Real-World Sexually Transmitted Infection Testing Patterns Among Individuals Using Pre-Exposure Prophylaxis for HIV-1 Prevention in the United States

Xiwen Huang*, Juan Yang, Edward Gemson, Wenyi Wang, Li Tao, Dylan Mezzio, Chris Nguyen, Sandra I. McCoy, Joshua Gruber

Gilead Sciences, Inc., Foster City, CA, USA

P-287

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- This real-world observational study demonstrated that the frequency of testing for sexually transmitted infections (STI) increased in people who were using HIV-1 pre-exposure prophylaxis (PrEP) compared with before they were using PrEP
- Despite more frequent testing post-PrEP initiation, only one-third of PrEP users tested for STIs every ≤6 months, the minimum frequency recommended by the Centers for Disease Control and Prevention (CDC)
- Overall, testing rates were lowest in the South and among those aged ≥45 years, and highest in those using Medicaid
 - Unexpectedly, testing rates were similar among males and females, although it is unclear whether women undergo asymptomatic screening more often than the recommended minimum frequency or are testing more due to symptomatic STIs
- STI prevalence increased slightly after PrEP initiation, which may be partially due to the increased STI testing detecting asymptomatic infections that would otherwise go undiagnosed or self-resolve
- Additional work is needed to determine the optimal STI testing frequency in the era of long-acting PrEP and to support people using PrEP in receiving appropriate testing

Plain Language Summary

- HIV prevention medications, also called “PrEP”, lower people’s chances of getting HIV when taken as directed
- However, PrEP does not protect against STIs, such as chlamydia
- The Centers for Disease Control and Prevention recommends that people using PrEP should test for STIs at least every 6 months
- This study looked at how often people were testing for STIs before and after starting PrEP
- The study showed that people tested for STIs more often when using PrEP compared with before using PrEP
 - However, only three out of every 10 people using PrEP tested for STIs at least every 6 months, as recommended
- More people had STIs after starting PrEP compared with before starting PrEP; however, this was probably due to testing for STIs more often when using PrEP
- More work is needed to support people using PrEP to get regular STI tests, especially as new types of PrEP become available that need to be given less often

Background

- The CDC generally recommends routine STI testing every 3–6 months for people taking HIV-1 PrEP, as part of comprehensive HIV-1/STI prevention services[1,2]
- The CDC recommends the following:[1,2]
 - Before initiating PrEP, all sexually active adults are recommended to be screened for chlamydia, gonorrhea, and syphilis
 - At least every 3 months, sexually active individuals with signs or symptoms of an STI should be tested, and asymptomatic men who have sex with men (MSM) with increased likelihood of recurrent STIs should be screened
 - At least every 6 months, all sexually active individuals should be screened; PrEP users should be tested for syphilis and gonorrhea, and MSM and transgender women should be tested for chlamydia, even if asymptomatic
 - At least every 12 months, heterosexually active individuals should be screened for chlamydia, even if asymptomatic
- Despite these recommendations and the increase in PrEP uptake,[3,4] the incidence of STIs remains high and disparities persist in the US[5]
- However, there is a lack of real-world evidence on STI testing rates among individuals using PrEP in the US

Objective

- To describe real-world patterns of STI testing among individuals using PrEP in the US

Methods

- Naïve PrEP users aged ≥18 years with an initial PrEP prescription between October 2020 and December 2023 were identified using the HealthVerity database
 - PrEP regimens included daily oral emtricitabine/tenofovir disoproxil fumarate or emtricitabine/tenofovir alafenamide, or intramuscular injections of cabotegravir every 2 months
 - Eligible individuals were required to have ≥12 months of continuous observation pre- and post-PrEP initiation, ≥9 months of cumulative PrEP use, and no evidence of HIV-1 infection within 12 months post-PrEP initiation
- Chlamydia, gonorrhea, and syphilis testing frequencies and diagnoses were assessed over the 12-month periods preceding and following PrEP initiation
 - Testing frequency was calculated as the average interval between tests, categorized as every ≤3, >3 to 6, >6 to 9, and >9 months
 - Presence of an STI was defined as an STI diagnosis or positive laboratory result within each 12-month observation window

Results

Study Population Disposition

- A total of 42,408 PrEP users were included in the analysis (**Figure 1**); most PrEP users were male (94%) and mean (SD) age was 36.9 (11.8) years (**Table 1**)

Figure 1. Study Population Disposition

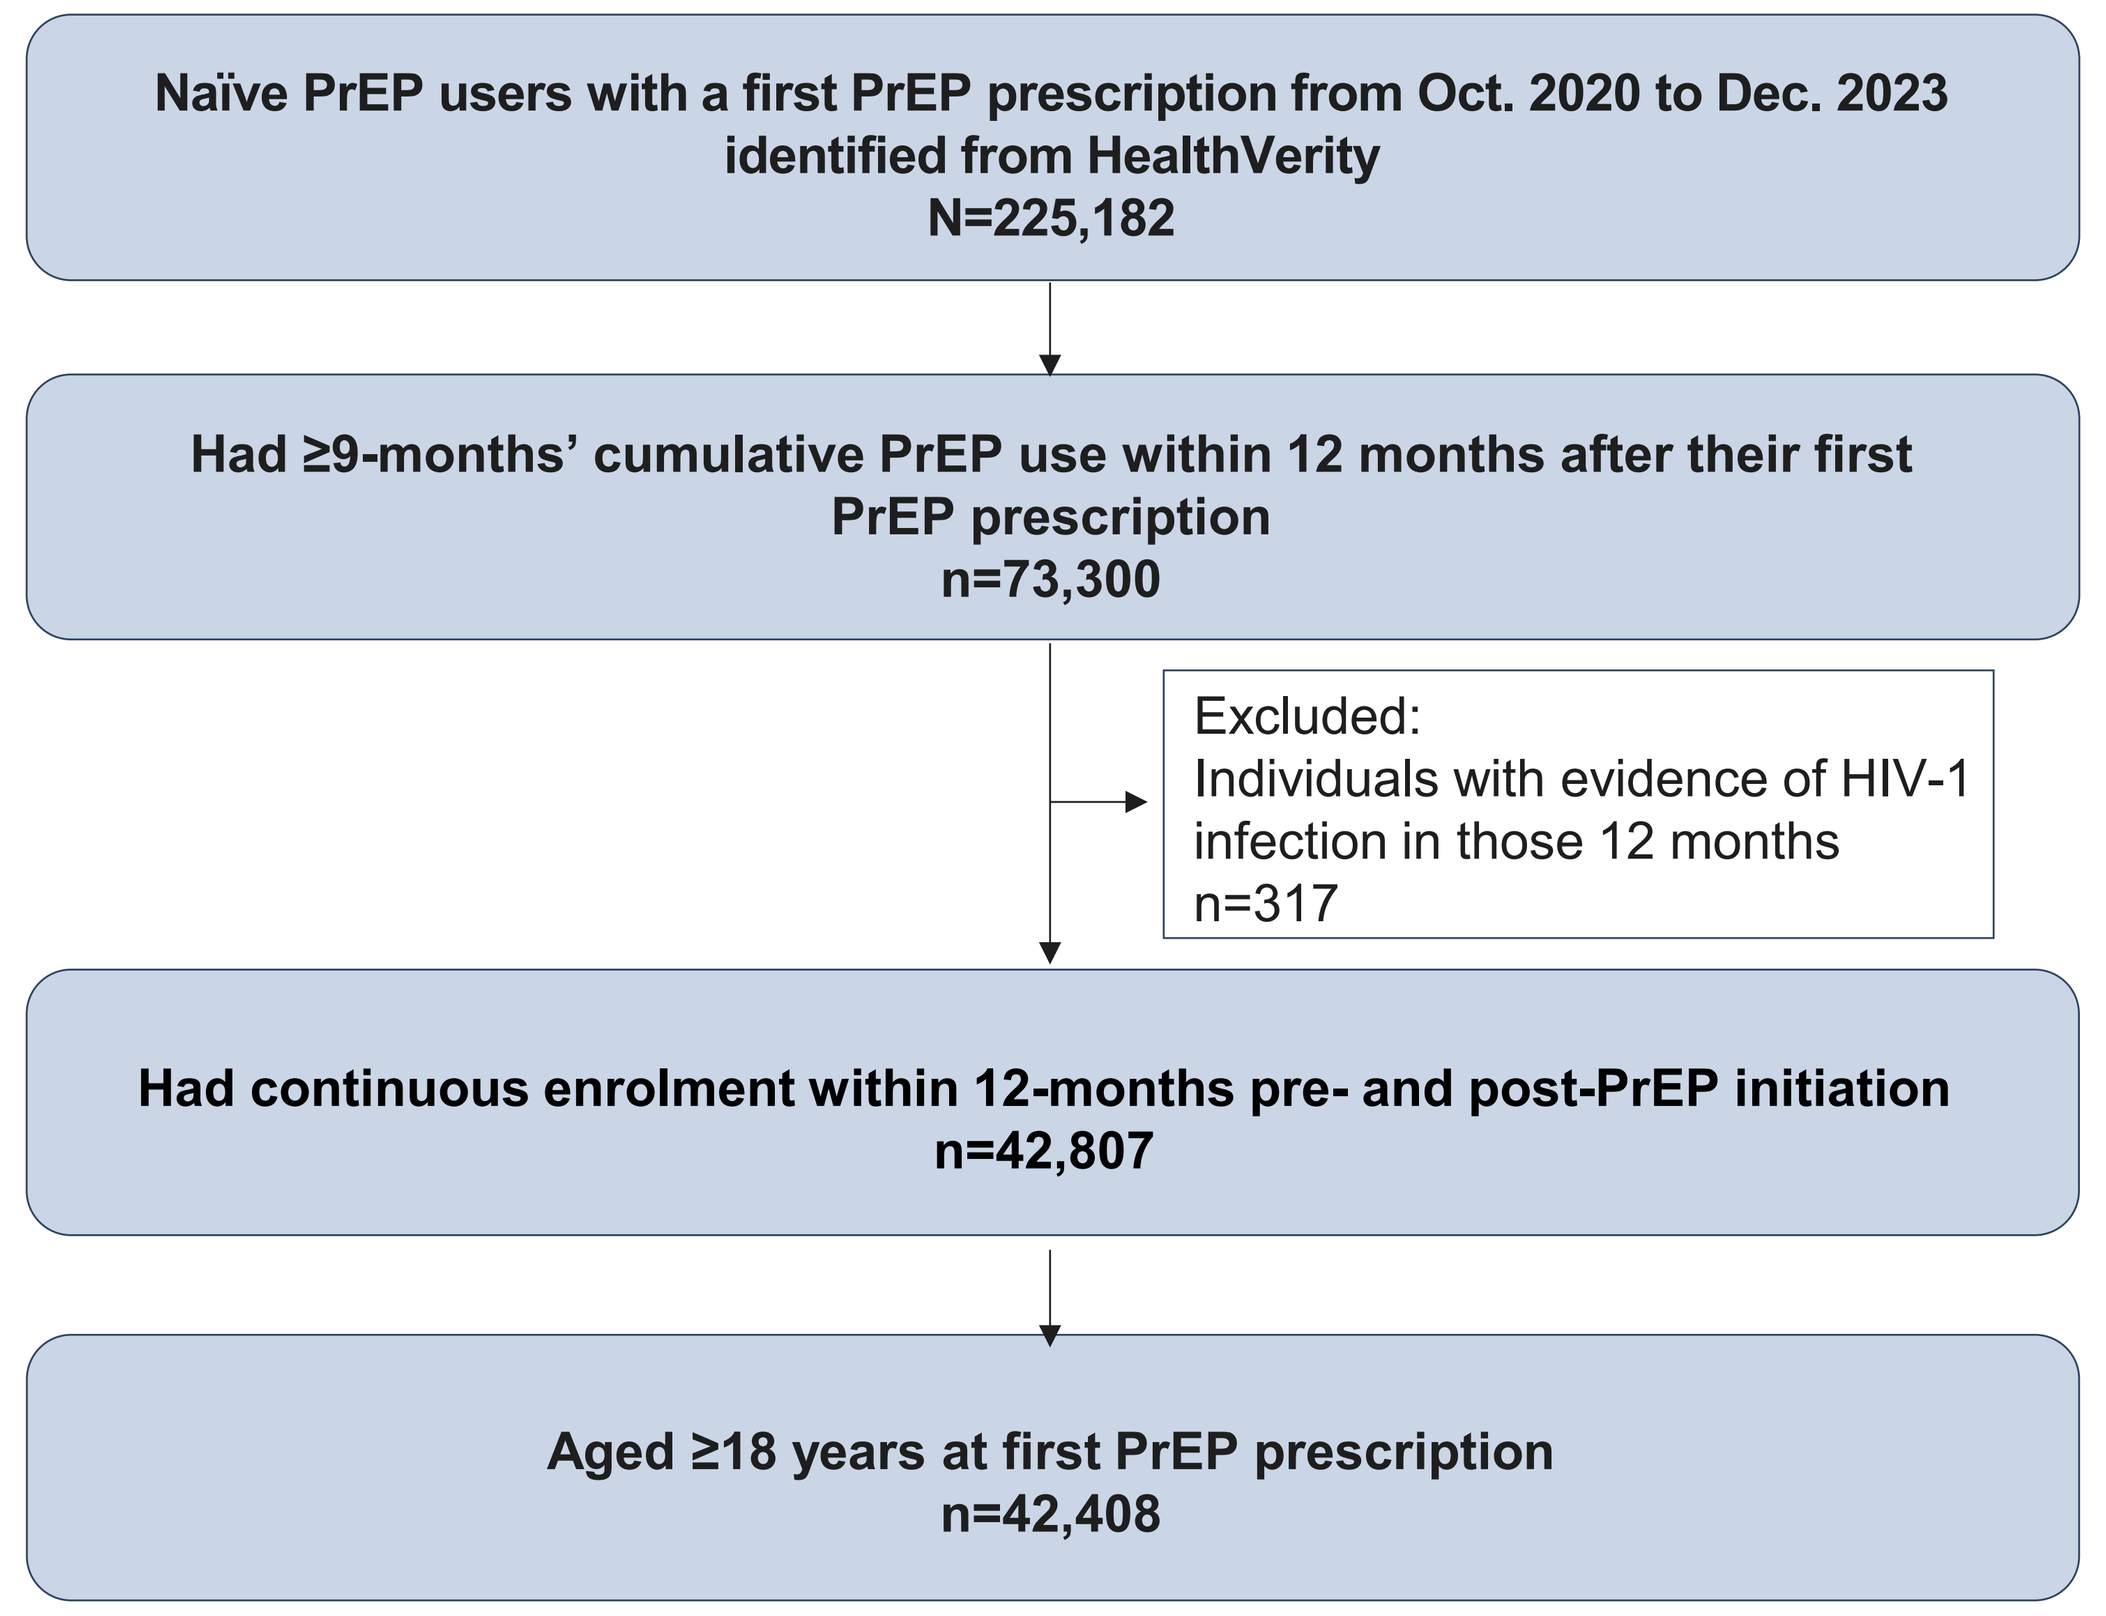

Naïve PrEP users with a first PrEP prescription from Oct. 2020 to Dec. 2023 identified from HealthVerity
N=225,182
Had ≥9-months’ cumulative PrEP use within 12 months after their first PrEP prescription
n=73,300
Excluded:
Individuals with evidence of HIV-1 infection in those 12 months
n=317
Had continuous enrolment within 12-months pre- and post-PrEP initiation
n=42,807
Aged ≥18 years at first PrEP prescription
n=42,408

PrEP, pre-exposure prophylaxis.

Table 1. Demographic Characteristics of Study Population (N=42,408)

Demographic characteristic	n (%)
Age at index date, years	
18–24	5786 (14)
25–34	15,771 (37)
35–44	10,323 (24)
≥45	10,528 (25)
Sex at birth	
Female	2744 (6)
Male	39,651 (94)
Insurance type	
Commercial	32,127 (76)
Medicaid	7832 (19)
Medicare	575 (1)
Region	
Midwest	7657 (18)
Northeast	10,573 (25)
South	13,348 (32)
West	10,274 (24)

Missing or unknown data are not shown in the table. Index date is defined as the date of first PrEP initiation.
PrEP, pre-exposure prophylaxis.

STI Testing

- Overall, 27% of all PrEP users tested for STIs within a 30-day window around PrEP initiation (initiation date ± 15 days)
- The proportion of individuals with ≥1 STI test increased from 26% pre-PrEP to 47% post-PrEP initiation (**Table 2**)
 - The proportion of individuals with ≥2 tests and the frequency of ≤6-monthly testing in these individuals increased post-PrEP initiation
 - Only 33% of all PrEP users (n=13,903) tested every ≤6 months

Table 2. STI Testing in Adults Newly Prescribed PrEP Within 12-Months Pre- and Post-PrEP Initiation

Population	Pre-PrEP initiation n (%)	Post-PrEP initiation n (%)
All	**42,408 (100)**	**42,408 (100)**
Individuals with no STI test	31,477 (74)	22,493 (53)
Individuals with ≥1 STI test	10,931 (26)	19,915 (47)
Individuals with 1 STI test only	6435 (15)	4537 (11)
Individuals with ≥2 STI tests	4496 (11)	15,378 (36)
Testing frequency among individuals with ≥2 tests		
All	4496 (100)	15,378 (100)
Every ≤3 months	1,892 (42)	6,098 (40)[a]
Every >3 to 6 months	1,901 (42)	7,805 (51)[a]
Every >6 to 9 months	540 (12)	1012 (7)
Every >9 months	163 (4)	463 (3)

[a]The number of PrEP users who tested every <6 months was calculated as 6098 + 7805 = 13,903.
STI testing was assessed 12 months pre- and post-PrEP initiation. STI tests conducted within 15 days of PrEP initiation (before or after) were counted in post-PrEP categories. Testing frequency was reported only in those individuals with ≥2 tests.
PrEP, pre-exposure prophylaxis; STI, sexually transmitted infections.

STI Testing Stratified by Demographics Among Individuals With ≥1 STI Test

- Among individuals with ≥1 STI test, STI testing frequency post-PrEP initiation increased across all demographics to approximately 30% testing every ≤3 months and 70% testing every ≤6 months, with variation observed by demographic characteristics (**Figure 2**)
 - Post-PrEP initiation, ≤3-monthly and ≤6-monthly testing frequencies were lowest in the South (27% and 66%, respectively) and among those aged ≥45 years (25% and 66%), and highest in those using Medicaid (35% and 73%)
- Pre-PrEP initiation, individuals residing in the South or Midwest, aged ≥45 years, or with commercial insurance had the lowest testing frequencies

Figure 2. Frequency of STI Testing Among Individuals With ≥1 STI Test Within the 12-Month Observation Window Pre- and Post-PrEP Initiation

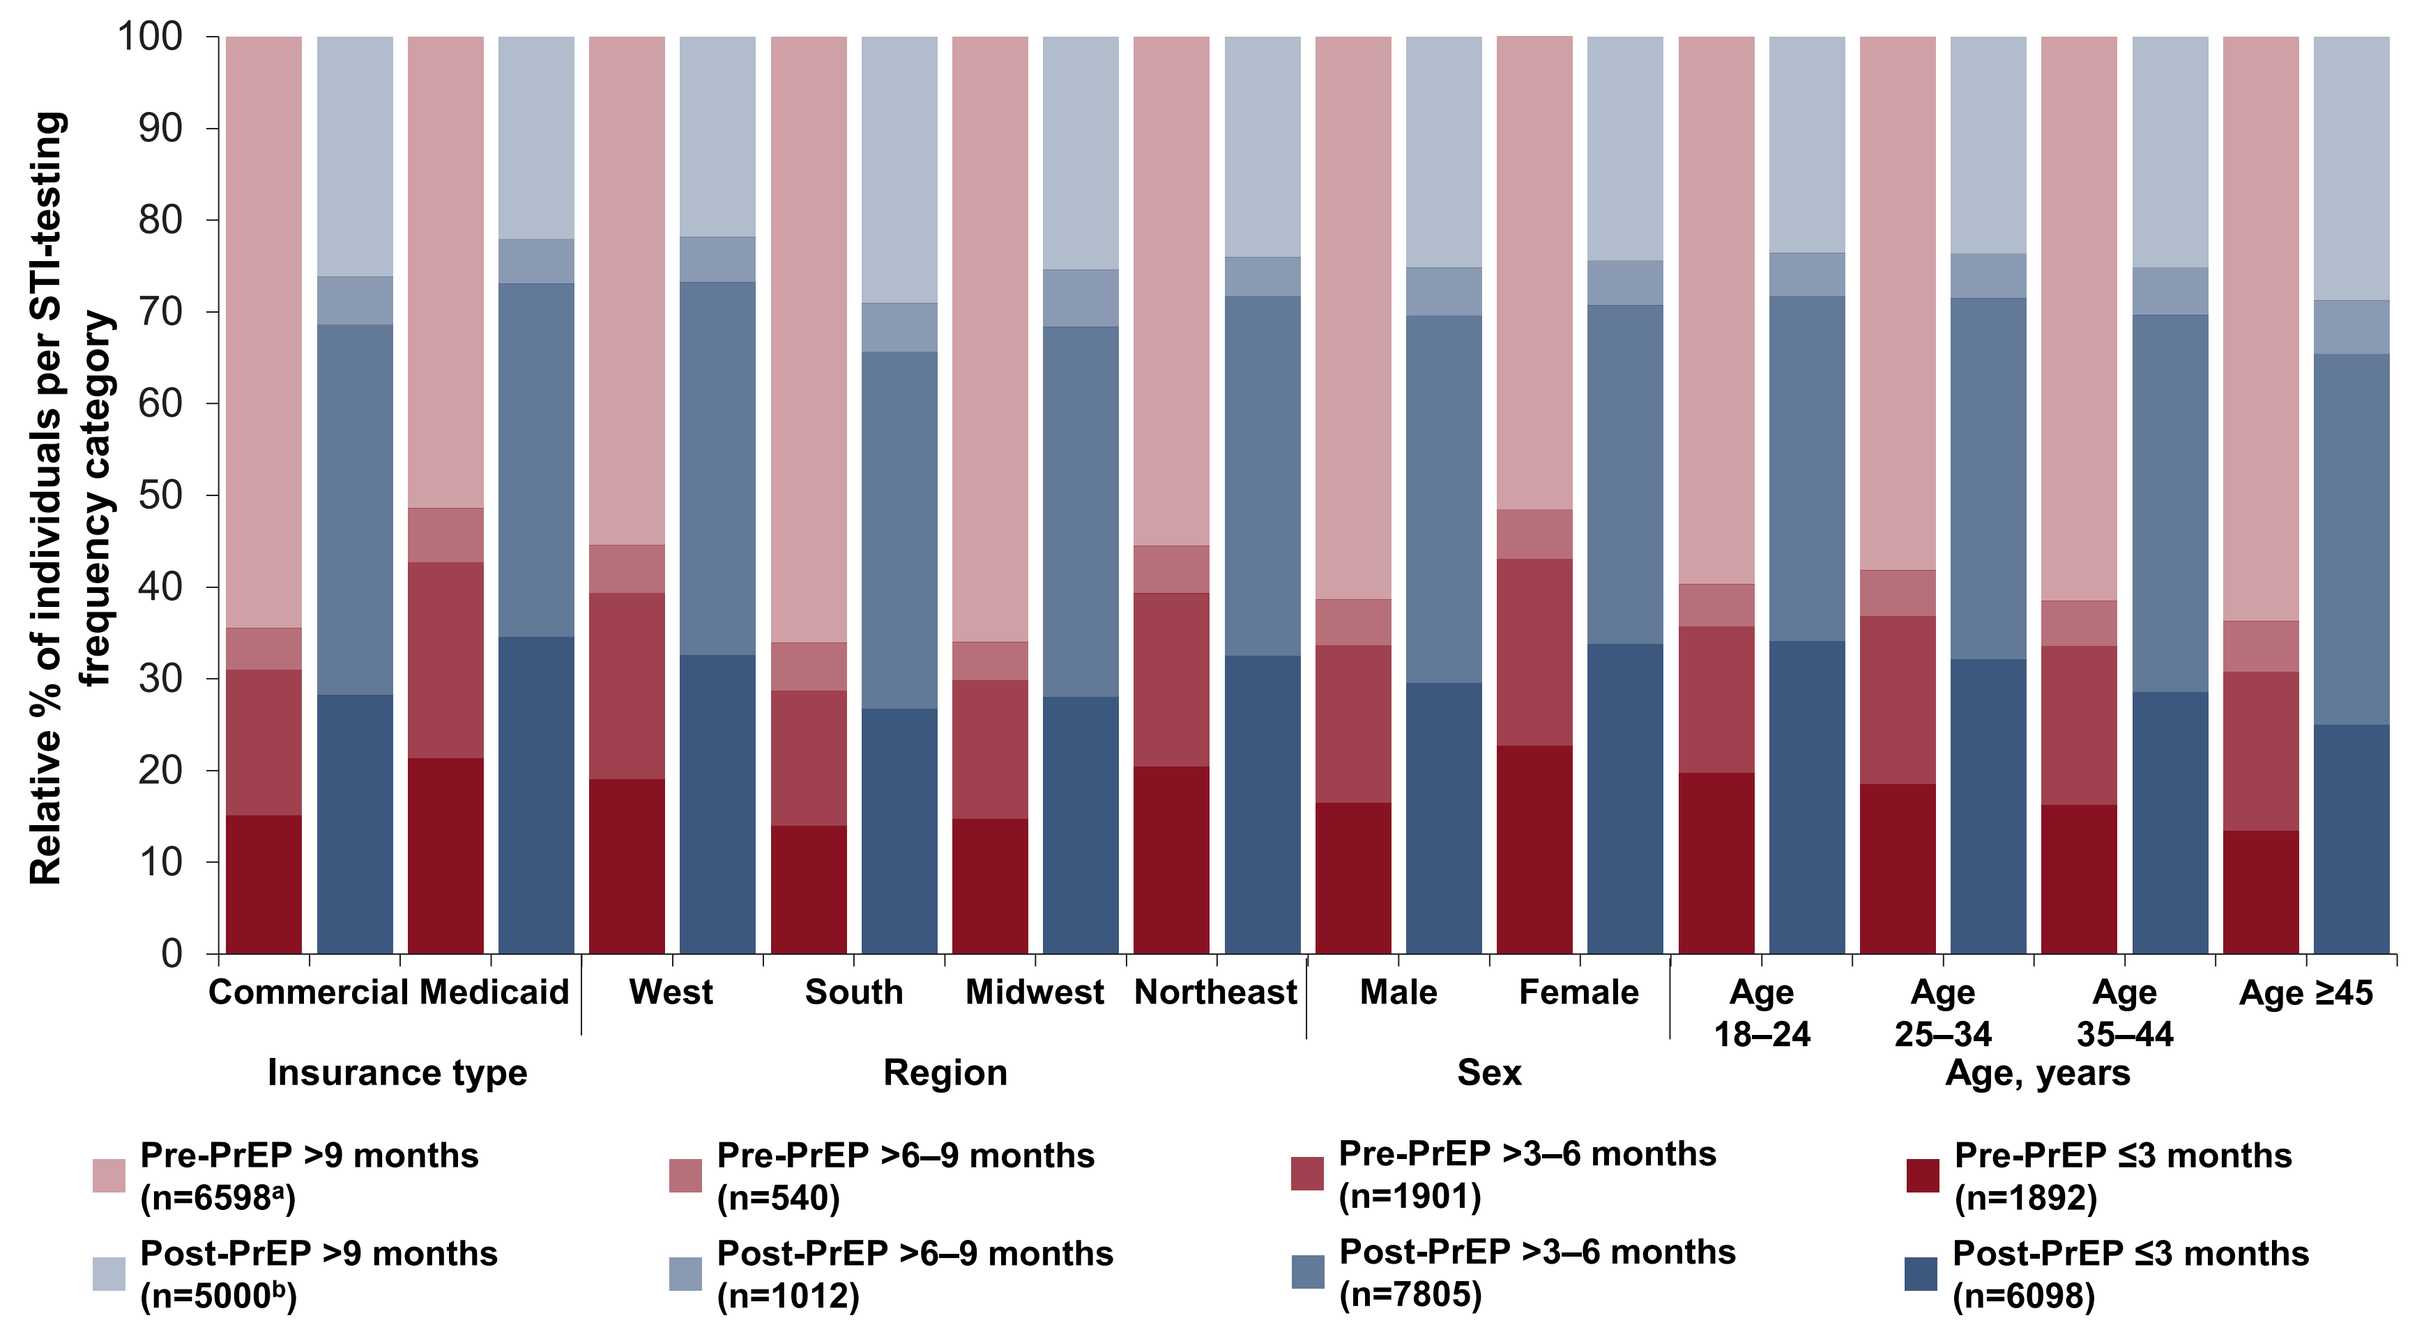

Relative % of individuals per STI-testing frequency category
100
90
80
70
60
50
40
30
20
10
0
Commercial
Medicaid
West
South
Midwest
Northeast
Male
Female
Age 18–24
Age 25–34
Age 35–44
Age ≥45
Insurance type
Region
Sex
Age, years
Pre-PrEP >9 months (n=6598[a])
Post-PrEP >9 months (n=5000[b])
Pre-PrEP >6–9 months (n=540)
Post-PrEP >6–9 months (n=1012)
Pre-PrEP >3–6 months (n=1901)
Post-PrEP >3–6 months (n=7805)
Pre-PrEP ≤3 months (n=1892)
Post-PrEP ≤3 months (n=6098)

STI tests conducted within 15 days of PrEP initiation (before or after) were counted in post-PrEP categories. [a]Included individuals with 1 STI test only (n=6435) and individuals with a testing frequency >9 months (n=163). [b]Included individuals with one STI test only (n=4537) and individuals with a testing frequency >9 months (n=463).
PrEP, pre-exposure prophylaxis; STI, sexually transmitted infection.

STI Testing Intervals Among Individuals With ≥2 STI Tests

- Mean testing intervals shortened slightly from 3.8 to 3.7 months pre- and post-PrEP initiation, respectively, among individuals with ≥2 STI tests (**Figure 3**)
 - The mean testing interval was slightly longer in male vs female individuals pre- and post-PrEP use
- The average time intervals pre- and post-PrEP use were concentrated around 3 months across overall, male, and female populations
- The distribution of testing intervals exhibited high variability and was skewed toward longer time intervals between tests

Figure 3. Violin Plots to Display Mean STI-Testing Intervals Among Individuals With ≥2 STI Tests in the 12-Months Pre- and Post-PrEP Initiation

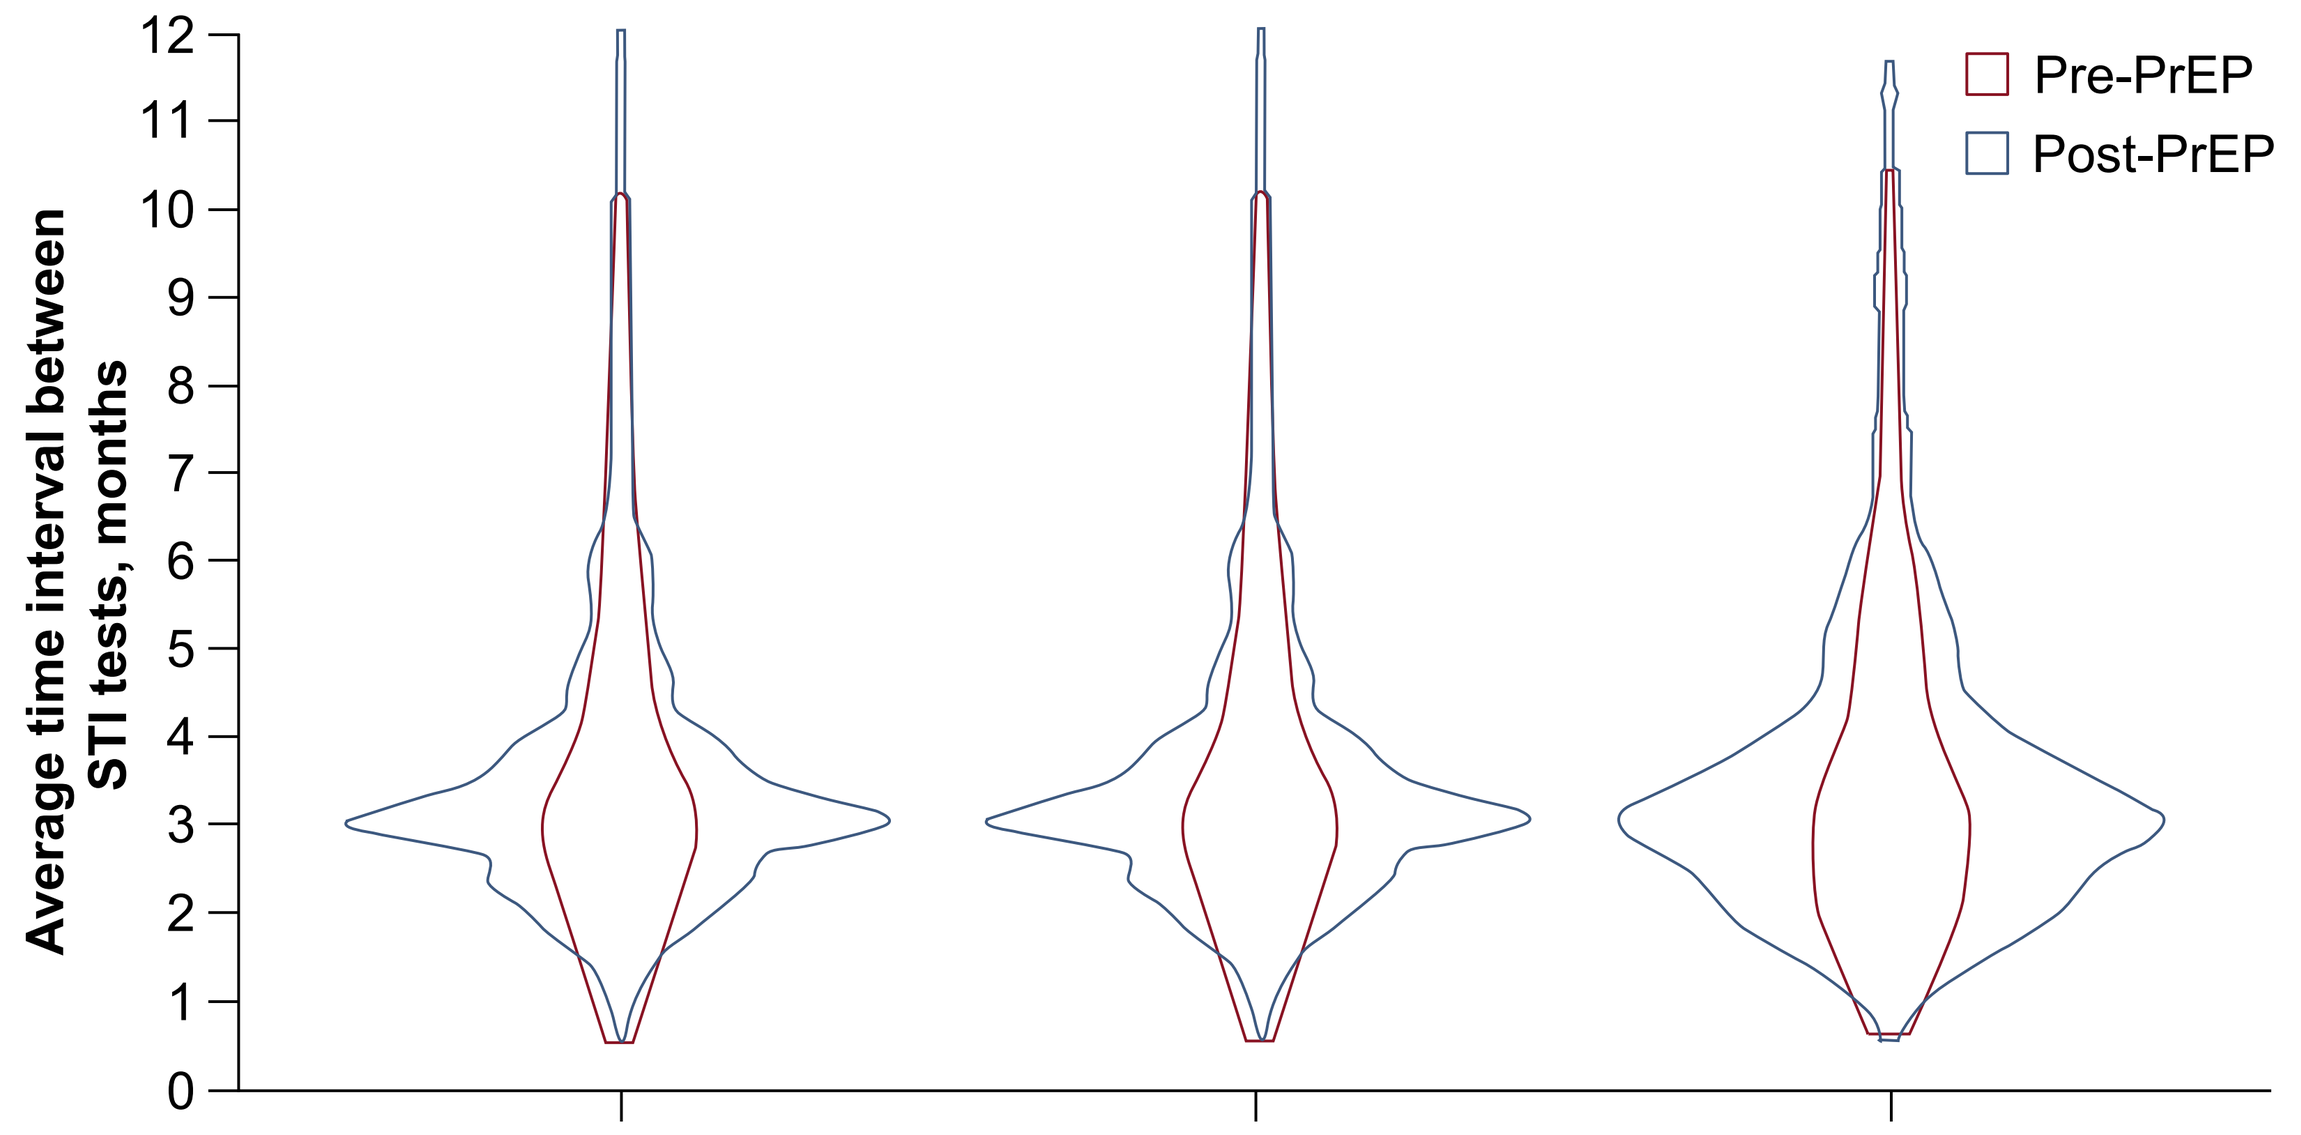

Average time interval between STI tests, months
12
11
10
9
8
7
6
5
4
3
2
1
0
Pre-PrEP
Post-PrEP

Mean testing interval, months	Overall[a]	Male	Female
Pre-PrEP	3.81 (n=4495)	3.83 (n=3935)	3.68 (n=560)
Post-PrEP	3.68 (n=15,374)	3.69 (n=14,102)	3.59 (n=1272)

STI tests conducted within 15 days of PrEP initiation (before or after) were counted in post-PrEP categories. [a]Individuals with unknown sex at birth were excluded.
PrEP, pre-exposure prophylaxis; STI, sexually transmitted infection.

STI Prevalence

- The measured prevalence of STIs was 12% pre-PrEP initiation and 18% post-PrEP initiation
 - The highest prevalence of STIs was observed among individuals aged 18–24 years (21%), those enroled in Medicaid (23%), and those living in the West (22%)

Limitations

- This claims-based analysis did not capture clinical data on STI-related symptoms; however, there may be important differences between testing for symptomatic and asymptomatic STIs
- These data may underestimate STI tests if individuals were tested outside their insurance plan (e.g., home testing) or at a health system that does not contribute to the dataset
- Individual-level PrEP adherence data were not available for this study; therefore, the correlation between PrEP adherence and STI testing patterns could not be assessed

References: 1. Centers for Disease Control and Prevention. 2021. Preexposure prophylaxis for the prevention of HIV infection in the United States - 2021 update. Available at: https://stacks.cdc.gov/view/cdc/112360. Accessed September 2025. **2.** Centers for Disease Control and Prevention. 2025. Clinical Guidance for PrEP. Available at: https://www.cdc.gov/hivnexus/hcp/prep/index.html#cdc_clinical_guidance_managing_disease-managing-patients-on-prep. Accessed September 2025. **3.** The Global PrEP Tracker: PrEP Watch. AVAC. 2022. Available at: https://data.prepwatch.org. Accessed September 2025. **4.** The urgency of now: AIDS at a crossroads. Geneva: Joint United Nations Programme on HIV/AIDS. 2024. Available at: https://www.unaids.org/sites/default/files/media_asset/2024-unaids-global-aids-update_en.pdf. Accessed September 2025. **5.** Centers for Disease Control and Prevention. 2024. Sexually Transmitted Infections Surveillance, 2023. Available at: https://www.cdc.gov/sti-statistics/annual/index.html. Accessed September 2025.

Acknowledgments: This study was funded by Gilead Sciences, Inc.
Medical writing and editorial support were provided by Amy Watkins, PhD, and Sherriden Beard, MA, of Ashfield MedComms (Macclesfield, UK), an Inizio company, and funded by Gilead Sciences, Inc.

Conflicts of Interest: **Xiwen Huang, Juan Yang, Edward Gemson, Wenyi Wang, Li Tao, Dylan Mezzio, Chris Nguyen, Sandra I. McCoy,** and **Joshua Gruber** are all employees and shareholders of Gilead Sciences, Inc.

***Correspondence:** Xiwen Huang, Xiwen.Huang7@gilead.com

IDWeek 2025, October 19–22, 2025. Atlanta, Georgia, USA