Pooled Safety and Tolerability of Twice-yearly Lenacapavir with Teropavimab and Zinlirvimab For HIV-1 Treatment

P-365

Kimberly Workowski[1], Peter J Ruane[2], Anthony Mills[3], Mehri S. McKellar[4]*, Olayemi Osiyemi[5], Linda Gorgos[6], Gordon E. Crofoot[7], Moti Ramgopal[8], Cynthia Brinson[9], Kwad Mponponsuo[10], Sean E. Collins[10], Hui Liu[10], Nan Zhang[10], Keith Flower[10], Tina Chakrabarti[10], Onyema Ogbuagu[11]

[1]Department of Medicine, Emory University, Atlanta, GA, USA; [2]Ruane Clinical Research, Los Angeles, CA, USA; [3]Men's Health Foundation, Los Angeles, CA, USA; [4]Division of Infectious Diseases, Department of Medicine, Duke University School of Medicine, Durham, NC, USA; [5]Triple O Research Institute PA, West Palm Beach, FL, USA; [6]AXCES Research Group, Santa Fe, NM, USA; [7]The Crofoot Research Center, Houston, TX, USA; [8]Midway Immunology and Research Center, Fort Pierce, FL, USA; [9]Central Texas Clinical Research, Austin, TX, USA; [10]Gilead Sciences, Inc., Foster City, CA, USA; [11]Yale School of Medicine, New Haven, CT, USA.

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- In Phase 1b and Phase 2 studies, the safety profile of lenacapavir (LEN), teropavimab (TAB), and zinlirvimab (ZAB) was similar to oral antiretroviral therapy (ART) up to Week 26, with the exception of injection site reactions (ISRs) related to subcutaneous (SC) LEN administration
 - Overall, 67% of participants who received LEN, TAB, and ZAB in the pooled analysis experienced an adverse event (AE; excluding ISRs) versus 59% of Phase 2 participants who remained on their stable baseline oral ART regimen
- Most injection site reactions (ISRs) were mild (Grade 1); one participant experienced an infusion-related reaction (Grade 1), which resolved without intervention
- Grade ≥3 AEs and laboratory abnormalities were infrequent, and there were no serious AEs or AEs leading to study drug discontinuation
- Anti-drug antibodies against TAB and ZAB, detected in 13% and 18% of participants, respectively, were generally low in titer, and did not impact safety
- The ongoing Phase 2 study extension phase will continue to evaluate twice-yearly LEN, TAB, and ZAB treatment of HIV-1

Plain Language Summary

- Healthcare providers give lenacapavir as an injection under the skin to treat HIV in people who have tried many HIV medicines that no longer work well
- Researchers are testing teropavimab and zinlirvimab, antibodies given through a vein (infusion), as a new type of HIV treatment in combination with lenacapavir
- People can receive lenacapavir, teropavimab, and zinlirvimab once every 6 months, unlike most HIV treatments that require daily pill intake
- Researchers tested this combination in two studies with people who had viral suppression (very low or no HIV in their blood)
- They looked at side effects in everyone who received the combination treatment
- The combination showed favorable safety. People had side effects at similar rates as those taking approved daily HIV pills, except for skin reactions at the lenacapavir injection site
- The most common side effects were skin reactions at the lenacapavir injection site
- No one stopped treatment because of side effects

Introduction

- LEN, the first-in-class HIV-1 capsid inhibitor, can be administered subcutaneously twice-yearly and is approved for the treatment of multidrug-resistant HIV-1 and for HIV-1 prevention[1–5]
- TAB and ZAB are broadly neutralizing antibodies (bNAbs) targeting the CD4-binding site and a non-overlapping epitope on the V3 glycan of the HIV-1 envelope trimer, respectively **(Figure 1)**[6]
 - TAB and ZAB have extended half-lives that allow for twice-yearly dosing
- The combination of LEN, TAB, and ZAB is being investigated as the first complete twice-yearly treatment regimen for HIV-1
 - At Week 26, high rates of virologic suppression were maintained for participants receiving LEN, TAB and ZAB in Phase 1b and Phase 2 studies[7,8]

Figure 1. HIV-1 Envelope Trimer Antibody Binding Sites

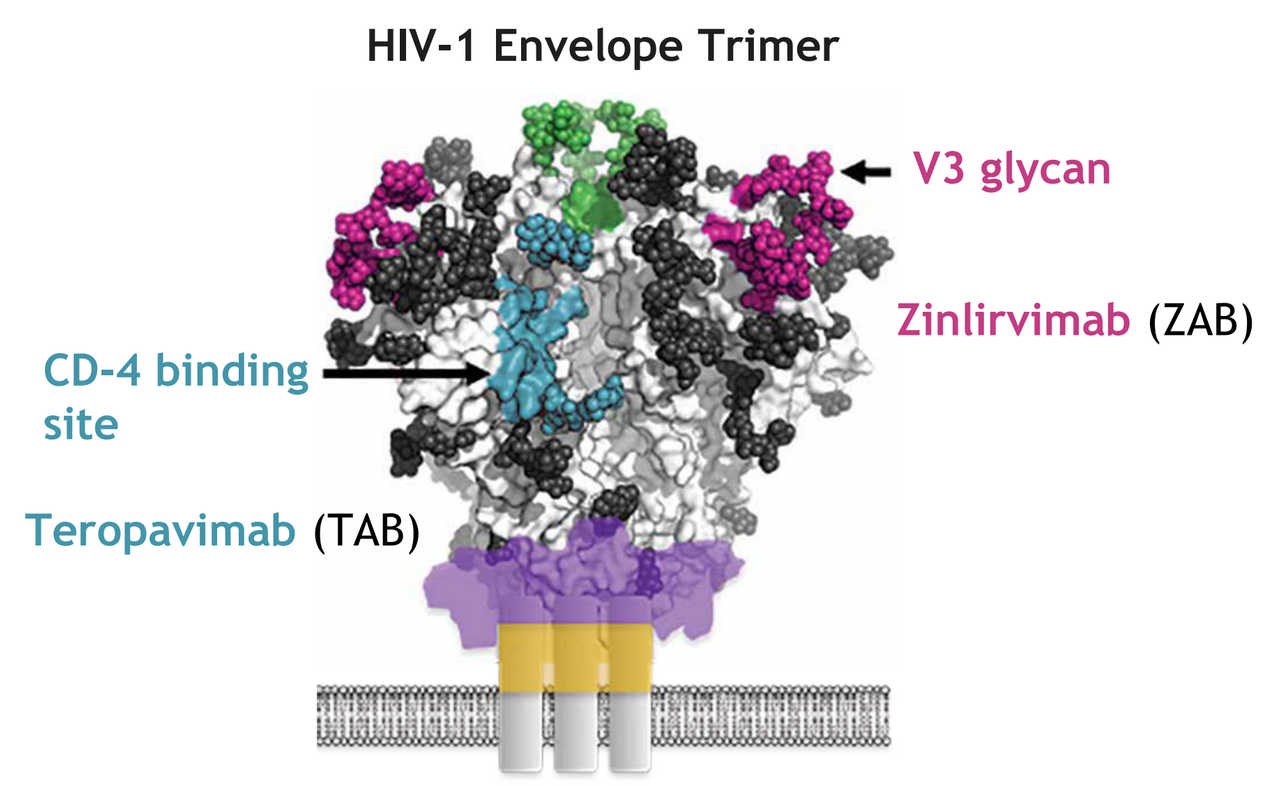
HIV-1 Envelope Trimer
V3 glycan
Zinlirvimab (ZAB)
CD-4 binding site
Teropavimab (TAB)

Objective

- To evaluate pooled Phase 1b and Phase 2 safety and tolerability of LEN, TAB, and ZAB after one dose, including the effect of anti-drug antibodies against TAB and ZAB on safety

Methods

- Both the Phase 1b **(Figure 2A)** and Phase 2 **(Figure 2B)** studies enrolled virologically suppressed (HIV-1 RNA <50 copies/mL) adults with HIV-1

Figure 2. Phase 1b (A) and Phase 2 (B) Study Designs

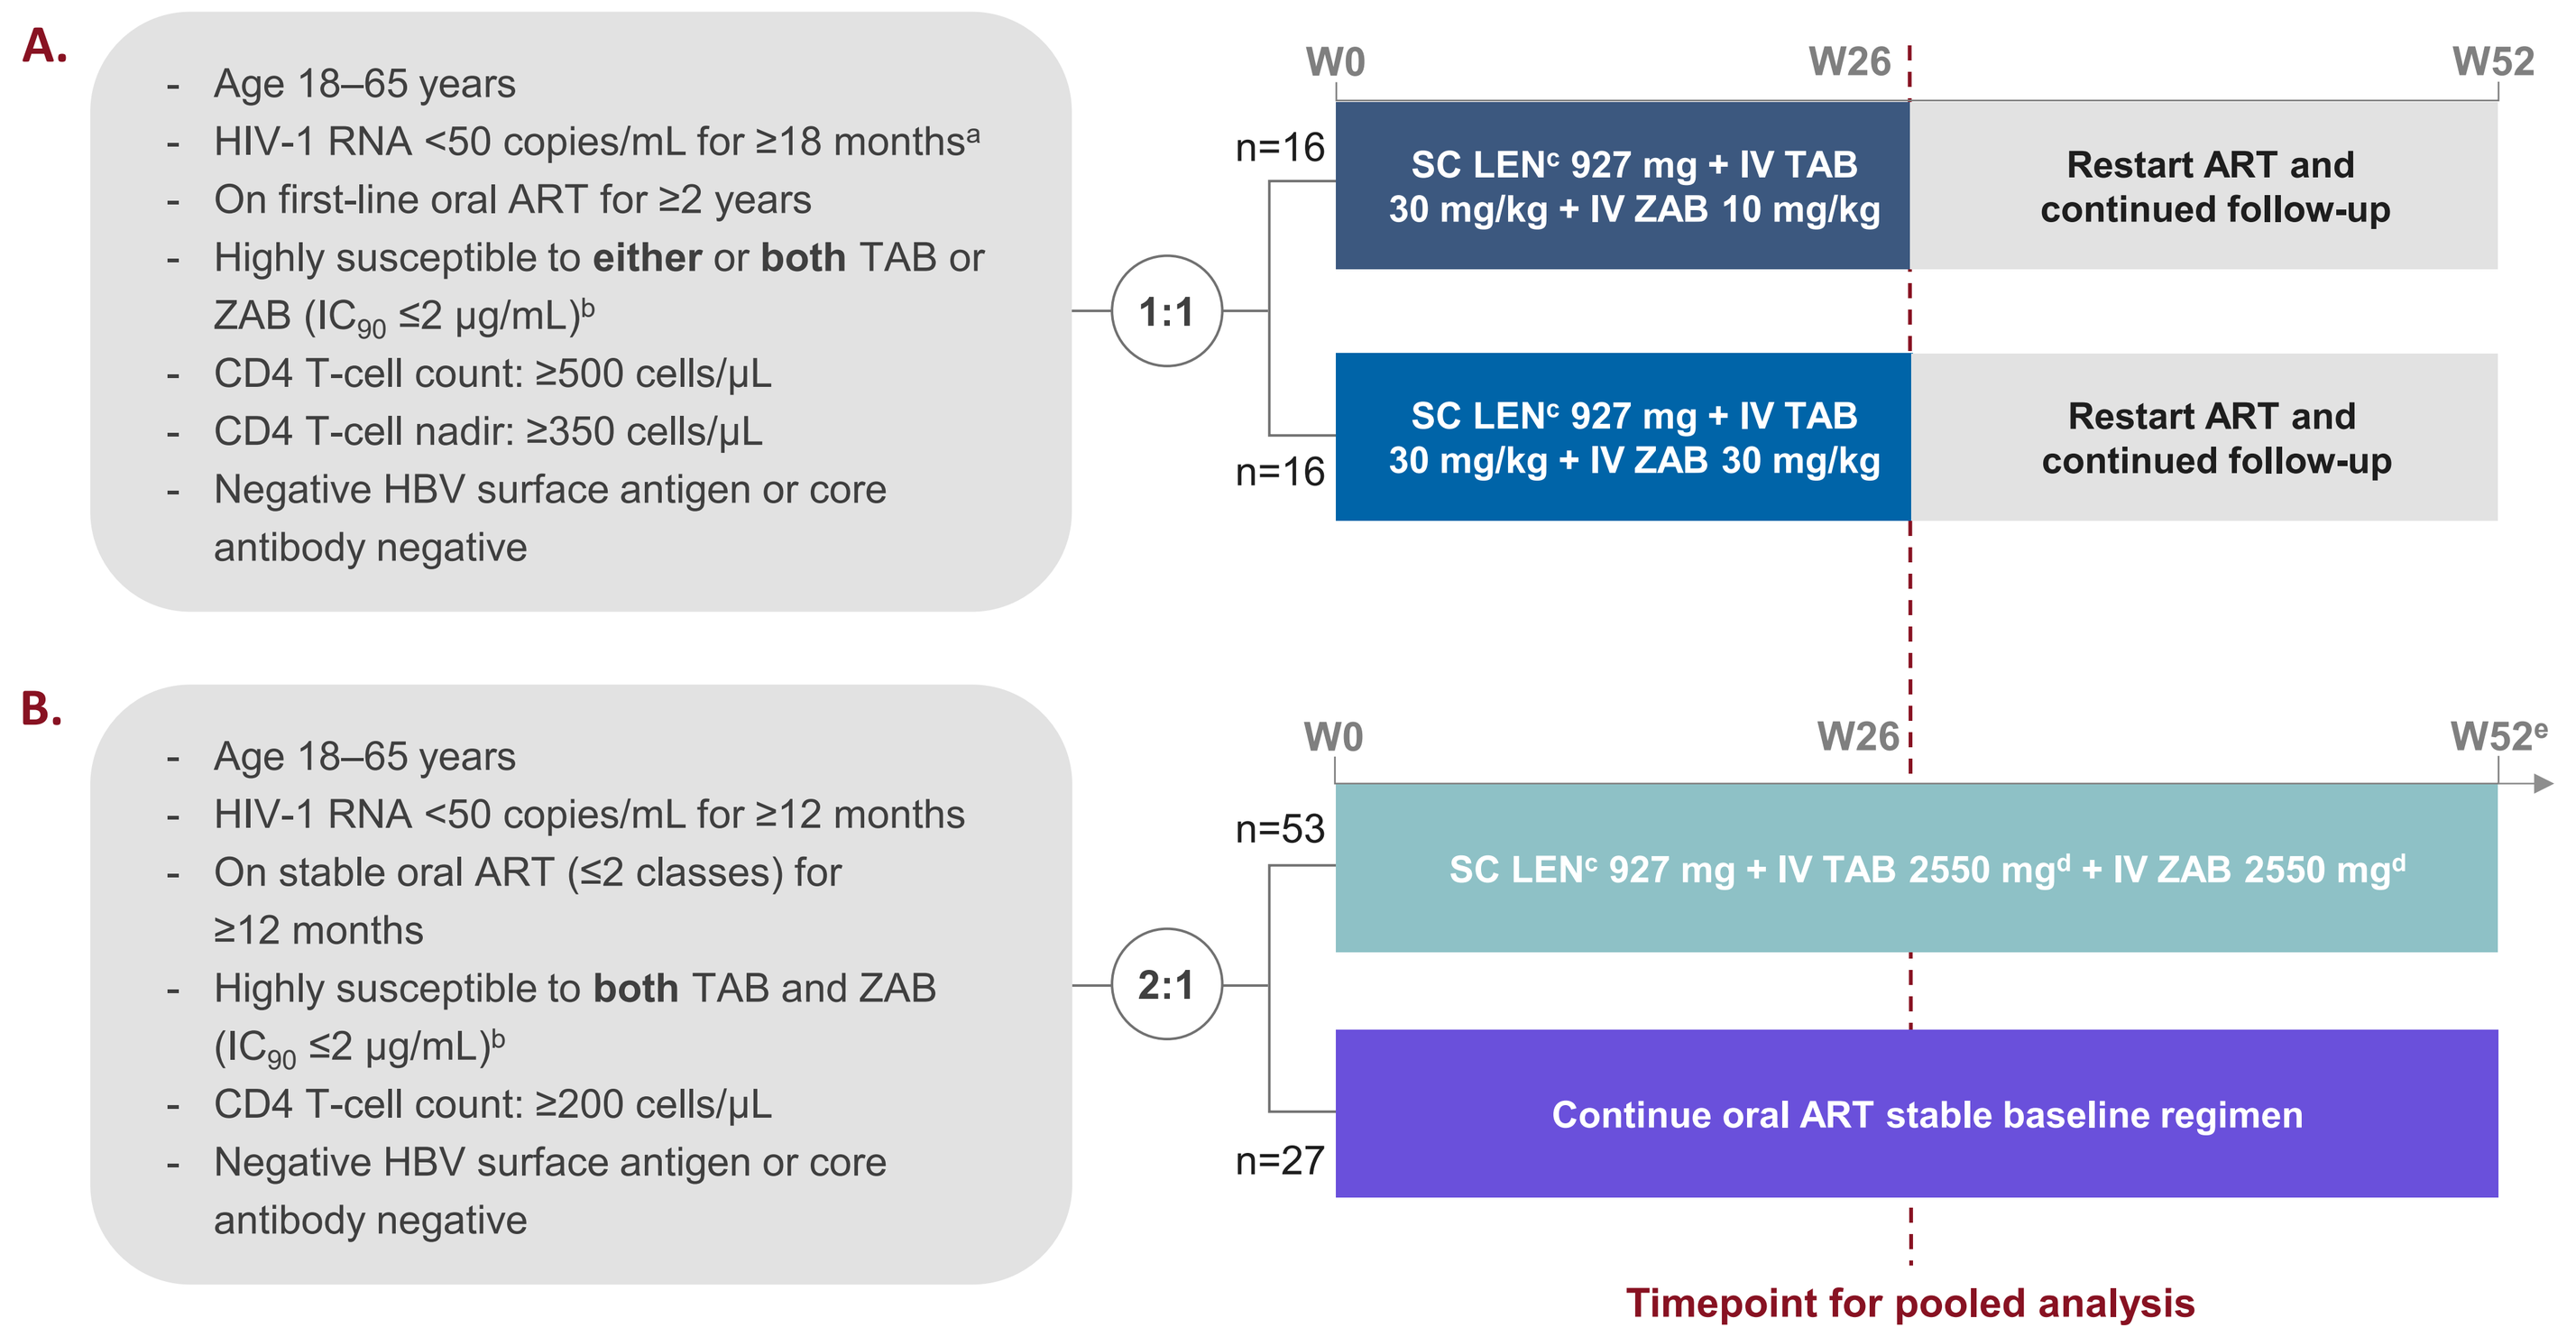
A.
- Age 18–65 years
- HIV-1 RNA <50 copies/mL for ≥18 months[a]
- On first-line oral ART for ≥2 years
- Highly susceptible to either or both TAB or ZAB (IC_{90} ≤2 µg/mL)[b]
- CD4 T-cell count: ≥500 cells/µL
- CD4 T-cell nadir: ≥350 cells/µL
- Negative HBV surface antigen or core antibody negative
1:1
W0
W26
W52
n=16
SC LEN[c] 927 mg + IV TAB 30 mg/kg + IV ZAB 10 mg/kg
Restart ART and continued follow-up
n=16
SC LEN[c] 927 mg + IV TAB 30 mg/kg + IV ZAB 30 mg/kg
Restart ART and continued follow-up
B.
- Age 18–65 years
- HIV-1 RNA <50 copies/mL for ≥12 months
- On stable oral ART (≤2 classes) for ≥12 months
- Highly susceptible to both TAB and ZAB (IC_{90} ≤2 µg/mL)[b]
- CD4 T-cell count: ≥200 cells/µL
- Negative HBV surface antigen or core antibody negative
2:1
W0
W26
W52[e]
n=53
SC LEN[c] 927 mg + IV TAB 2550 mg[d] + IV ZAB 2550 mg[d]
n=27
Continue oral ART stable baseline regimen
Timepoint for pooled analysis

[a]Previous virologic failure was allowed if participants were virologically suppressed (HIV-1 RNA <50 copies/mL) for ≥18 months prior to screening. [b]By PhenoSense® mAb Assay (Monogram Biosciences). [c]Plus LEN 600 mg orally on Days 1 and 2 for pharmacological loading. [d]The fixed doses of 2550 mg for TAB and ZAB in the Phase 2 study were selected to produce similar exposures to the 30 mg/kg weight-based dose in the Phase 1b study. [e]Followed by a post-Week 52 extension phase in which all participants received LEN, TAB, and ZAB.
ART, antiretroviral therapy; **HBV**, Hepatitis B virus; **IC_{90}**, 90% inhibitory concentration; **IV**, intravenous; **LEN**, lenacapavir; **SC**, subcutaneous; **TAB**, teropavimab; **W**, Week; **ZAB**, zinlirvimab.

- Data from all randomized participants who received one dose of the complete LEN, TAB, and ZAB regimen were pooled in this analysis **(Figure 3)**
- Safety was assessed by incidence of AEs and laboratory abnormalities through Week 26
 - Infusion-related reactions were identified by investigator assessment
- Biologic medications, such as bNAbs, may elicit an immune response against the drug, thereby potentially affecting drug exposure, efficacy, and/or safety[9]; therefore, AEs occurring in participants with anti-drug antibodies were reviewed individually for potential relationships between anti-drug antibodies and safety
 - Anti-drug antibodies against TAB and ZAB were determined from samples taken at baseline and at protocol-specified visits after dosing with LEN, TAB and ZAB using validated bioanalytical methods[10]
- Stable baseline ART group data from the Phase 2 study were used as a reference group for standard-of-care regimens and are not intended for direct comparison

Figure 3. Pooled Analysis of Phase 1b and Phase 2 LEN, TAB, and ZAB Participants

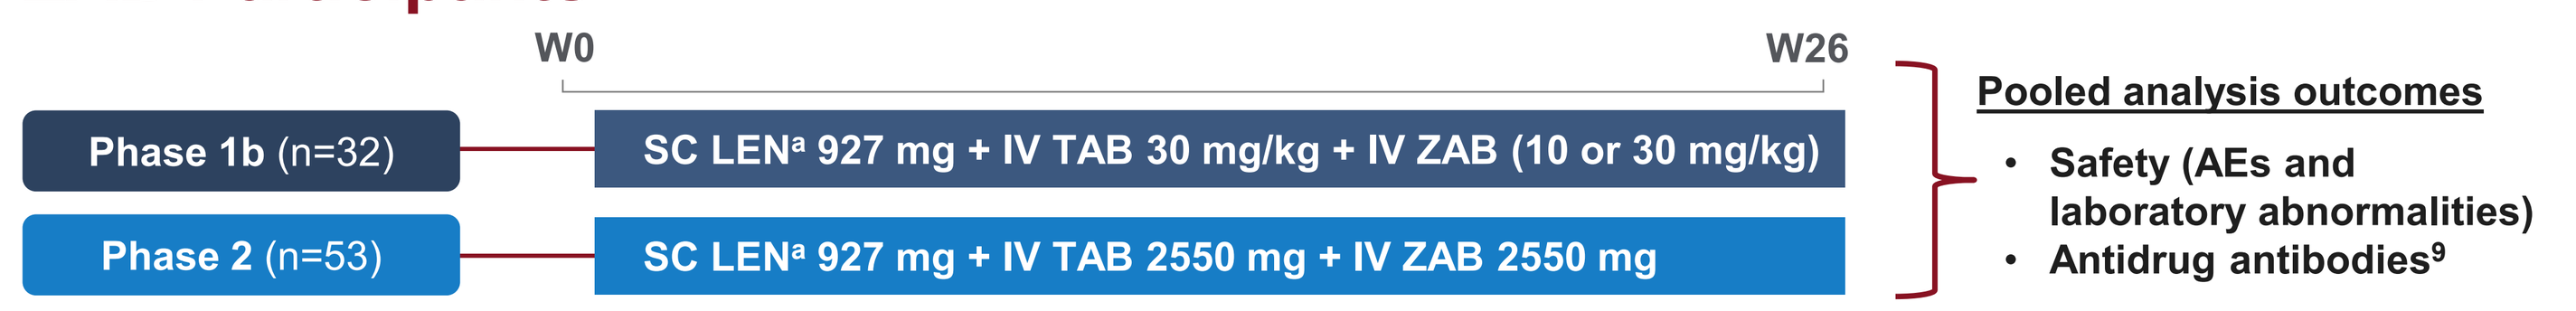
W0
W26
Phase 1b (n=32)
SC LEN[a] 927 mg + IV TAB 30 mg/kg + IV ZAB (10 or 30 mg/kg)
Phase 2 (n=53)
SC LEN[a] 927 mg + IV TAB 2550 mg + IV ZAB 2550 mg
Pooled analysis outcomes
- Safety (AEs and laboratory abnormalities)
- Antidrug antibodies[9]

[a]Plus LEN 600 mg orally on Days 1 and 2 for pharmacological loading.
AE, adverse event; **IV**, intravenous; **LEN**, lenacapavir; **SC**, subcutaneous; **TAB**, teropavimab; **W**, Week; **ZAB**, zinlirvimab.

Results

Baseline Characteristics

- A total of 84 participants were included in the pooled analysis (Phase 1b, n=31; Phase 2, n=53)
 - One participant from the Phase 1b study received oral LEN only and was excluded from the analysis
- The pooled participants had a median age of 46 years and 17% were assigned female at birth **(Table 1)**, compared with 57 years and 15%, respectively, in the stable oral ART group

Table 1. Baseline Characteristics

	Pooled LEN, TAB, and ZAB (n=84)	Phase 2 – Stable Oral ART (n=27)
Median (range) age, years	46 (20–65)	57 (28–65)
Assigned female sex at birth, n (%)	14 (17)	4 (15)
Mean (SD) CD4 T-cells/µL	838 (309.8)	768 (260.0)
Race, n (%)		
Asian	3 (4)	1 (4)
Black	28 (33)	8 (30)
White	46 (55)	16 (59)
Other	7 (8)	2 (7)
Hispanic or Latine ethnicity, n (%)	22 (26)	7 (26)
US region, n (%)	79 (94)	19 (70)
Median (IQR) weight, kg	90 (81–106)	87 (58–157)
Median (IQR) BMI, kg/m²	29.0 (25.8–34.1)	29.2 (25.5–32.0)
Median (IQR) time since diagnosis, years[a]	12.7 (7.1–19.2)	18.4 (12.4–27.3)
Median (IQR) time since ART initiation, years[a]	11.4 (7.1–17.4)	16.4 (10.4–23.4)

[a]Self-reported by participants.
ART, antiretroviral therapy; **BMI,** body mass index; **IQR,** interquartile range; **LEN**, lenacapavir; **TAB**, teropavimab; **ZAB**, zinlirvimab.

Safety

- Median duration of study drug exposure at time of analysis was 26 weeks in both the pooled cohort and the stable oral ART group
- In the pooled cohort, 56 (67%) participants experienced an AE, excluding ISRs **(Table 2)**
 - There were no serious AEs, or AEs leading to study drug or study discontinuation
- One participant experienced an infusion-related reaction (Grade 1) which resolved without intervention

Table 2. AE Overview

Participants, n (%)	Pooled LEN, TAB, and ZAB (n=84)	Phase 2 - Stable Oral ART (n=27)
AEs, excluding ISRs	56 (67)[a]	16 (59)
Grade ≥2	26 (31)	3 (11)
Grade ≥3	2 (2)[b]	2 (7)
Treatment-related AEs, excluding ISRs	9 (11)[c]	0
Grade ≥2	3 (4)[d]	0
Grade ≥3	1 (1)[e]	0
Serious AEs	0	1 (4)[f]
AEs leading to study or study drug discontinuation	0	1 (4)[f]

Exclusion of ISRs only refers to the pooled LEN, TAB, and ZAB group only, as it is not applicable to the stable oral ART group. [a]Including ISRs, 71 participants (85%) had an AE. [b]Acute pyelonephritis, ureteritis, and nephrolithiasis in one participant; injection site cellulitis in one participant. [c]Treatment-related AEs are defined as AEs considered related to study drug by the investigator. Including ISRs, 57 participants (68%). Increased lacrimation, diarrhea, headache, nausea, abnormal dreams, insomnia, device dislocation, infusion-related reaction, injection site cellulitis (n=1 each). [d]Injection site cellulitis, diarrhea, insomnia (n=1 each). [e]Injection site cellulitis. [f]Metastatic pancreatic cancer.
AE, adverse event; **ART**, antiretroviral therapy; **ISR**, injection site reaction; **LEN**, lenacapavir; **TAB**, teropavimab; **ZAB**, zinlirvimab.

- ISRs related to SC LEN were reported in 55 (65%) pooled cohort participants, the majority of whom (85%; 47/55) experienced mild (Grade 1) ISRs **(Figure 4)**
 - One Grade 3 ISR of erythema was reported which resolved without intervention
 - Pain and erythema had a median duration of 11 and 15 days, respectively, until resolution **(Table 3)**
 - Nodules and induration had longer median durations **(Table 3)**; these decreased in size over time, with resolution by Week 26 in 9/28 (32%) people with nodules and 10/16 (63%) with induration
- Grade ≥3 laboratory abnormalities occurred in 5 (6%) participants from the LEN, TAB, and ZAB group (0% in the stable oral ART group), were considered unrelated to study drug, and were not deemed clinically significant by the investigator
 - Grade 3 laboratory abnormalities were high creatine kinase (n=2), glycosuria (n=2), and non-fasting hyperglycaemia (n=1); one Grade 4 laboratory abnormality of high lipase occurred

Figure 4. SC LEN-Related ISRs[a] Occurring in >10% of Pooled Cohort Participants (n=84)

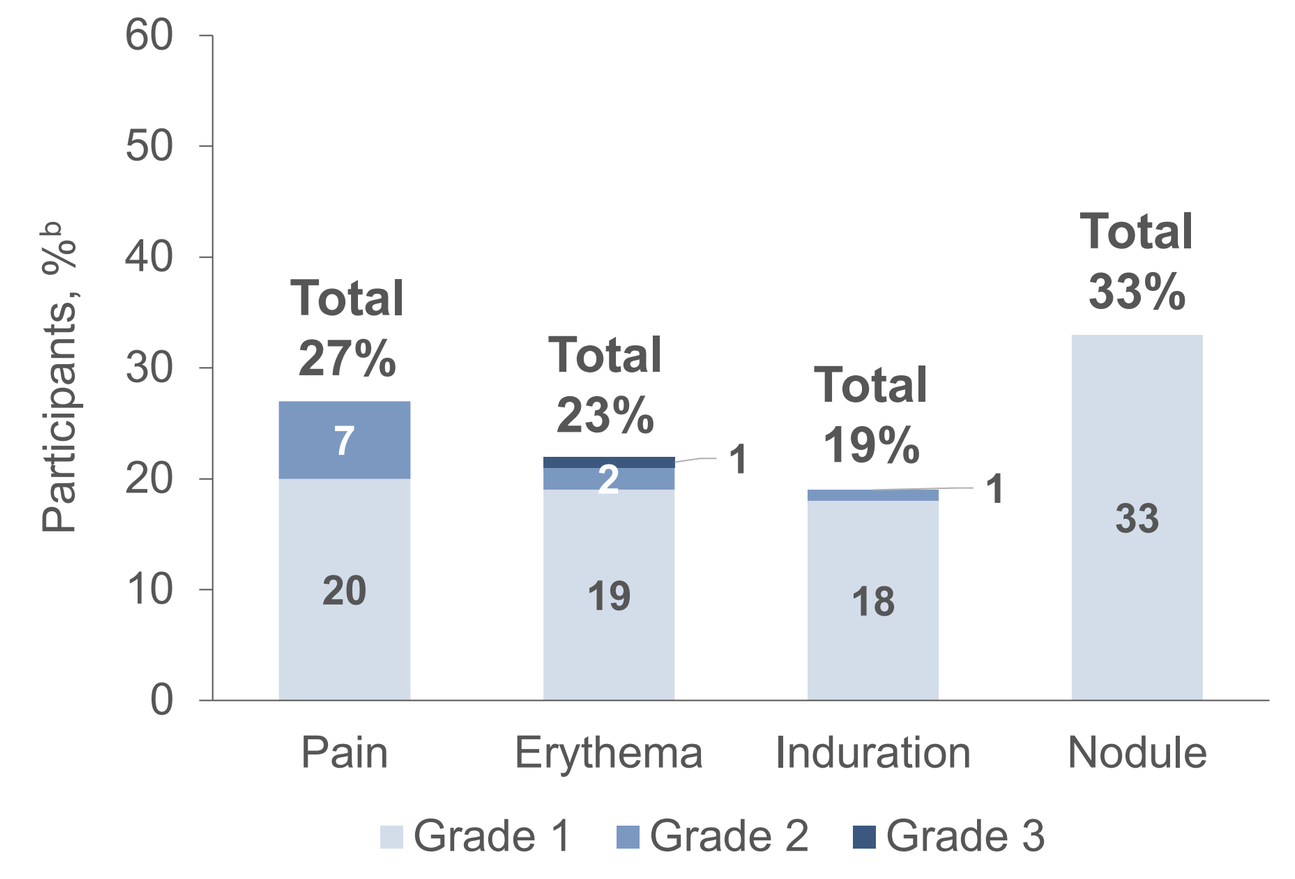
Participants, %[b]
60
50
40
30
20
10
0
Total 27%
7
20
Total 23%
2
1
19
Total 19%
1
18
Total 33%
33
Pain
Erythema
Induration
Nodule
Grade 1
Grade 2
Grade 3

Values have been rounded to a whole percent, therefore columns may not add to the total values. [a]ISRs were defined as AEs matching the Medical Dictionary of Regulatory Activities (MedDRA) v27.1[11] high-level terms of "Injection site reactions".
AE, adverse event; **ISR**, injection site reaction; **LEN**, lenacapavir; **SC**, subcutaneous.

Table 3. Median Duration of SC LEN-Related ISRs[a] Occurring in >10% of Participants

	Median (range) duration, days LEN, TAB, and ZAB (n=55)
Any ISR	39 (1–386)
Pain	11 (1–232)
Erythema	15 (1–265)
Induration	232 (9–365)
Nodule	246 (5–386)

[a]ISRs were defined as AEs matching the Medical Dictionary of Regulatory Activities (MedDRA) v27.1[11] high-level terms of "Injection site reactions".
AE, adverse event; **ISR**, injection site reaction; **LEN**, lenacapavir; **SC**, subcutaneous.

Anti-Drug Antibodies

- Anti-drug antibodies against TAB were detected in 11/83 (13%) participants and against ZAB in 15/83 (18%) participants
 - Anti-drug antibodies against TAB and ZAB, respectively, were persistent (positive result from two samples at least 16 weeks apart, or at the last sample prior to end of study) in 9/11 and 13/15 participants and transient in two participants each
- Anti-drug antibodies were generally observed at low titers
- There was no identifiable effect of anti-drug antibodies on drug exposure and safety; for further data on anti-drug antibodies in the Phase 2 study, presented in IDWeek 2025 Poster P-1248[10], please scan the QR code on this poster to view

References: **1.** Subramanian R, et al. *Mol Pharm.* 2023;20:6213-25. **2.** Sunlenca® Prescribing Information. Available at: https://www.gilead.com/-/media/files/pdfs/medicines/hiv/sunlenca/sunlenca_pi.pdf (Accessed October 2025). **3.** Sunlenca® Summary of Product Characteristics. Available at: https://www.ema.europa.eu/en/documents/product-information/sunlenca-epar-product-information_en.pdf (Accessed October 2025). **4.** Yeztugo® Prescribing information. Available at: https://www.gilead.com/-/media/files/pdfs/medicines/hiv/yeztugo/yeztugo_pi.pdf (Accessed October 2025). **5.** Yeytuo ® Summary of Product Characteristics. Available at: https://www.ema.europa.eu/en/documents/product-information/yeytuo-epar-product-information_en.pdf. (Accessed October 2025). **6.** Gautam R, et al. Nat Med. 2018;24:610–6. **7.** Little S, et al. Presented at HIV Glasgow 2024, November 10–13, Glasgow, UK; Presentation O23. **8.** Ogbuagu O, et al. Presented at CROI 2025, March 9–12, San Francisco, California, US; Presentation 151. **9.** Swanson SJ. *Front Immunol.* 2024;15:1401178. **10.** Zhang N, et al. Presented at IDWeek 2025, October 19–22, Atlanta, Georgia, USA; Poster P-1248. **11.** Maintenance and Support Services Organization. *Medical Dictionary for Regulatory Activities (MedDRA) version 27.1*. Available at: https://www.meddra.org/. (Accessed October 2025)

Acknowledgments: We extend our thanks to the participants and their families, and to the participating investigators and staff
This study was funded by Gilead Sciences, Inc., Foster City, CA, USA.
Medical writing support was provided by Jessica Woods of Ashfield MedComms (Macclesfield, UK), an Inizio company, and funded by Gilead Sciences, Inc.

Disclosures: **Kimberly Workowski:** Gilead Sciences, Inc. (research funding). **Peter J Ruane:** Gilead Sciences, Inc. and ViiV (advisor/consultant and honoraria). **Mehri S. McKellar:** Gilead Sciences, Inc. (research funding). **Anthony Mills:** no disclosures to report. **Olayemi Osiyemi:** Gilead Sciences, Inc. (advisor/consultant, speaker, stock), ViiV (advisor/consultant and speaker), and Merck (advisor/consultant). **Linda Gorgos:** Gilead Sciences, Inc., Merck, GSK, and ViiV (grants/contract payments); AIDS Education and Training Center – South Central (honoraria). **Gordon E. Crofoot:** AbbVie, Gilead Sciences, Inc., Janssen Pharmaceuticals, Merck, and ViiV (grants/contract payments), and Gilead Sciences, Inc. (support for attending meetings). **Moti Ramgopal:** Gilead Sciences, Inc. (advisor/consultant, research support, and honoraria), ViiV (advisor/consultant, honoraria), Merck and Abbott (advisor/consultant), and AbbVie (honoraria). **Cynthia Brinson:** Gilead Sciences, Inc. (advisor/consultant, grant/research support, honoraria, medical writing funds), ViiV (advisor/consultant, honoraria), and GSK (honoraria). **Kwad Mponponsuo, Sean E. Collins, Hui Liu, Nan Zhang, Keith Flower, and Tina Chakrabarti** are all employees and shareholders of Gilead Sciences, Inc. **Onyema Ogbuagu:** Gilead Sciences, Inc. and ViiV (advisory boards).

Correspondence: Tina Chakrabarti: tina.chakrabarti@gilead.com

IDWeek 2025, October 19–22, 2025; Atlanta, GA, USA