7 (13.5)	1 (3.7)	18 (14.0)
Study drug–related serious TEAE	1[b] (2.0)	0	0	1 (0.8)
TEAE leading to study drug discontinuation	2 (4.0)	1 (1.9)	0	3[c] (2.3)
TEAE leading to death	1[d] (2.0)	0	0	1 (0.8)

[a]Grade 3 TEAE of chorioretinitis related to study drug reported in one participant. [b]Visual impairment and uveitis in one participant were reported as serious TEAEs related to study drug. [c]Three participants discontinued study drug due to adverse events unrelated to study drug. [d]The TEAE leading to death was due to a road traffic accident unrelated to study drug. TEAE, treatment-emergent adverse event; TN, treatment naïve; VS, virologically suppressed.

- The most common study drug–related TEAEs were vomiting (11.6%), nausea (8.5%), and abdominal pain (7.8%)

Change in Growth Parameters

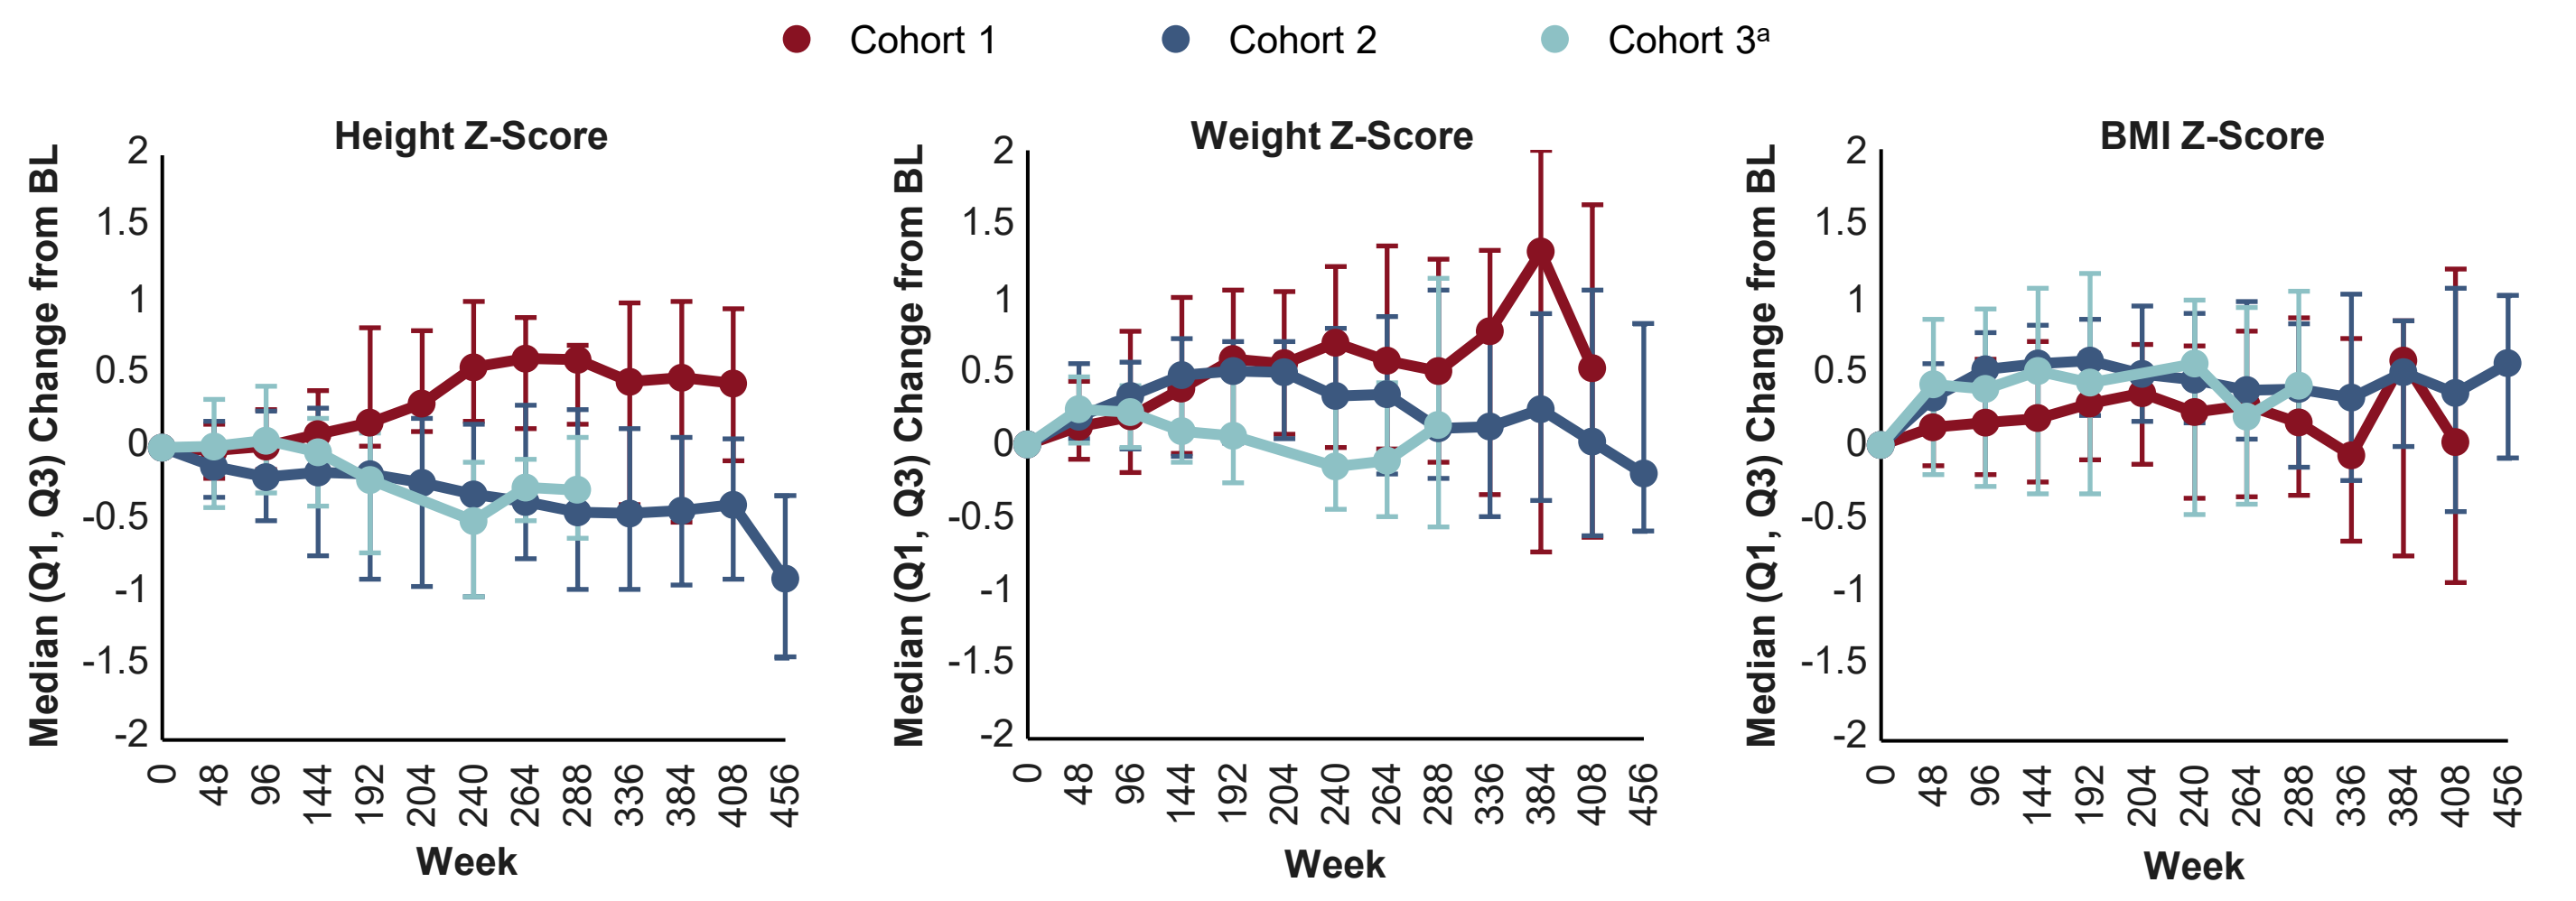

Cohort 1
Cohort 2
Cohort 3[a]
Height Z-Score
Weight Z-Score
BMI Z-Score
Median (Q1, Q3) Change from BL
Week
0 48 96 144 192 204 240 264 288 336 384 408 456

Height Z-Score:
Cohort 1 n = 50 48 46 33 28 25 21 20 17 10 7 4
Cohort 2 n = 52 51 46 41 40 39 16 9 37 34 33 30 13
Cohort 3 n = 27 27 27 26 24 22 21 10

Weight Z-Score:
Cohort 1 n = 50 48 46 33 28 25 21 20 17 10 7 4
Cohort 2 n = 52 51 46 41 40 39 36 37 37 34 33 30 13
Cohort 3 n = 27 27 27 26 24 22 21 10

BMI Z-Score:
Cohort 1 n = 50 48 46 33 28 25 21 20 17 10 7 4
Cohort 2 n = 52 51 46 41 40 39 36 37 37 34 33 30 13
Cohort 3 n = 27 27 27 26 24 22 21 10

Z-scores generated using the 2000 US CDC growth charts. [a]There are no data for Cohort 3 at Week 204.
BL, baseline; BMI, body mass index; CDC, Centers for Disease Control and Prevention; Q, quartile.

- Changes from baseline in height, weight, and BMI Z-scores were not clinically significant

Change in Bone Mineral Density

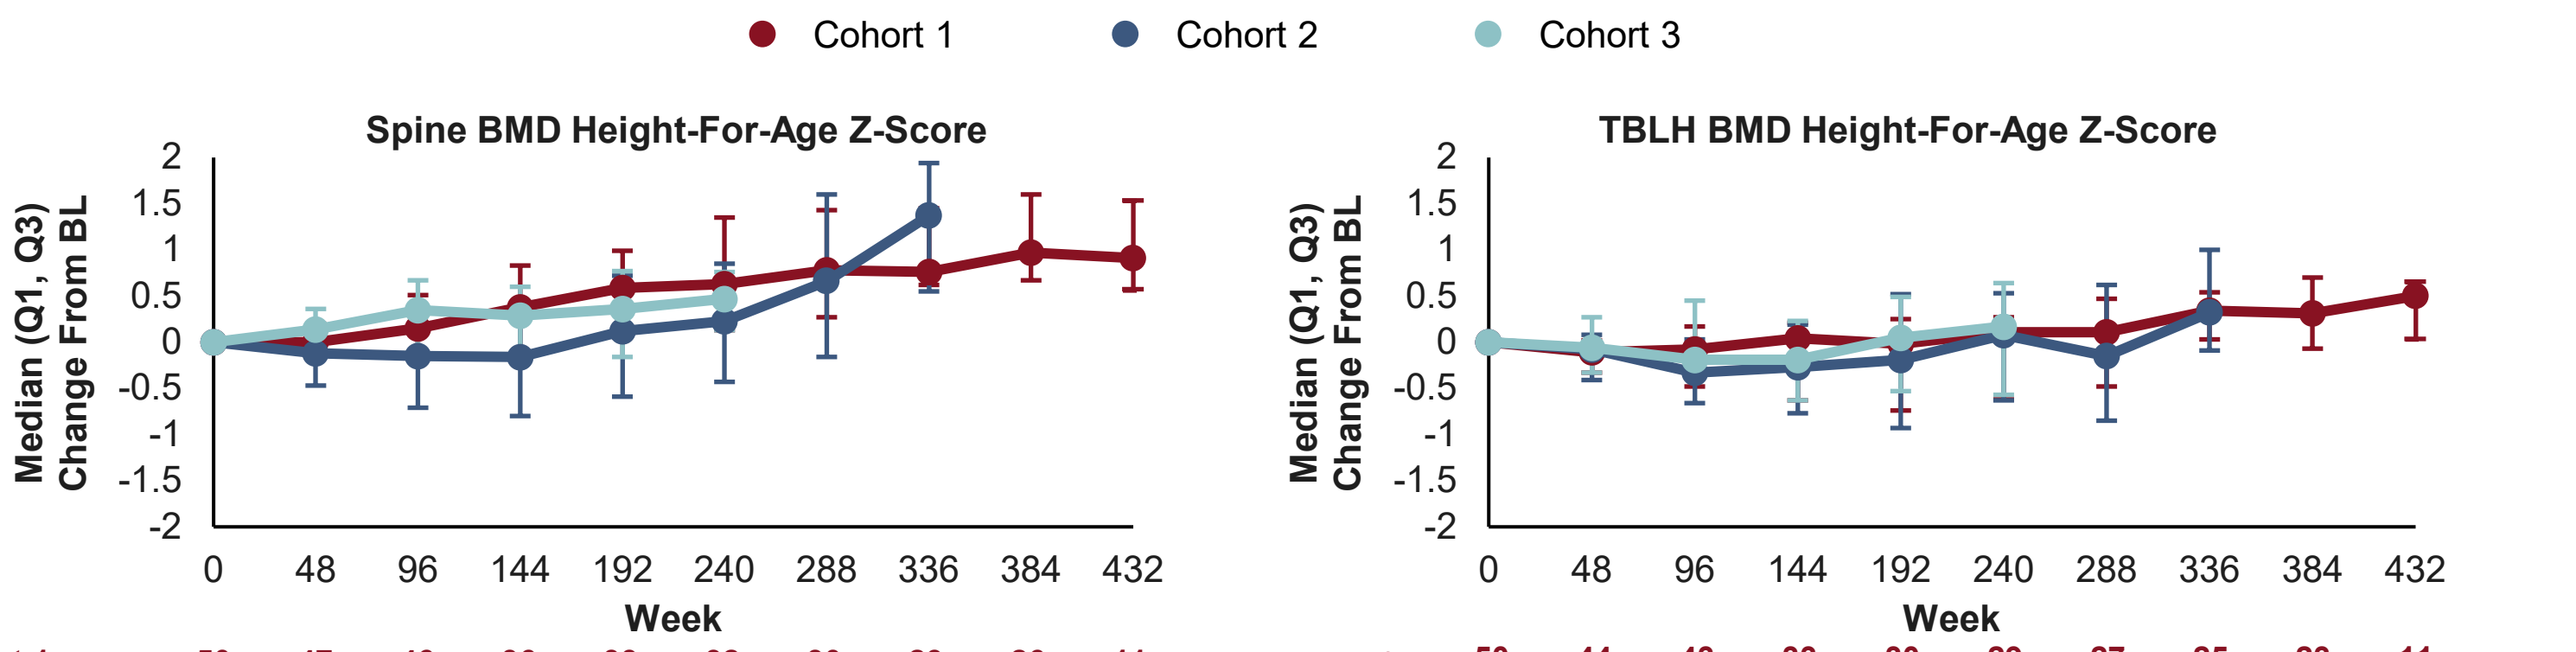

Cohort 1
Cohort 2
Cohort 3
Spine BMD Height-For-Age Z-Score
TBLH BMD Height-For-Age Z-Score
Median (Q1, Q3) Change From BL
Week
0 48 96 144 192 240 288 336 384 432

Spine BMD	0	48	96	144	192	240	288	336	384	432
Cohort 1 n =	50	47	46	36	33	32	30	29	26	11
Cohort 2 n =	52	49	43	40	37	33	16	9		
Cohort 3 n =	27	27	25	25	23	22				

TBLH BMD	0	48	96	144	192	240	288	336	384	432
n =	50	44	43	33	30	29	27	25	23	11
n =	52	51	45	44	39	36	17	10		
n =	27	27	25	24	23	22				

Spine BMD height-for-age Z-scores and TBLH BMD height-for-age Z-scores are extrapolated. Some participants had missing height-for-age Z-scores because their heights were outside the median height in the CDC growth chart, or the height-for-ages were outside the BMD reference data for Z-scores.
BL, baseline; BMD, bone mineral density; CDC, Centers for Disease Control and Prevention; Q, quartile; TBLH, total body less head.

- Height-for-age–adjusted BMD Z-scores increased or remained stable

Median (Q1, Q3)	Cohort 1 ≥ 12 to < 18 years; ≥ 35 kg			Cohort 2 6 to < 12 years; ≥ 25 kg			Cohort 3 ≥ 2 years; ≥ 14 to < 25 kg		
	Baseline	Week 336[a]	Percent Change From BL	Baseline	Week 240[a]	Percent Change From BL	Baseline	Week 240[a]	Percent Change From BL
Spine BMD, g/cm²	0.78 (0.68, 0.94) n = 47	0.96 (0.88, 0.99) n = 29	36 (23, 47) n = 29	0.62 (0.54, 0.67) n = 50	0.87 (0.71, 0.94) n = 33	35 (27, 46) n = 33	0.46 (0.40, 0.48) n = 27	0.57 (0.48, 0.70) n = 22	28 (21, 33) n = 22
TBLH BMD, g/cm²	0.88 (0.82, 0.96) n = 45	0.94 (0.89, 1.00) n = 25	11 (7, 21) n = 25	0.67 (0.64, 0.71) n = 52	0.83 (0.79, 0.89) n = 36	21 (17, 30) n = 36	0.51 (0.45, 0.57) n = 27	0.69 (0.63, 0.72) n = 22	36 (31, 40) n = 22

[a]Visits were included up to last timepoint where ≥ 50% of participants for each cohort were continuing follow-up in the optional open-label extension and had available data. BL, baseline; BMD, bone mineral density; Q, quartile; TBLH, total body less head.

- Spine and TBLH BMD increased over time

Change in eGFR From Baseline

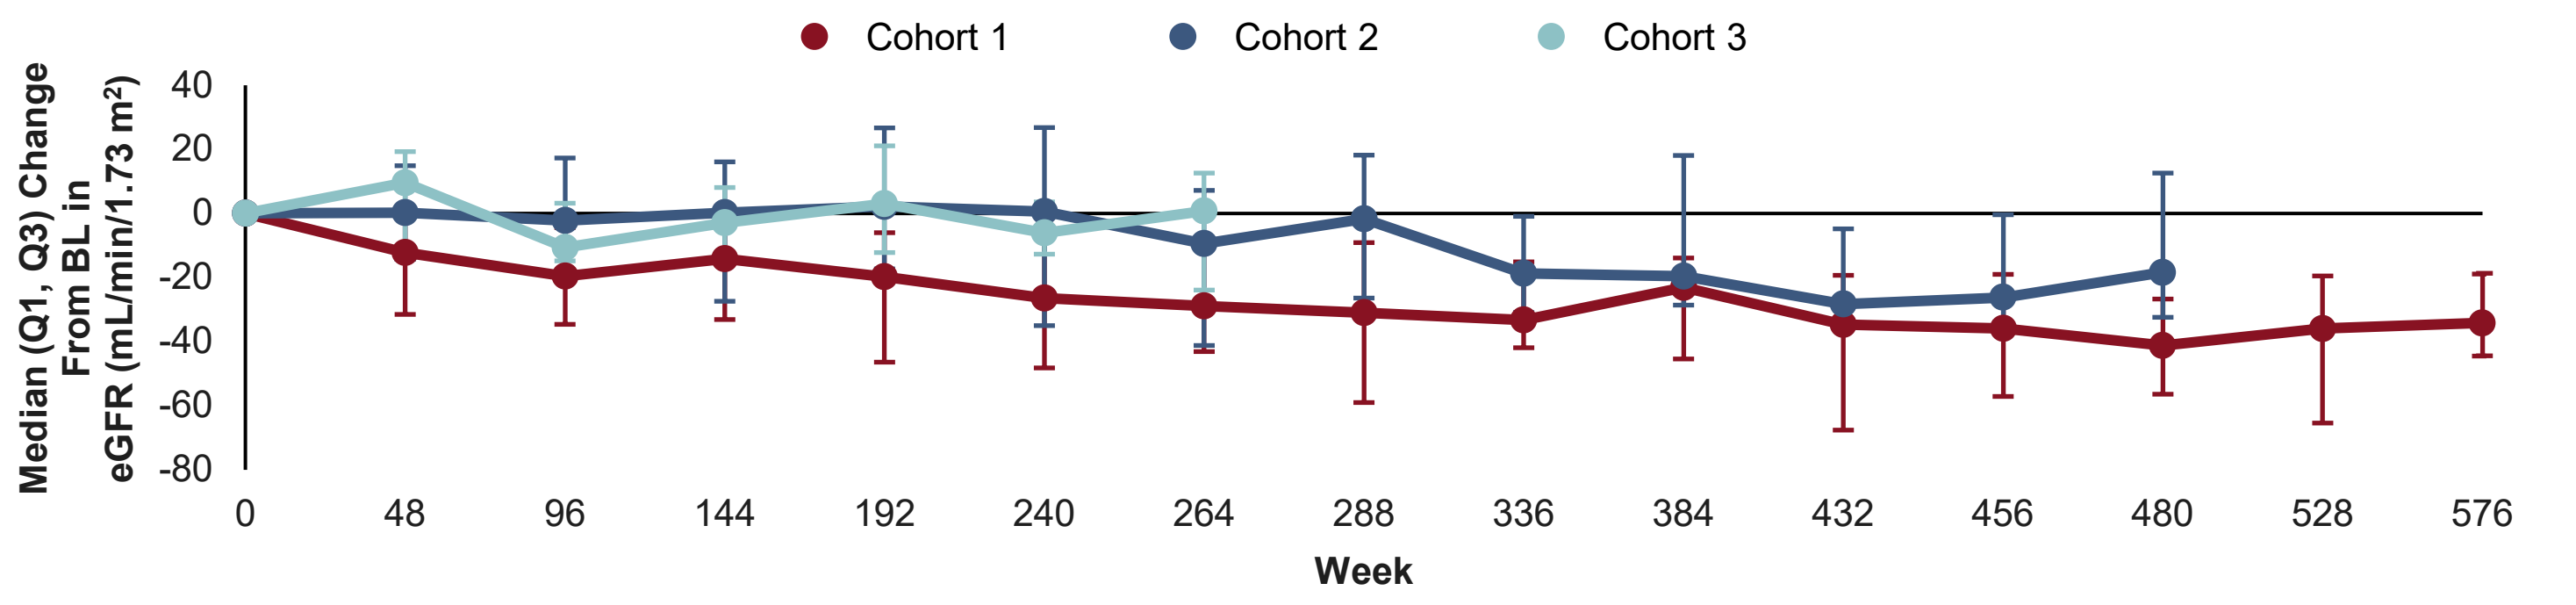

Cohort 1
Cohort 2
Cohort 3
Median (Q1, Q3) Change From BL in eGFR (mL/min/1.73 m²)
40
20
0
-20
-40
-60
-80
Week

	0	48	96	144	192	240	264	288	336	384	432	456	480	528	576
Cohort 1 n =	50	48	46	35	33	32	31	31	17	29	25	25	18	21	7
Cohort 2 n =	52	52	46	37	40	23	37	37	28	32	16	16	14		
Cohort 3 n =	27	27	17	26	23	21	14								

eGFR by Schwartz formula. eGFR, estimated glomerular filtration rate; Q, quartile.

- Initial decreases in eGFR may be the result of cobicistat inhibiting tubular secretion of creatinine, which is not clinically significant[7]

Absolute eGFR Over Time

Median (Q1, Q3)	Cohort 1 ≥ 12 to < 18 years; ≥ 35 kg			Cohort 2 6 to < 12 years; ≥ 25 kg			Cohort 3 ≥ 2 years; ≥ 14 to < 25 kg		
	Baseline	Week 48	Week 456[a]	Baseline	Week 48	Week 384[a]	Baseline	Week 48	Week 264[a]
eGFR[b], mL/min/1.73 m²	156 (129, 185) n = 50	146 (119, 166) n = 48	139 (123, 142) n = 25	150 (138, 165) n = 52	155 (134, 166) n = 51	144 (123, 158) n = 32	147 (139, 159) n = 27	162 (142, 174) n = 27	144 (121, 160) n = 14

[a]Visits were included up to last timepoint where ≥ 50% of participants for each cohort were continuing follow-up in the optional open-label extension and had available data. [b]eGFR by Schwartz formula. eGFR, estimated glomerular filtration rate; Q, quartile.

- eGFR remained within age-appropriate ranges, with no cases of proximal tubulopathy observed

References: **1.** Genvoya. Package Insert. Gilead Sciences, Inc. 2022. **2.** DeJesus E, et al. *AIDS Res Hum Retroviruses*. 2018;34:337-42. **3.** Department of Health and Human Services. https://clinicalinfo.hiv.gov/en/guidelines/pediatric-arv/ (accessed April 30, 2026). **4.** Gaur AH, et al. *Lancet HIV*. 2016;3:e561-8. **5.** Natukunda E, et al. *Lancet Child Adolesc Health*. 2017;1:27-34. **6.** Natukunda E, et al. *J Int AIDS Soc*. 2025;28:e26414. **7.** Pozniak A, et al. *J Acquir Immune Defic Syndr*. 2016;71:530-37.

Acknowledgments: This study was sponsored by Gilead Sciences, Inc. We thank all study participants, their families, and all participating study investigators and staff. Medical writing support was provided by Jessica Adams, PharmD, AAHIVE (Gilead Sciences, Inc.). Production assistance was provided by Aspire Scientific Ltd (UK) and was funded by Gilead Sciences, Inc. Funding for medical writing support was provided by Gilead Sciences.

Disclosures: **NR** reports research funding paid to their institution by Gilead Sciences, Inc., GSK, MSD, and Penta; and consulting fees for participating in an advisory board for ViiV Healthcare. **AG** reports research grants from Gilead Sciences, Inc., Merck, and ViiV Healthcare; and participation on an advisory board for ViiV Healthcare. **PK**, **EH**, **SA**, and **EN** have no conflicts of interest to report. **RS** reports research funding paid to their institution by Gilead Sciences, Inc., GSK, MSD, and Penta. **AL** reports trial funding from GSK and MSD. **MRH**, **KK**, **XL**, **BO**, and **VAV** are employees of, and hold stocks in, Gilead Sciences, Inc.

Correspondence: Natella Rakhmanina, nrakhman@childrensnational.org.

AIDS 2026, the 26th International AIDS Conference; July 26–31, 2026; Rio de Janeiro, Brazil