Reaching the Hard-to-Reach via the ACCELERATE Model of HIV Care: Telehealth, Rapid Restart and Follow-up

Hilal Abdessamad, MD[1], Thomas T. Deem, MSN, FNP-BC, AAHIVS[2], Blair A. Thedinger[3], MD, Wassim Jamaleddine[1], MD, Anis Buttar-Miller[1], Ransome Drexler[1], Dima Dandachi, MD, MPH[1]

[1]University of Missouri – Columbia, Columbia, MO, USA, [2]Gilead Sciences, Inc, Foster City, CA, USA, [3]KC Care Health Center, Kansas City, MO, USA

Background

- Patient retention in HIV care is key for Ending the HIV Epidemic (EHE).
- Missouri (MO) is a Phase I jurisdiction due to high rural HIV transmission.
- ACCELERATE is an on-going, mixed-methods implementation study across MO that assesses the acceptability and effectiveness of an intervention to identify and rapidly relink people with HIV (PWH) who are out of care (OOC) to health services.

Methodology

- We identified and engaged adult OOC PWH in 4 clinics across MO through a data to care analysis and enhanced contact algorithm. (Figure 1).
- OOC was defined as a lack of medical visit in the last 6 months and off antiretroviral therapy (ART) for at least 1 month.
- Prior to an in-person visit, individuals participated in a telehealth (TLH) provider appointment.
- After TLH they had a rapid start of a 30-day supply of study-provided Bictegravir/Emtricitabine/Tenofovir Alafenamide (BIC/FTC/TAF) without obtaining baseline labs.
- Select surveys were completed at baseline and after visits at 1 month.

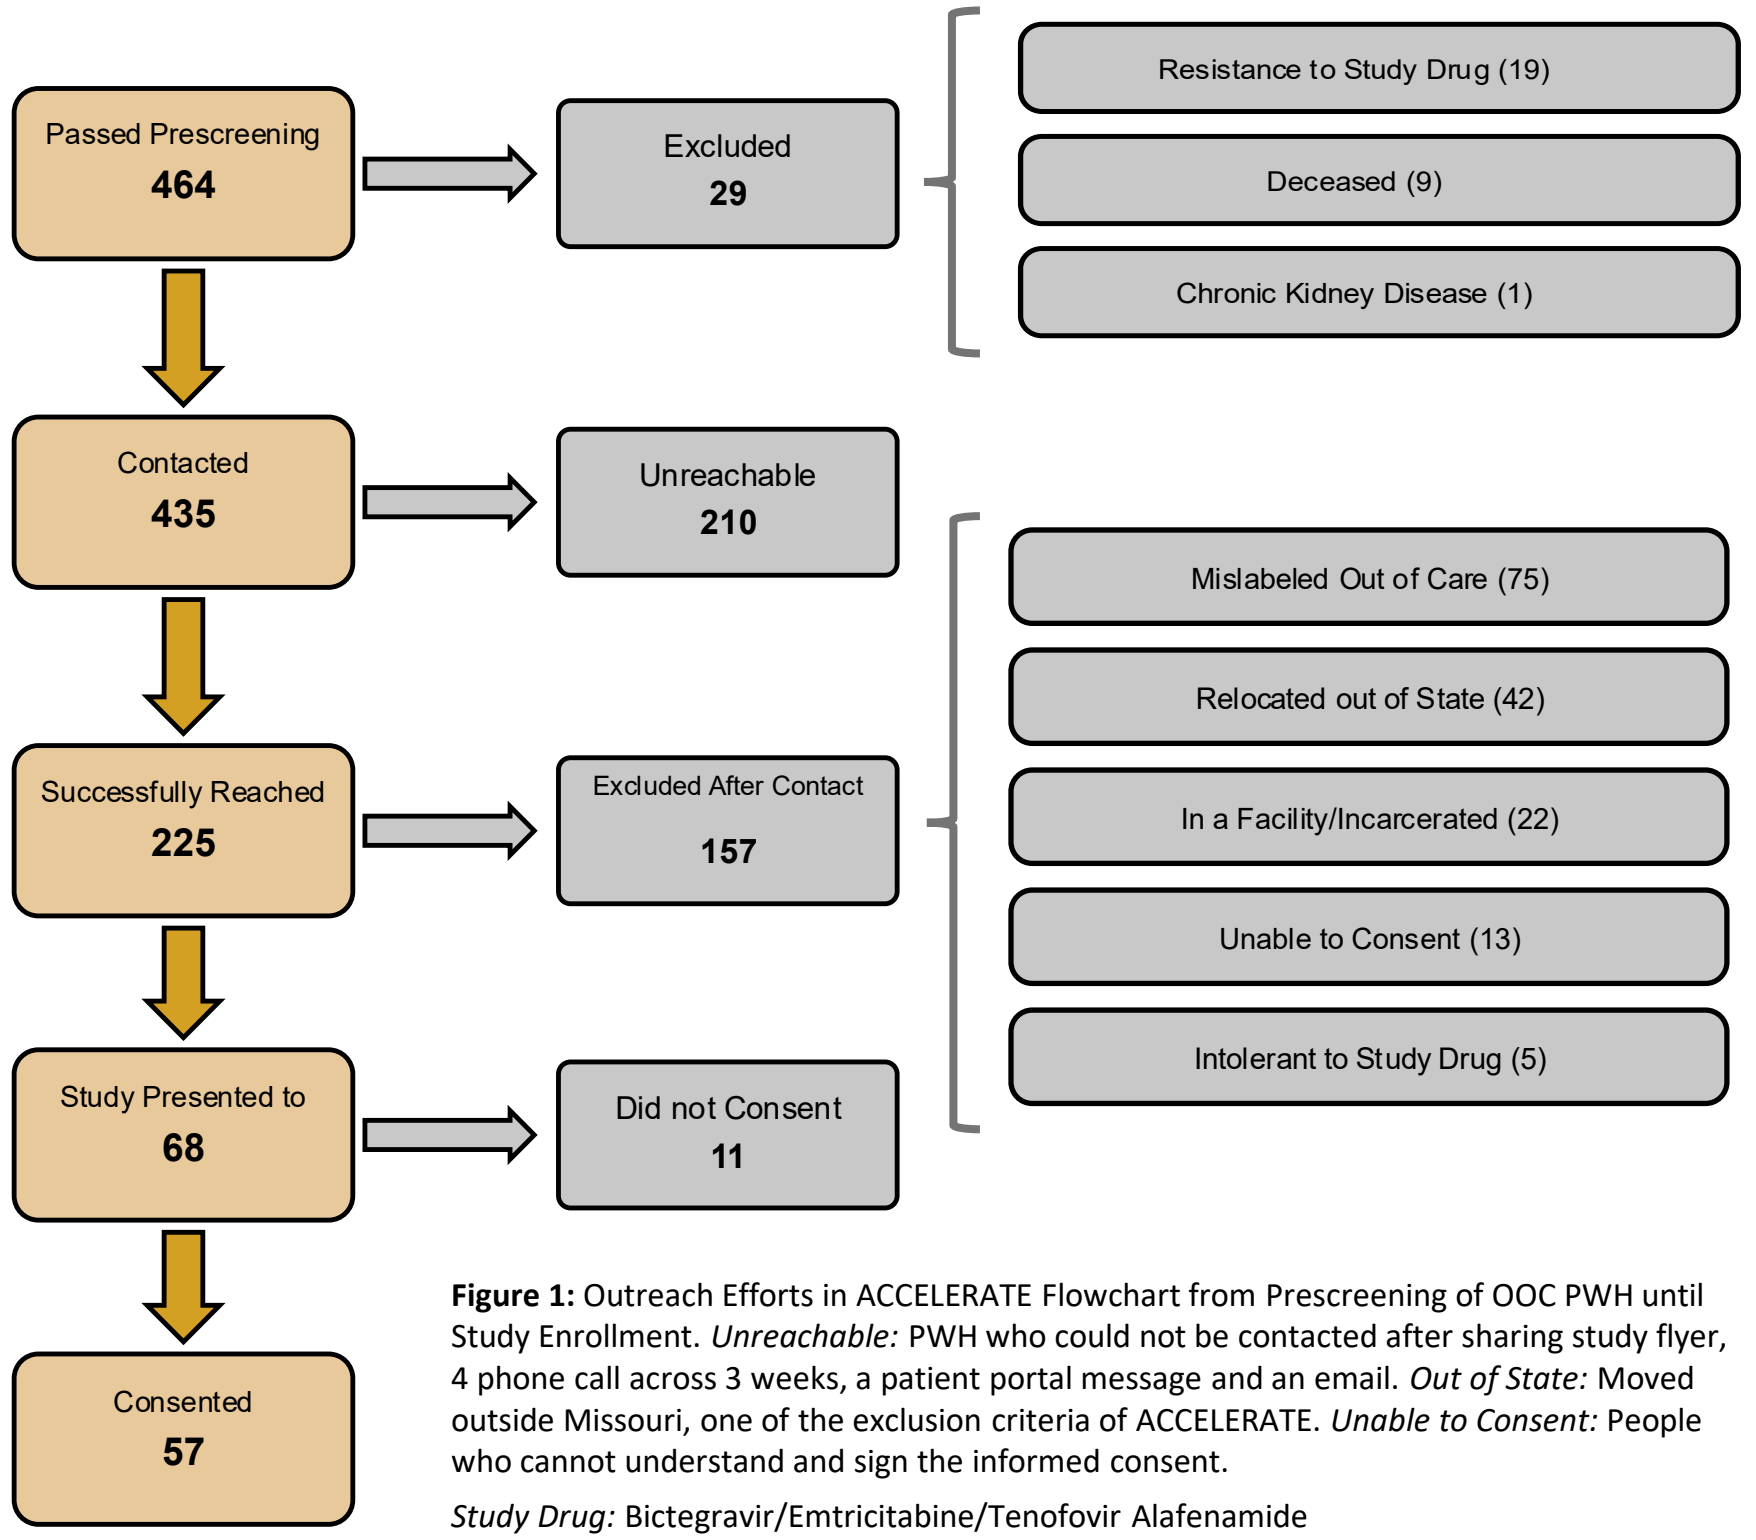
Passed Prescreening
464
Excluded
29
Resistance to Study Drug (19)
Deceased (9)
Chronic Kidney Disease (1)
Contacted
435
Unreachable
210
Successfully Reached
225
Excluded After Contact
157
Mislabeled Out of Care (75)
Relocated out of State (42)
In a Facility/Incarcerated (22)
Unable to Consent (13)
Intolerant to Study Drug (5)
Study Presented to
68
Did not Consent
11
Consented
57

Figure 1: Outreach Efforts in ACCELERATE Flowchart from Prescreening of OOC PWH until Study Enrollment. *Unreachable:* PWH who could not be contacted after sharing study flyer, 4 phone call across 3 weeks, a patient portal message and an email. *Out of State:* Moved outside Missouri, one of the exclusion criteria of ACCELERATE. *Unable to Consent:* People who cannot understand and sign the informed consent.

Study Drug: Bictegravir/Emtricitabine/Tenofovir Alafenamide

Results

- Phone calls were the most successful contact method.
- We reached 225/464 (49%) PWH, 150/225 (63%) confirmed OOC, 68/150 (45%) were eligible, and 57/68 (84%) were enrolled.
- On average, participants have been off ART for 15 months (SD 22).
- Mean age of 42 (SD 11), 80% male sex at birth and 36% Black.
- At baseline:
 - 52% reported substance use
 - 48% had an Internalized AIDS Stigma Scale score of 5+
 - 62% screened positive for anxiety with the Generalized Anxiety Disorder-2 survey
 - 60% screened positive for depression with the Patient Health Questionnaire-2 survey.
 - Among barriers to care, mental health causes ranked highest, followed by financial concerns. (Figure 2).
- Using the Customer Assessment of Health Care Providers and Systems survey, 33/43 (77%) of PWH rated their TLH visit of 8 or above (scale 1 - 10).
- Of PWH who have reached 1-month follow-up since enrollment (n=41):
 - 80% attended their clinic visit
 - 30/31 (97%) reported missing less than 5 doses in 30 days
 - Most reported satisfaction with their ART using the HIV Treatment Satisfaction Questionnaire. (HIVTSQ, Table 1).

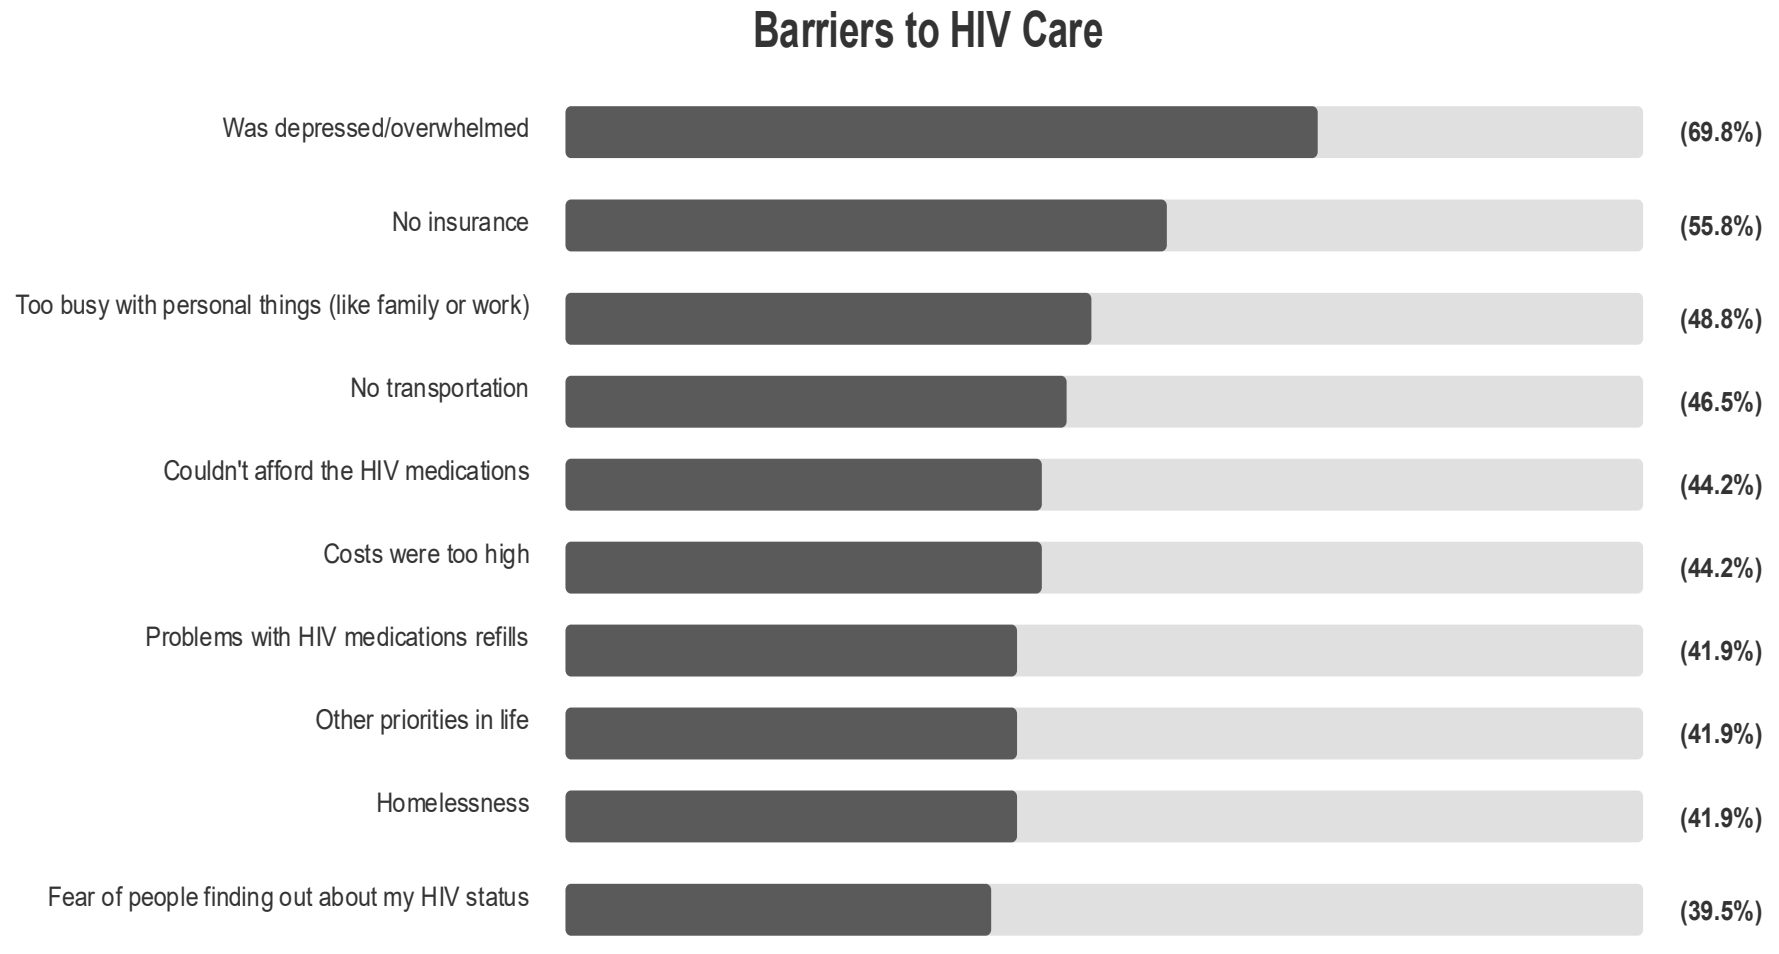
Barriers to HIV Care
Was depressed/overwhelmed (69.8%)
No insurance (55.8%)
Too busy with personal things (like family or work) (48.8%)
No transportation (46.5%)
Couldn't afford the HIV medications (44.2%)
Costs were too high (44.2%)
Problems with HIV medications refills (41.9%)
Other priorities in life (41.9%)
Homelessness (41.9%)
Fear of people finding out about my HIV status (39.5%)

Figure 2: Top 10 barriers to HIV care, selected by PWH who were OOC (%, N=43). As part of the baseline survey, respondents chose which barriers were important reasons to fall out of care. For each barrier, the percentage of respondents who identified it as "important" was calculated.

HIV Treatment Satisfaction Questionnaire	Mean (SD) (N=31)
How satisfied are you with your current treatment?	5.64 (0.66)
How well controlled do you feel your HIV has been recently?	5.22 (1.14)
How satisfied are you with any side effects of your present treatment?	5.32 (1.24)
How satisfied are you with the demands made by your current treatment?	5.71 (0.59)
How convenient have you been finding your treatment to be recently?	5.65 (0.71)
How flexible have you been finding your treatment to be recently?	5.58 (0.76)
How satisfied are you with your understanding of your HIV?	5.81 (0.4)
How satisfied are you with the extent to which the treatment fits in with your lifestyle?	5.64 (0.61)
Would you recommend your present treatment to someone else who is being offered this HIV treatment?	5.71 (0.7)
How satisfied would you be to continue with your present form of treatment?	5.65 (0.66)

Table 1: Ten-item HIV Treatment Satisfaction Questionnaire (HIVTSQ).

Findings at 1 Month.

The responses to the HIVTSQ questionnaire are scaled from 1 to 6, where 1 represents the least favorable response and 6 represents the most favorable option.

Conclusion

- Despite the high prevalence of financial difficulties, mental health challenges and substance use among participants, ACCELERATE was acceptable overall to OOC PWH.
- This integrated, rapid-start telehealth intervention shows promise as a re-engagement strategy for OOC PWH that ACCELERATEs progress towards achieving the targets of the EHE initiative in the US.

Notes

- Baseline data included demographics, substance use, barriers to care, Internalized AIDS Stigma Scale, the Customer Assessment of Health Care Providers and Systems, Generalized Anxiety Disorder-and Patient Health Questionnaire-2 surveys.
- Month 1 data included clinic visit attendance, medication compliance and HIVTSQ questionnaire.

Health Care