Barriers to Oral HIV Pre-Exposure Prophylaxis Perceived by Those Receiving an Initial Prescription: USA Survey Analysis

P-326

Patrick S. Sullivan[1]*, JeanPierre Coaquira Castro[2], Krisha Patel[3], Alice Hsiao[2], Iyue Sung[3†], Jessica Citronberg[3], Michael Bogart[2], Woodie Zachry[2]

[1]Emory University Rollins School of Public Health, Atlanta, GA, USA; [2]Gilead Sciences, Inc., Foster City, CA, USA; [3]Walgreens Co., Deerfield, IL, USA.

*Presenting Author. †At time of analysis.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- Results of this cross-sectional survey revealed that many individuals picking up an initial HIV-1 oral pre-exposure prophylaxis (PrEP) prescription encountered obstacles that were associated with delayed or no pickup
- Cost concerns and insurance coverage issues were significantly more prevalent among individuals who did not pick up their first PrEP prescription within 14 days of when their healthcare provider first prescribed it versus those that did pick up their prescription within 14 days
- Overall, 21% of individuals were told their prescription could not be processed properly, nearly half (43%) of whom were not given a reason; among those given a reason, lack of insurance coverage and the need for prior authorization (PA) were the most commonly reported
- These findings suggest that efforts to address barriers to PrEP at the pharmacy level should target reducing out-of-pocket (OOP) costs and streamlining medical authorization processes to further encourage PrEP initiation

Plain Language Summary

- Medications that prevent HIV, also called "PrEP", are available in the USA. But some people who need or want to take PrEP have problems getting it
- Most insurance plans cover PrEP. However, coverage amounts vary, and some plans ask for healthcare providers to get "prior authorization" – special approval from the insurance company before they pay for the medicine
- We asked people picking up their first PrEP prescription about what they knew about PrEP, how they felt about taking it, and any costs or problems they faced at the pharmacy
- Researchers compared answers between people who picked up their PrEP within 14 days of their healthcare provider prescribing it and those who did not
- Many people had problems when trying to pick up their prescription at the pharmacy, such as insurance not covering it or needing prior authorization from their healthcare provider
- People who did not pick up their prescription within 14 days were more likely to have issues with cost, insurance coverage, and their overall budget
- To help more people take PrEP in the USA, we need to make it easier and less expensive to pick up a first prescription – especially for those worried about money

Background

- The Ending the HIV Epidemic (EHE) initiative, introduced by the US Department of Health and Human Services in 2019, targets a 90% reduction in new HIV-1 cases in the US by 2030[1]
 - — The US Preventive Services Task Force (USPSTF) provided a Grade A recommendation in 2019 that PrEP be made available to any person at high risk of HIV-1 acquisition, and in 2023, to any person at increased risk[2,3]
 - — Under the Affordable Care Act (ACA), most private health insurance plans are required to cover USPSTF Grade A and B recommendations with zero OOP costs and no PA[4]
- Between 2023 to 2024, PrEP use in the US increased by 17% to 591,475 PrEP users; uptake, however, remains disproportionately low in individuals who are young, female, Black or Hispanic/Latine, and who live in the South[5]
 - — Barriers to PrEP uptake include a lack of PrEP awareness, concerns about side effects, stigma, limited geographic access, and costs[6] (**Figure 1**)
- Despite USPSTF recommendations and ACA insurance coverage requirements, OOP costs and PA vary widely across plans[7,8]
- There is a lack of research on the barriers to PrEP perceived by individuals picking up their first PrEP prescription
 - — We previously reported the results of a pilot study identifying cost and insurance coverage problems as the most reported issues associated with delays in picking up an initial PrEP prescription[9]
 - — Here we present the final results of this survey study

Figure 1. A Chronology of Various Factors Impacting Effective PrEP Uptake

Know about PrEP and be willing to try it	Obtain a prescription for PrEP	Successfully pick up PrEP at pharmacy	Have good adherence and persistence
• Knowledge of PrEP • Safety concerns • Perception of risk	• Access to HCP who will prescribe	• Cost concerns • Access to pharmacy • Insurance coverage and restrictions	• Access to HCP and pharmacy • Perception of risk • Daily pill burden

Persistent factors: PrEP-related stigma, trust in healthcare

HCP, healthcare provider; PrEP, pre-exposure prophylaxis.

Objective

- To describe factors associated with failure to pick up a first PrEP prescription within 14 days among people prescribed PrEP for the first time

Methods

Survey Design

- Adults with an initial PrEP prescription and without evidence of an HIV-1 infection were invited to complete an electronically-deployed, IRB-approved survey in English or Spanish
 - — Eligible individuals had a first prescription for PrEP at a Walgreens pharmacy in the USA ≤7 months prior to being invited to participate in the pilot phase (March–April 2024) or final study phase (July–December 2024) of the survey
 - — Individuals were contacted by text, email, or both, consented to participate, and were compensated upon survey completion
- A prescreener questionnaire assessed respondent eligibility and collected additional respondent information
 - — Eligibility information included age, prior HIV-1 diagnosis, and indication and timing of first PrEP prescription
 - — Information on insurance type, cost of PrEP, and state where dispensing pharmacy was located were obtained retrospectively from the pharmacy database
- The survey comprised 35 multiple choice and Likert scale questions on topics including attitudes toward PrEP, OOP expenses at pickup, and prescription fill rejection reasons
 - — Responses to Likert scale questions were dichotomized to increase statistical power

Data Analysis

- Data were combined across the pilot and final survey
 - — No major differences were found between study phases in question content, data collection, or distribution of responses
- Responses were compared between respondents who picked up their prescription within 14 days of when the order was first received at the pharmacy (prompt pickup [PP]) and those who did not pick up within 14 days (delayed/no pickup [D/NP]) using Chi-square/Fisher's exact tests (significance threshold $p \leq 0.05$)
- Survey responses were grouped into five key topics: Demographics and health-related social needs, PrEP attitudes and stigma, self-efficacy, healthcare experience, and cost and coverage of PrEP
- Univariate logistic regression analysis was used to calculate odds ratios (OR) and confidence intervals (CI) for the association between pickup status and predictor variables
 - — OR values indicate the odds of observing a predictor variable among those with PP versus D/NP, where OR>1 favors PP and OR<1 favors D/NP
 - — Results were considered statistically significant when the 95% CI excluded a value of 1.0
- Associations were also assessed with a multivariate logistic regression model, adjusted for age, sex, race, payer type, and pharmacy region

Results

Respondent Overview

- Of the 39,596 individuals invited to complete the survey, 1432 registered (4%) to participate; of these, 824 (58%) individuals were eligible for inclusion and 773 (54%) completed the survey (**Figure 2A**)
 - — Among respondents, 199 (26%) participated in the pilot and 574 (74%) in the final study
 - — In total, 625 (81%) respondents picked up promptly and 148 (19%) had delayed or no pickup
- Individuals who registered for the survey most often used a pharmacy in the South (34%), followed by the Midwest (29%), and West (22%), and least often in the Northeast (14%) (**Figure 2B**)
 - — 49% of respondents' pharmacies were located in a state with a PrEP drug assistance program (DAP) (**Figure 2B**)

Figure 2. Respondent Overview (A) and Geographic Locations of Walgreens Pharmacies (B)

A

Individuals in Walgreens' database with an initial oral PrEP prescription[a] within 7 months of invitation period
N=39,596

↓

Invited individuals who completed the survey registration (pilot and final study, combined)
n=1432

→ **Excluded:[a]** Not prescribed PrEP ≤7 months[b] (n=565); Medication not prescribed for HIV-1 prevention[c] (n=63)

↓

Eligible individuals
n=824

→ **Excluded:** Did not complete the survey (n=51)

↓

Eligible individuals who completed the survey
n=773

↓

PP: Prescription filled, dispensed within 14 days
n=625
Includes 160 (26%) from pilot

D/NP: Prescription filled, not dispensed by Day 14
n=148
Includes 39 (26%) from pilot

B

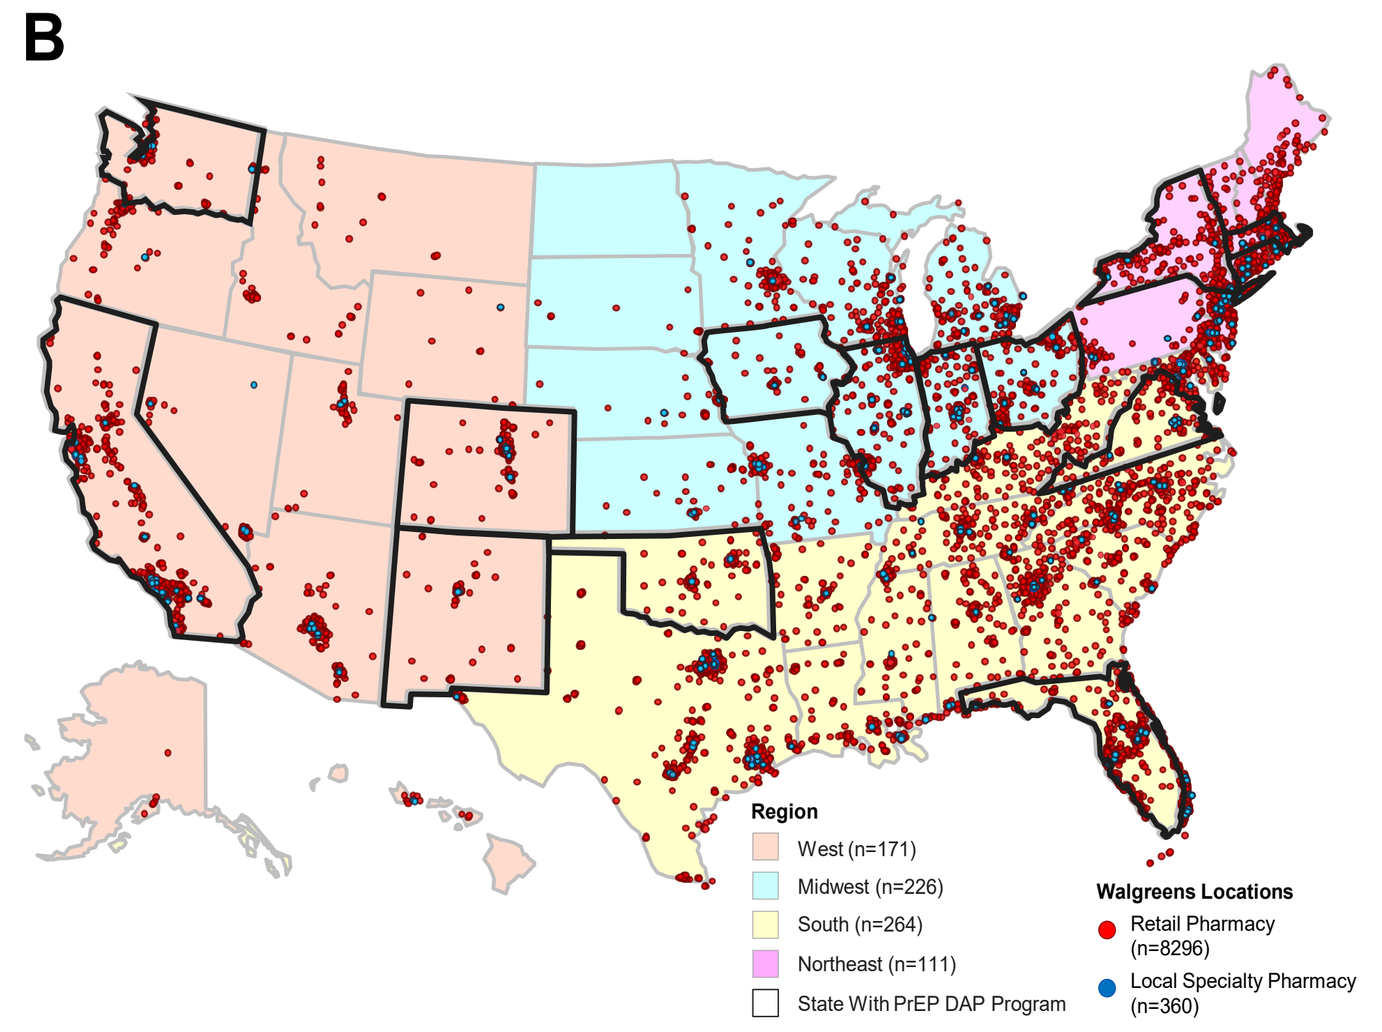
Region
West (n=171)
Midwest (n=226)
South (n=264)
Northeast (n=111)
State With PrEP DAP Program
Walgreens Locations
Retail Pharmacy (n=8296)
Local Specialty Pharmacy (n=360)

Figure B shows the US states and number of survey respondents in each region; states boxed in black have PrEP DAP programs.
[a]Exclusion items were not mutually exclusive. [b]GPI codes specific for PrEP medications (including F/TDF and F/TAF) were used to identify individuals for survey invitation; GPI codes specific to antiretroviral treatment were used to exclude individuals with prior HIV-1 treatment. [c]Medication prescribed for post-exposure prophylaxis or treatment of HIV-1 or hepatitis B virus.
DAP, drug assistance program; D/NP, delayed/no pickup; F/TAF, emtricitabine/tenofovir alafenamide fumarate; F/TDF, emtricitabine/tenofovir disoproxil fumarate; GPI, generic product identifier; PP, prompt pickup; PrEP, pre-exposure prophylaxis.

Study Population (Table 1)

- Respondent characteristics were similar between groups, with the exception of OOP costs
 - — 84% of PP respondents had $0 OOP costs for PrEP compared with 47% of D/NP respondents
 - — 46% of D/NP respondents had no cost data, primarily due to no pickup and thus no charge data, compared with 2% of PP respondents
- The frequency of respondents using a pharmacy in a state without a PrEP DAP was slightly higher in the D/NP group (57%) than the PP group (50%)
- No differences in region of pharmacy were observed between dispensation groups

Table 1. Characteristics of Survey Respondents by PrEP Dispensation Group, USA, March–December, 2024

Characteristic, n (%)		Total (N=773)	Prompt pickup (n=625)	Delayed/no pickup (n=148)
Age (y)	<25	99 (12.8)	79 (12.6)	20 (13.5)
	25–34	248 (32.1)	190 (30.4)	58 (39.2)
	35–44	227 (29.4)	188 (30.1)	39 (26.4)
	45–54	103 (13.3)	91 (14.6)	12 (8.1)
	55–65	63 (8.2)	57 (9.1)	6 (4.1)
	>65	33 (4.3)	20 (3.2)	13 (8.8)
Biological sex	Male	626 (81.0)	505 (80.8)	121 (81.8)
	Female	133 (17.2)	110 (17.6)	23 (15.5)
	Other[a]	14 (1.8)	10 (1.6)	4 (2.7)
Race	Asian	39 (5.0)	28 (4.5)	11 (7.4)
	Black	79 (10.2)	58 (9.3)	21 (14.2)
	Hispanic/Latine	91 (11.8)	71 (11.4)	20 (13.5)
	White	452 (58.5)	384 (61.4)	68 (45.9)
	Multiple races	96 (12.4)	75 (12.0)	21 (14.2)
	Other[b]	16 (2.1)	9 (1.4)	7 (4.7)
Educational level	High school or less	98 (12.7)	77 (12.3)	21 (14.2)
	1–3 y college	250 (32.3)	202 (32.3)	48 (32.4)
	≥4 y college	415 (53.7)	338 (54.1)	77 (52.0)
	Prefer not to answer	10 (1.3)	8 (1.3)	2 (1.4)
Payer type[c]	Commercial	489 (63.3)	407 (65.1)	82 (55.4)
	Government funded	200 (25.9)	162 (25.9)	38 (25.7)
	Cash	0 (0)	0 (0)	0 (0)
	Discount card, PAP, or other	81 (10.5)	56 (9.0)	25 (16.9)
Region	Northeast	111 (14.4)	91 (14.6)	20 (13.5)
	Midwest	226 (29.2)	178 (28.5)	48 (32.4)
	South	264 (34.2)	216 (34.6)	48 (32.4)
	West	171 (22.1)	140 (22.4)	31 (20.9)
PrEP DAP State[d]	PrEP DAP	379 (49.0)	315 (50.4)	64 (43.2)
	Non-PrEP DAP	394 (51.0)	310 (49.6)	84 (56.8)
PrEP OOP cost	$0	591 (76.5)	522 (83.5)	69 (46.6)
	>$0 to ≤$10	46 (6.0)	40 (6.4)	6 (4.1)
	>$10 to ≤$25	23 (3.0)	21 (3.4)	2 (1.4)
	>$25 to ≤$100	31 (4.0)	29 (4.6)	2 (1.4)
	>$100 to ≤$500	3 (0.4)	3 (0.5)	0 (0)
	No data[e]	79 (10.2)	10 (1.6)	69 (46.6)

[a]Includes none of the above and prefer not to answer. [b]Includes American Indian or Alaska Native, Middle Eastern or North African, and prefer not to answer. [c]Government funded includes Medicare Part D, Medicare Part B, managed Medicaid, and state Medicaid. [d]Includes California, Colorado, Washington DC, Florida, Illinois, Indiana, Iowa, Massachusetts, New Mexico, New York, Ohio, Oklahoma, Virginia, and Washington State. [e]Cost data unavailable due to no pickup within 30 days or otherwise missing data.
DAP, drug assistance program; OOP, out-of-pocket; PAP, patient assistance program; PrEP, pre-exposure prophylaxis; y, years.

PrEP Costs and Financial Burdens (Table 2)

- Prompt pickup was significantly ***less likely*** for individuals who:
 - — Reported that the cost of PrEP was important in their decision to pick up (OR, 0.46) or was difficult to pay (OR, 0.40)
 - — Had high versus moderate concern about their job and income stability (OR, 0.63)
- Prompt pickup was significantly ***more likely*** when individuals:
 - — Were "very much" versus "somewhat" able to meet their monthly expenses (OR, 1.82)
 - — Reported their OOP general medical costs as "not at all" versus "somewhat" more than expected (OR, 1.86)

Insurance Coverage and Claims Issues (Table 2)

- Prompt pickup was significantly ***less likely*** for individuals who:
 - — Were covered under Medicare (OR, 0.46) or who used cash/other means (OR, 0.45)
 - — Encountered insurance processing issues (OR, 0.21) and reported experiencing delays due to those issues (OR, 0.10)

Table 2. Relationship Between Prompt PrEP Dispensation and PrEP Cost, Insurance Coverage, and Health-Related Social Needs, USA, March–December, 2024

Domain	Question content	Category	Ref.	Univariable OR (95% CI)	Multivariable OR (95% CI)[a]
Cost and coverage, pharmacy data	Pharmacy region	Midwest	North-east	0.82 (0.45–1.44)	-
		South		0.99 (0.55–1.74)	-
		West		0.99 (0.53–1.84)	-
	Pharmacy in state with PrEP DAP program	No	Yes	0.75 (0.52–1.07)	0.71 (0.47–1.05)
	Payer type	Medicare	Com-mercial	0.46 (0.26–0.84)	-
		Medicaid		1.26 (0.75–2.22)	-
		Cash/other		0.45 (0.27–0.77)	-
Cost and coverage, survey responses	Had to pay for all or part of PrEP at first pickup attempt	Yes	No	0.73 (0.48–1.13)	0.78 (0.49–1.26)
	OOP cost	>$0	$0	1.23 (0.64–2.62)	1.4 (0.60–3.19)
	If told an OOP cost, how important was that cost in decision to pick up	Important	Not imp.	0.46 (0.20–0.93)	0.41 (0.16–0.89)
	Based on OOP cost, finding a way to pay for PrEP was/would be	Hard	Easy	0.40 (0.27–0.59)	0.46 (0.30–0.71)
		Not told cost		1.20 (0.68–2.20)	1.12 (0.62–2.10)
	Told that PrEP could not be processed or properly charged to insurance	Yes	No	0.21 (0.14–0.32)	0.20 (0.13–0.32)
	Issue with processing PrEP delayed/prevented pickup			0.10 (0.02–0.37)	0.06 (0.01–0.28)
Health-related social needs	Paying for basic necessities is	Hard	Easy	0.71 (0.49–1.02)	0.78 (0.52–1.17)
	Would describe current living situation as: none (no steady housing), worried (steady housing but worried about losing it), steady (steady place to live)	None	Steady	0.54 (0.25–1.27)	0.70 (0.29–1.87)
		Worried		0.68 (0.43–1.08)	0.76 (0.46–1.29)
	Transportation issues have delayed/prevented attending medical appointments	Yes	No	0.68 (0.42–1.10)	0.86 (0.50–1.54)
	Transportation issues have delayed/prevented picking up prescriptions			0.63 (0.39–1.03)	0.72 (0.42–1.27)
Financial burden	Able to meet my monthly expenses (within last 14 days)	Very much	Some–what	1.82 (1.23–2.69)	1.76 (1.15–2.69)
		Not at all		1.21 (0.68–2.26)	1.16 (0.62–2.26)
	OOP medical expenses are more than expected (within last 14 days)	Very much		0.70 (0.43–1.16)	0.76 (0.44–1.30)
		Not at all		1.86 (1.17–2.96)	1.78 (1.08–2.94)
	Concerned about keeping job and income (within last 14 days)	Very much		0.63 (0.40–0.99)	0.80 (0.48–1.31)
		Not at all		0.78 (0.50–1.22)	0.84 (0.52–1.34)

Univariate Analysis: More likely to delay/not pickup ← OR (95% CI) → More likely to pick up promptly (axis 0, 0.5, 1.0, 1.5, 2.0, 2.5, 3.0)

Statistical significance indicated by red text.
[a]Adjusted by age, sex, race, payer type, pharmacy region.
CI, confidence interval; DAP, drug assistance program; OOP, out-of-pocket cost; OR, odds ratio; PrEP, pre-exposure prophylaxis.

Health-Related Social Factors (Table 2)

- The likelihood of prompt pickup was consistently lower, but non-statistically significant, for individuals with unsteady housing, transportation barriers, and higher-than-expected OOP medical expenses in general versus respondents without these concerns

Other Assessed Factors

- No significant associations were found between pickup group and respondent demographics, perceived risk, PrEP attitudes, HIV-1 stigma, self-efficacy, and satisfaction in healthcare provider (**Table 3**)
- For all factors, directionality of multivariate logistic regression results were consistent with univariate logistic regression results (**Tables 2 and 3**)

Table 3. Relationship Between Prompt PrEP Dispensation and Other Assessed Variables, USA, March–December, 2024

Domain	Question content	Category	Ref.	Univariable OR (95% CI)	Multivariable OR (95% CI)[a]
Demo-graphics	Highest level of education	≤High school	≥4y college	0.84 (0.49–1.46)	0.91 (0.51–1.17)
		1–3 y college		0.96 (0.64–1.44)	1.05 (0.67–1.65)
	Sexual orientation	Heterosexual	LGB	1.36 (0.74–2.72)	1.76 (0.83–4.01)
Perceived risk	Believed risk of getting HIV without PrEP	High	Medium	1.04 (0.67–1.62)	1.20 (0.75–1.95)
		Low		1.25 (0.81–1.93)	1.14 (0.72–1.81)
PrEP attitudes	Believe that taking PrEP as told by doctor will protect against HIV	Agree	Neither agree nor disagree	1.22 (0.44–2.91)	1.36 (0.43–3.55)
		Disagree		1.14 (0.28–5.11)	0.99 (0.21–4.85)
	Believe PrEP is safe to take	Agree		1.11 (0.64–1.85)	1.09 (0.60–1.91)
		Disagree		0.68 (0.24–2.08)	0.61 (0.21–1.98)
	Believe people who take PrEP are responsible in protecting themselves from HIV	Agree		1.73 (0.83–3.38)	2.16 (1.01–4.39)
		Disagree		5.60 (0.95–107)	6.17 (1.00–120)
PrEP stigma	Would be concerned if PrEP use was discovered	Agree	Neither agree nor disagree	1.20 (0.65–2.21)	1.26 (0.65–2.47)
		Disagree		1.00 (0.61–1.58)	0.96 (0.56–1.58)
	Believe people who take PrEP are promiscuous	Agree		0.94 (0.53–1.74)	1.27 (0.66–2.52)
		Disagree		0.80 (0.53–1.19)	0.82 (0.53–1.27)
Self-efficacy	Difficulty finding information about PrEP	Hard	Easy	0.79 (0.47–1.35)	0.73 (0.43–1.31)
	Expected difficulty of taking PrEP as directed by doctor			0.98 (0.60–1.66)	1.05 (0.62–1.86)
	Difficulty visiting doctor who prescribed PrEP			0.80 (0.50–1.30)	0.79 (0.48–1.35)
	Difficulty of getting tested for HIV before starting PrEP			0.91 (0.48–1.84)	0.82 (0.42–1.70)
	Difficulty of attending recommended screenings every three months			0.87 (0.58–1.35)	0.83 (0.53–1.34)
	Talked with doctor about your sexual behaviors	No	Yes	0.84 (0.47–1.58)	0.90 (0.48–1.80)
	Degree of difficulty when talking to doctor about sexual behaviors	Hard	Easy	1.19 (0.73–2.01)	1.30 (0.76–2.33)
	Did not talk with doctor about sexual behaviors, expected degree of difficulty	Hard	Easy	0.86 (0.27–2.92)	0.48 (0.60–3.98)
	Discussed taking PrEP with most recent sexual partner(s)	No	Yes	0.73 (0.48–1.12)	0.93 (0.58–1.50)
	Talked with partner about PrEP and found it	Hard	Easy	0.91 (0.43–2.17)	0.79 (0.35–1.95)
Satisfaction in HCP	Doctor explained how to use PrEP in a way that I could understand	Agree	Neither agree nor disagree	0.39 (0.12–0.99)	0.39 (0.11–1.01)
		Disagree		0.30 (0.08–0.92)	0.36 (0.09–1.16)
	Doctor explained how to obtain PrEP in a way that I could understand	Agree		1.35 (0.68–2.52)	1.15 (0.52–2.33)
		Disagree		0.77 (0.30–1.98)	0.79 (0.27–2.25)

Univariate Analysis: More likely to delay/not pickup ← OR (95% CI) → More likely to pick up promptly (axis 0, 0.5, 1.0, 1.5, 2.0, 2.5, 3.0)

Statistical significance indicated by red text.
[a]Adjusted by age, sex, race, payer type, pharmacy region.
CI, confidence interval; DAP, drug assistance program; HCP, healthcare provider; LGB, lesbian, gay, or bisexual; OR, odds ratio; PrEP, pre-exposure prophylaxis; y, years.

Claims Processing Issues (Figure 3)

- Overall, 163 (21%) respondents reported encountering a claims processing issue
 - — Among these respondents,139 (85%) reported experiencing delays in pickup
 - — Many respondents (n=69, 42%) were not told why they encountered a processing issue
- Claims processing issues were reported significantly more often (p<0.01) in the D/NP group (n=67, 45%) than the PP group (n=96, 15%)
 - — These issues caused pickup delays in both groups, but delays were significantly more frequent (p<0.01) in the D/NP group (n=65, 97%) than the PP group (n=74, 77%)
 - — No differences were observed between pickup groups in the proportion of respondents given a reason for the processing issue

Figure 3. Claims Processing Issues by PrEP Pickup Group, USA, March–December, 2024

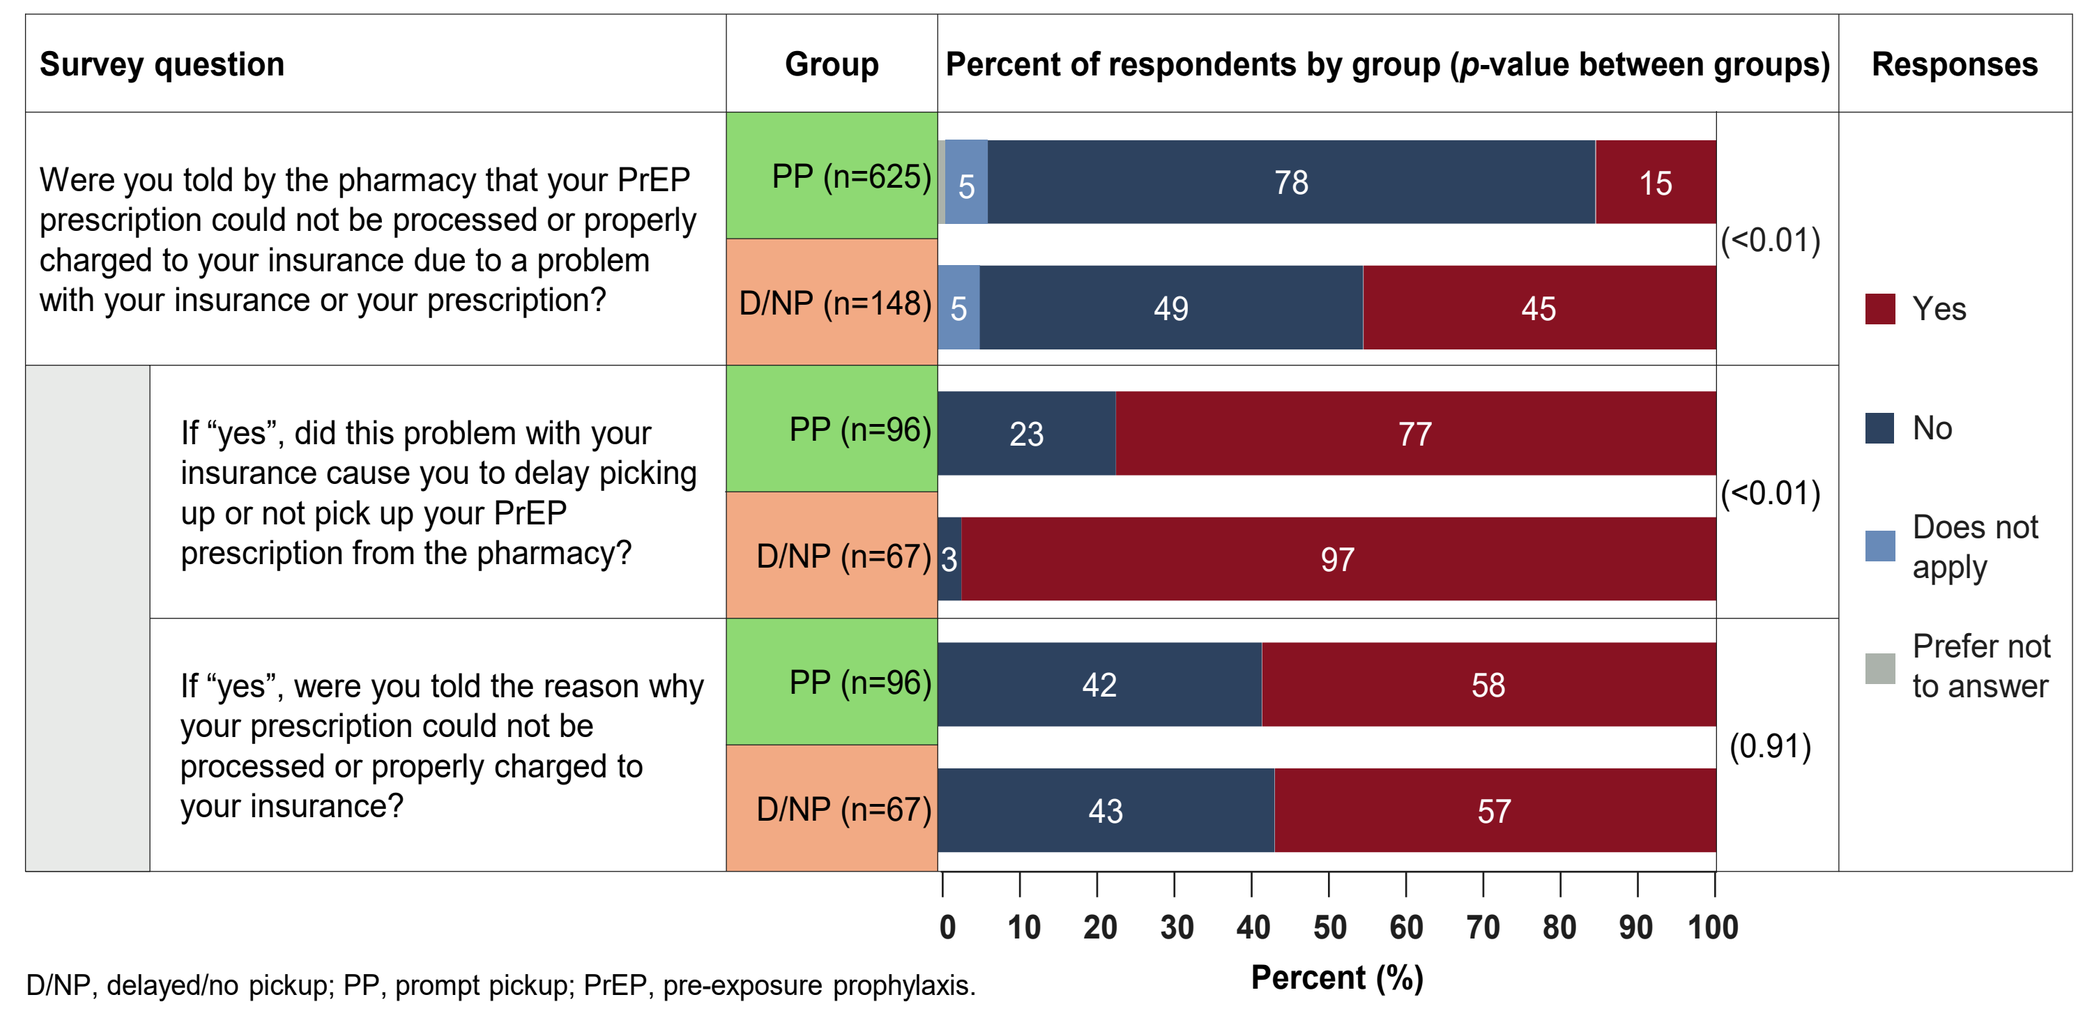
Survey question
Group
Percent of respondents by group (p-value between groups)
Responses
Were you told by the pharmacy that your PrEP prescription could not be processed or properly charged to your insurance due to a problem with your insurance or your prescription?
PP (n=625)
5
78
15
D/NP (n=148)
5
49
45
(<0.01)
If "yes", did this problem with your insurance cause you to delay picking up or not pick up your PrEP prescription from the pharmacy?
PP (n=96)
23
77
D/NP (n=67)
3
97
(<0.01)
If "yes", were you told the reason why your prescription could not be processed or properly charged to your insurance?
PP (n=96)
42
58
D/NP (n=67)
43
57
(0.91)
Yes
No
Does not apply
Prefer not to answer
0 10 20 30 40 50 60 70 80 90 100
Percent (%)

D/NP, delayed/no pickup; PP, prompt pickup; PrEP, pre-exposure prophylaxis.

Self-Reported Claims Issue Reasons (Figure 4)

- Among the 139 respondents with a claims processing issue, 94 (68%) respondents were given at least one reason, including 56 with prompt pickup and 38 with delayed/no pickup
- For the respondents with claims issues who were given a reason, self-reported prescription fill rejection reasons differed significantly (p<0.01) between pickup groups
 - — Respondents in the D/NP group compared with the PP group more often reported encountering the need for PA (32% versus 21%) and other insurance-related issues (34% versus 16%)
 - — Among 9% of PP respondents who encountered a claims issue and reported a reason, 48% reported being told their PrEP medication was not covered by insurance, compared with 34% in the D/NP group

Figure 4. Reasons for Claims Processing Issue for People Who Reported at Least One Issue, by PrEP Pickup Group, USA, March–December, 2024

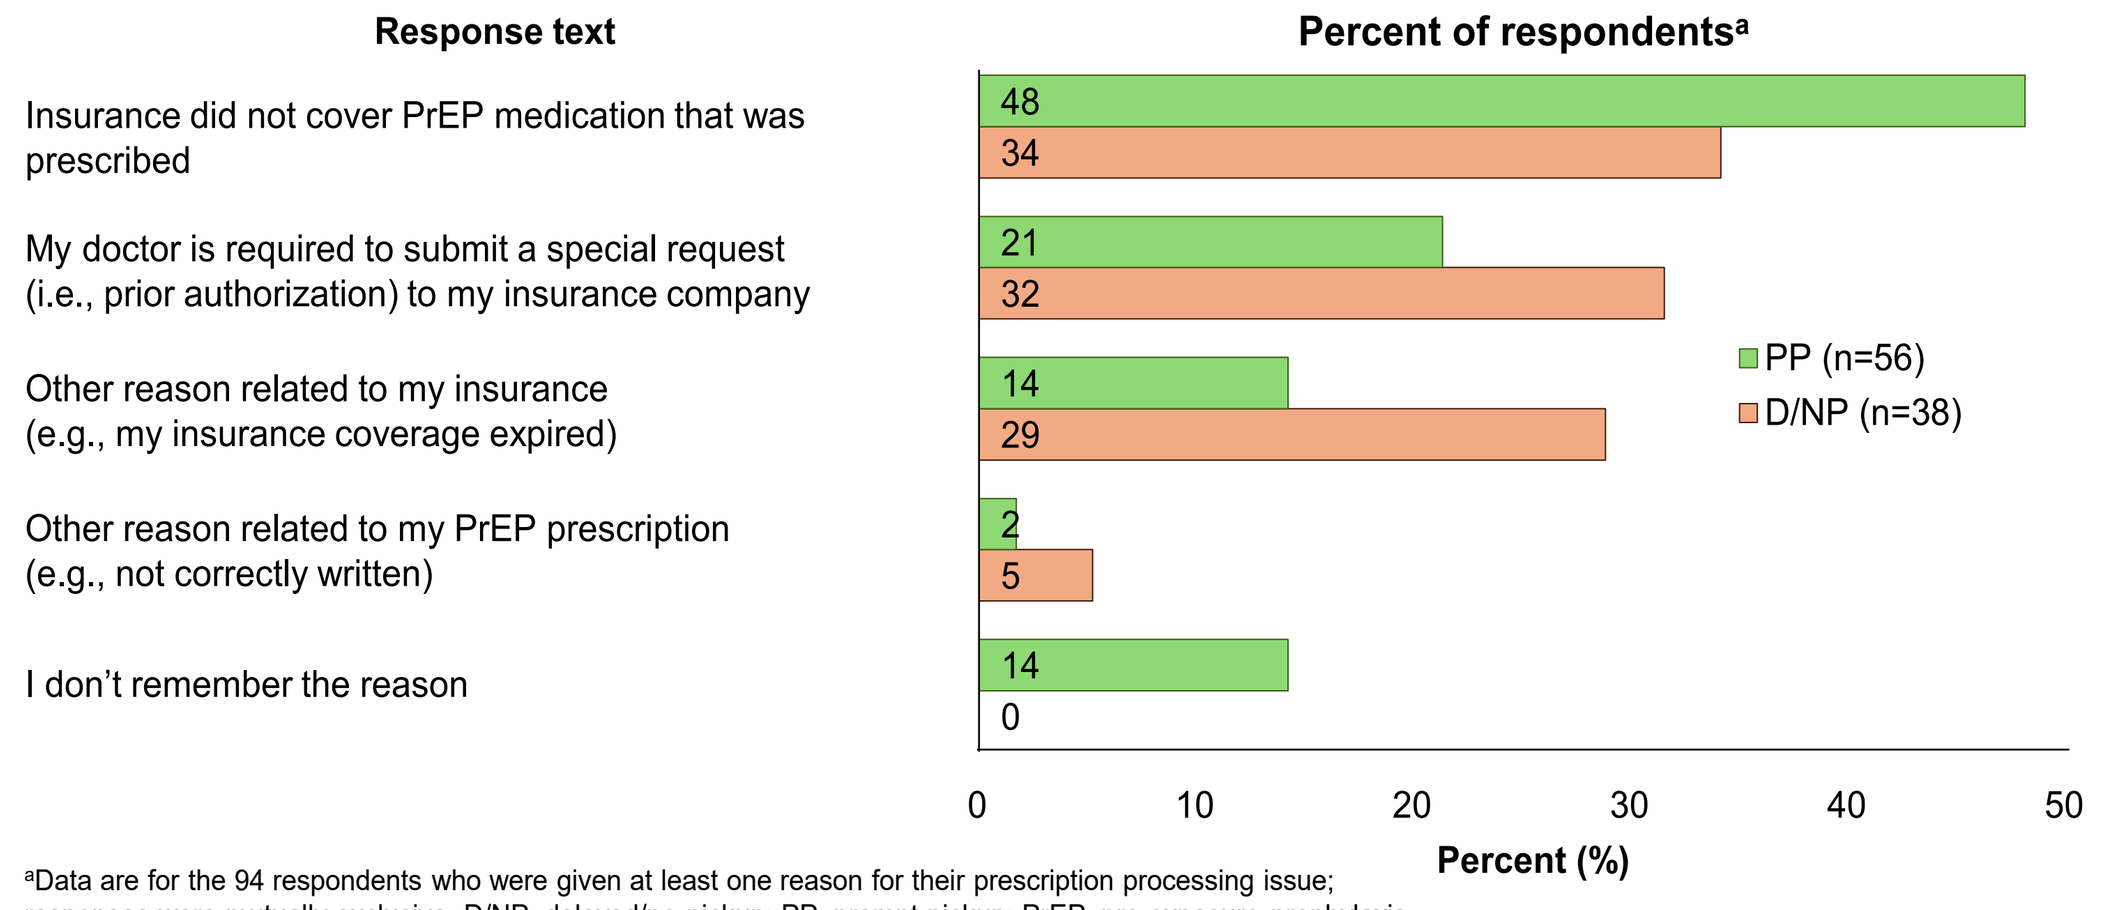
Response text
Percent of respondents[a]
Insurance did not cover PrEP medication that was prescribed
48
34
My doctor is required to submit a special request (i.e., prior authorization) to my insurance company
21
32
Other reason related to my insurance (e.g., my insurance coverage expired)
14
29
Other reason related to my PrEP prescription (e.g., not correctly written)
2
5
I don't remember the reason
14
0
PP (n=56)
D/NP (n=38)
0 10 20 30 40 50
Percent (%)

[a]Data are for the 94 respondents who were given at least one reason for their prescription processing issue; responses were mutually exclusive. D/NP, delayed/no pickup; PP, prompt pickup; PrEP, pre-exposure prophylaxis.

Limitations

- Possible biases due to the uneven distribution of Walgreens pharmacies, an unrepresentative sample due to non-participation, reliance on self-reported data, and inclusion of subjective questions may limit the generalizability of results to a broader population
- The cross-sectional survey design does not allow for the evaluation of factors that vary with time
- Barriers to PrEP dispensation are complex with interacting drivers that might not be identifiable in the survey
- Causal inferences cannot be drawn from this descriptive study, because many factors may contribute to variation between groups

References: **1.** CDC. Ending the HIV Epidemic in the US (EHE). Available at: https://www.cdc.gov/ehe/index.html (Accessed September 2025). **2.** US Preventative Services Task Force. Final Recommendation Statement: Preexposure Prophylaxis for the Prevention of HIV Infection. Available at https://www.uspreventiveservicestaskforce.org/uspstf/announcements/final-recommendation-statement-preexposure-prophylaxis-prevention-hiv-infection (Accessed September 2025). **3.** US Preventative Services Task Force. Prevention of Acquisition of HIV: Preexposure Prophylaxis, Final Recommendation Statement. Available at https://www.uspreventiveservicestaskforce.org/uspstf/recommendation/prevention-of-human-immunodeficiency-virus-hiv-infection-pre-exposure-prophylaxis (Accessed September 2025). **4.** CDC. Preventative Services Coverage. Available at https://www.cdc.gov/high-quality-care/hcp/resources/preventive-services-coverage.html (Accessed September 2025). **5.** AIDSVu. AIDSVu Releases 2024 PrEP Use Data Showing Growing Use Across the U.S. Available at: https://aidsvu.org/news-updates/aidsvu-releases-2024-prep-use-data-showing-growing-use-across-the-u-s (Accessed August 2025). **6.** Mayer KH, et al. *Adv Ther*. 2020;37(5):1778–811. **7.** Dean LT, et al. *Health Aff*. 2024;43(1):36–45. **8.** McManus KA, et al. *JAMA Network Open*. 2020;3(6):e207445. **9.** Patel K, et al. Perceived Barriers by Those Prescribed Initial HIV-1 Pre-Exposure Prophylaxis: US Pilot Survey Results. Poster presented at: AMCP: March 31–April 3, 2025; Houston, TX.

Acknowledgments: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Erin McMullin, PhD, and Sherriden Beard, MA, of Ashfield MedComms (Macclesfield, UK), an Inizio company, and funded by Gilead Sciences, Inc.

Conflicts of Interest: **KP** and **JC** are current or former employees and shareholders of Walgreens Co. **IS** was an employee of Walgreens at the time of the study. **JCC, AH, MB,** and **WZ** are employees and shareholders of Gilead Sciences, Inc. **PSS** has received research grants and speaker fees from Gilead Sciences Inc. He has also received research grants from Merck and ViiV Healthcare.

IDWeek 2025, October 19–22, 2025. Atlanta, GA, USA